Targeted epigenomic control of the MYC oncogene robustly inhibits survival and growth of Non-Small Cell Lung Cancer organoid and xenograft models

Eugine Lee, Defne Yarar, Houda Belaghzal, Kai-Yuan Chen, Cameron Vergato, Padraich Flahardy, Stephen Siecinski, Charles O'Donnell, Joseph Newman, Thomas McCauley

Omega Therapeutics, Cambridge, MA, USA

Gordon Research Conferences

BACKGROUND

Increasing interest has focused on the development of epigenetic therapies to modulate gene expression pre-transcriptionally by harnessing epigenetic processes such as DNA methylation and histone modification. Frequent mutations in epigenetic enzymes as well as non-coding genomic sites involved in epigenetic regulation of gene expression, such as promoters and enhancers, have been implicated in cancer and other diseases. Targeting these dysregulated epigenomic sites would allow the specific and effective modulation of a wide range of targets, including those deemed undruggable such as the oncogene MYC. Epigenetic modulation of such targets has been an area of increasing interest and there are therapies in development in cancer and other diseases.

MYC is a master transcription factor that is critical for multiple cell physiologies and whose activity is frequently dysregulated in cancer, including non-small cell lung cancer (NSCLC). NSCLC is the leading cause of global cancer-related mortality, making up almost 25% of all cancer deaths. The MYC gene and its regulatory elements reside alone within an insulated genomic domain (IGD), a chromatin-looping region mediated by CTCF and other factors. MYC represents an attractive target for pre-transcriptional gene modulation via an epigenetic approach, as epigenetic changes in MYC have been demonstrated in multiple cancers.

Here, we describe a NSCLC-specific MYC-targeted epigenomic mRNA therapy, an Omega epigenomic controller (OEC), (NSCLC MYC-OEC), targeting two sites within the MYC IGD in order to tune MYC expression pre-transcriptionally with high specificity and durability.

Figure 1. Structure and Mechanism of Action of OECs

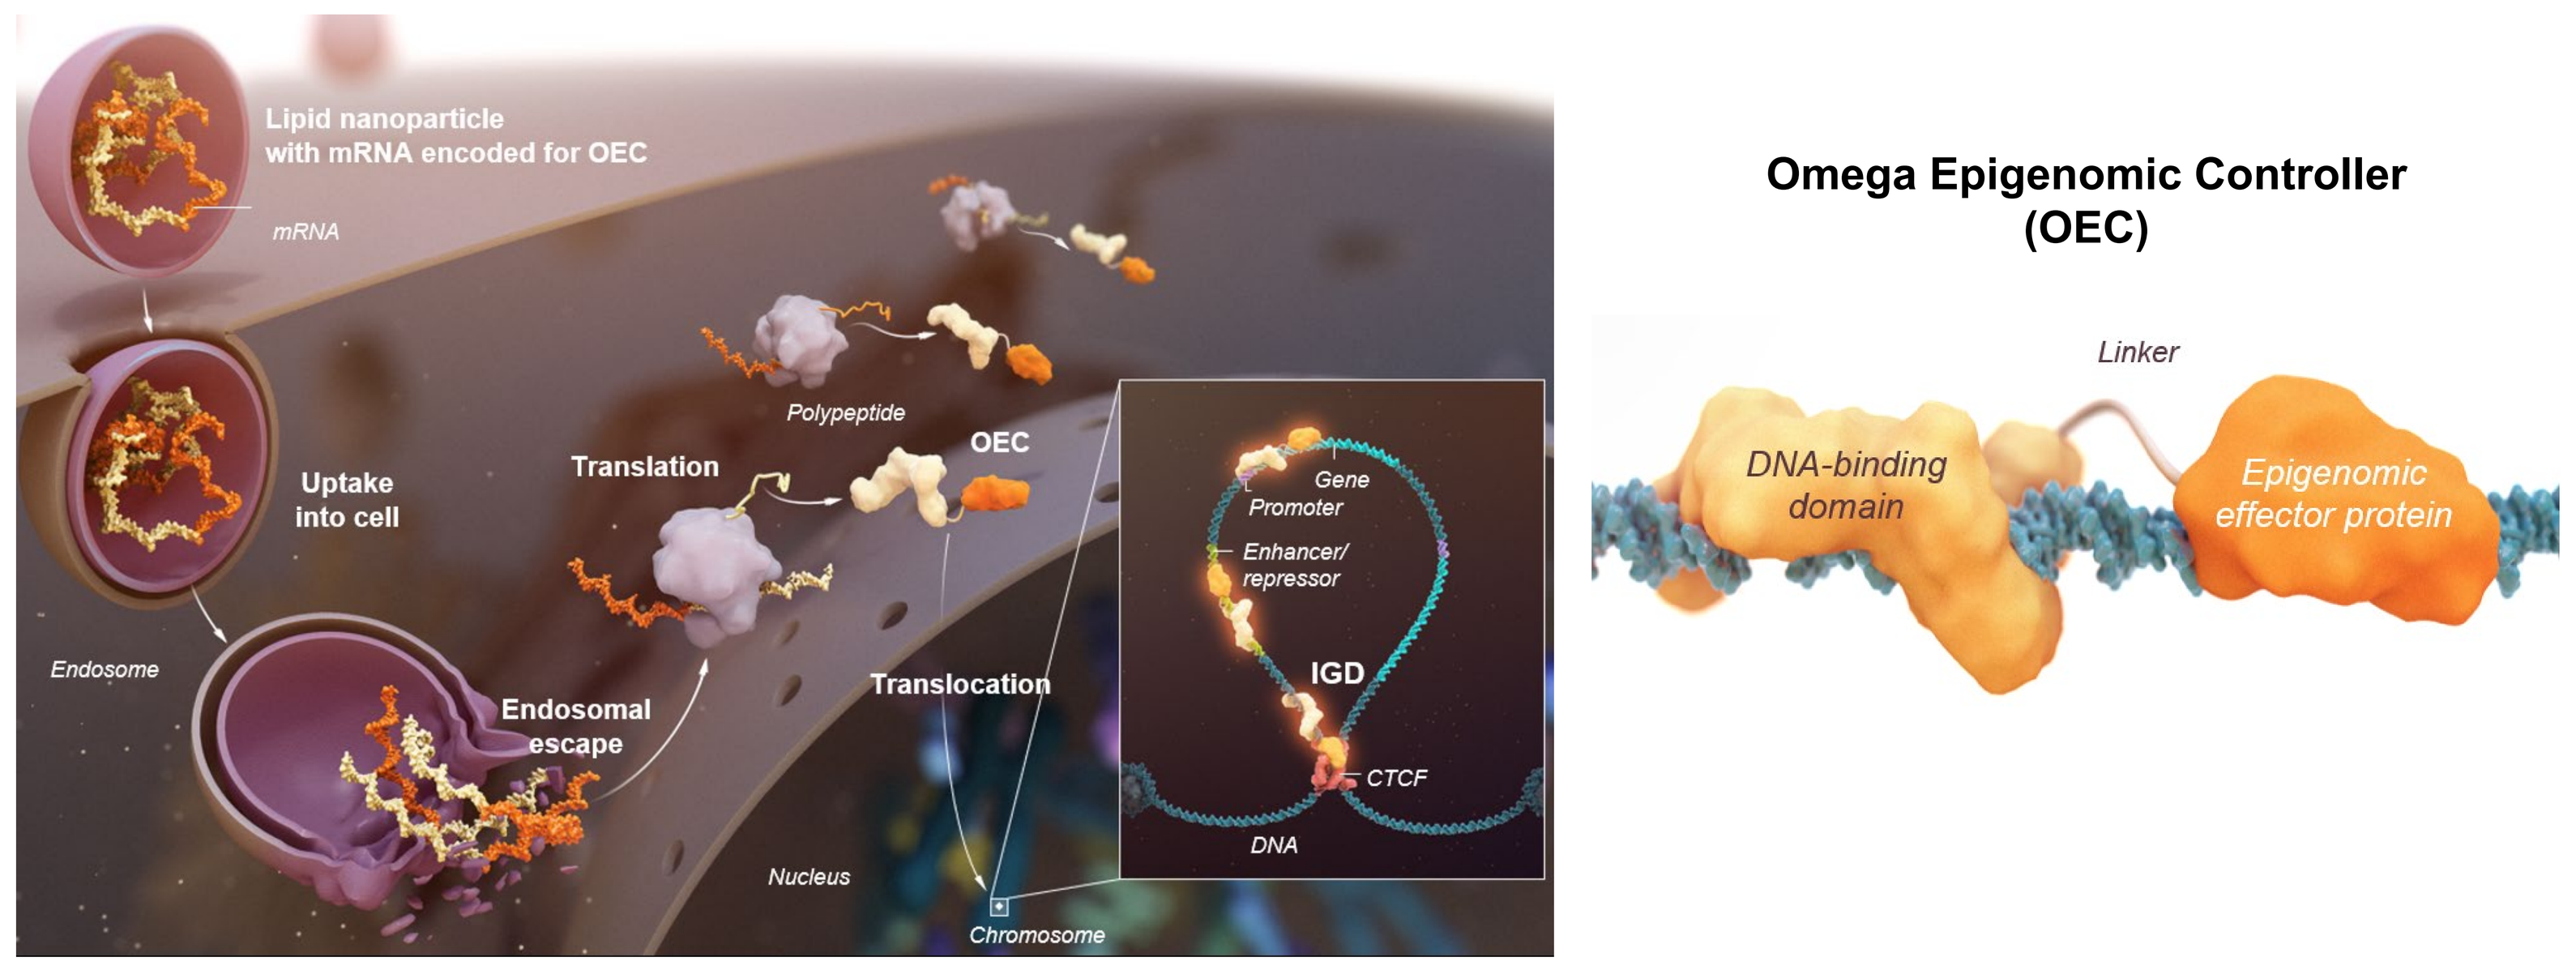
Lipid nanoparticle with mRNA encoded for OEC
mRNA
Uptake into cell
Translation
Polypeptide
OEC
Endosome
Endosomal escape
Translocation
Nucleus
Chromosome
Gene Promoter
Enhancer/ repressor
IGD
CTCF
DNA
Omega Epigenomic Controller (OEC)
Linker
DNA-binding domain
Epigenomic effector protein

Figure 2 & 3. On-target effect of a NSCLC MYC-OEC targeting two different sites within MYC IGD

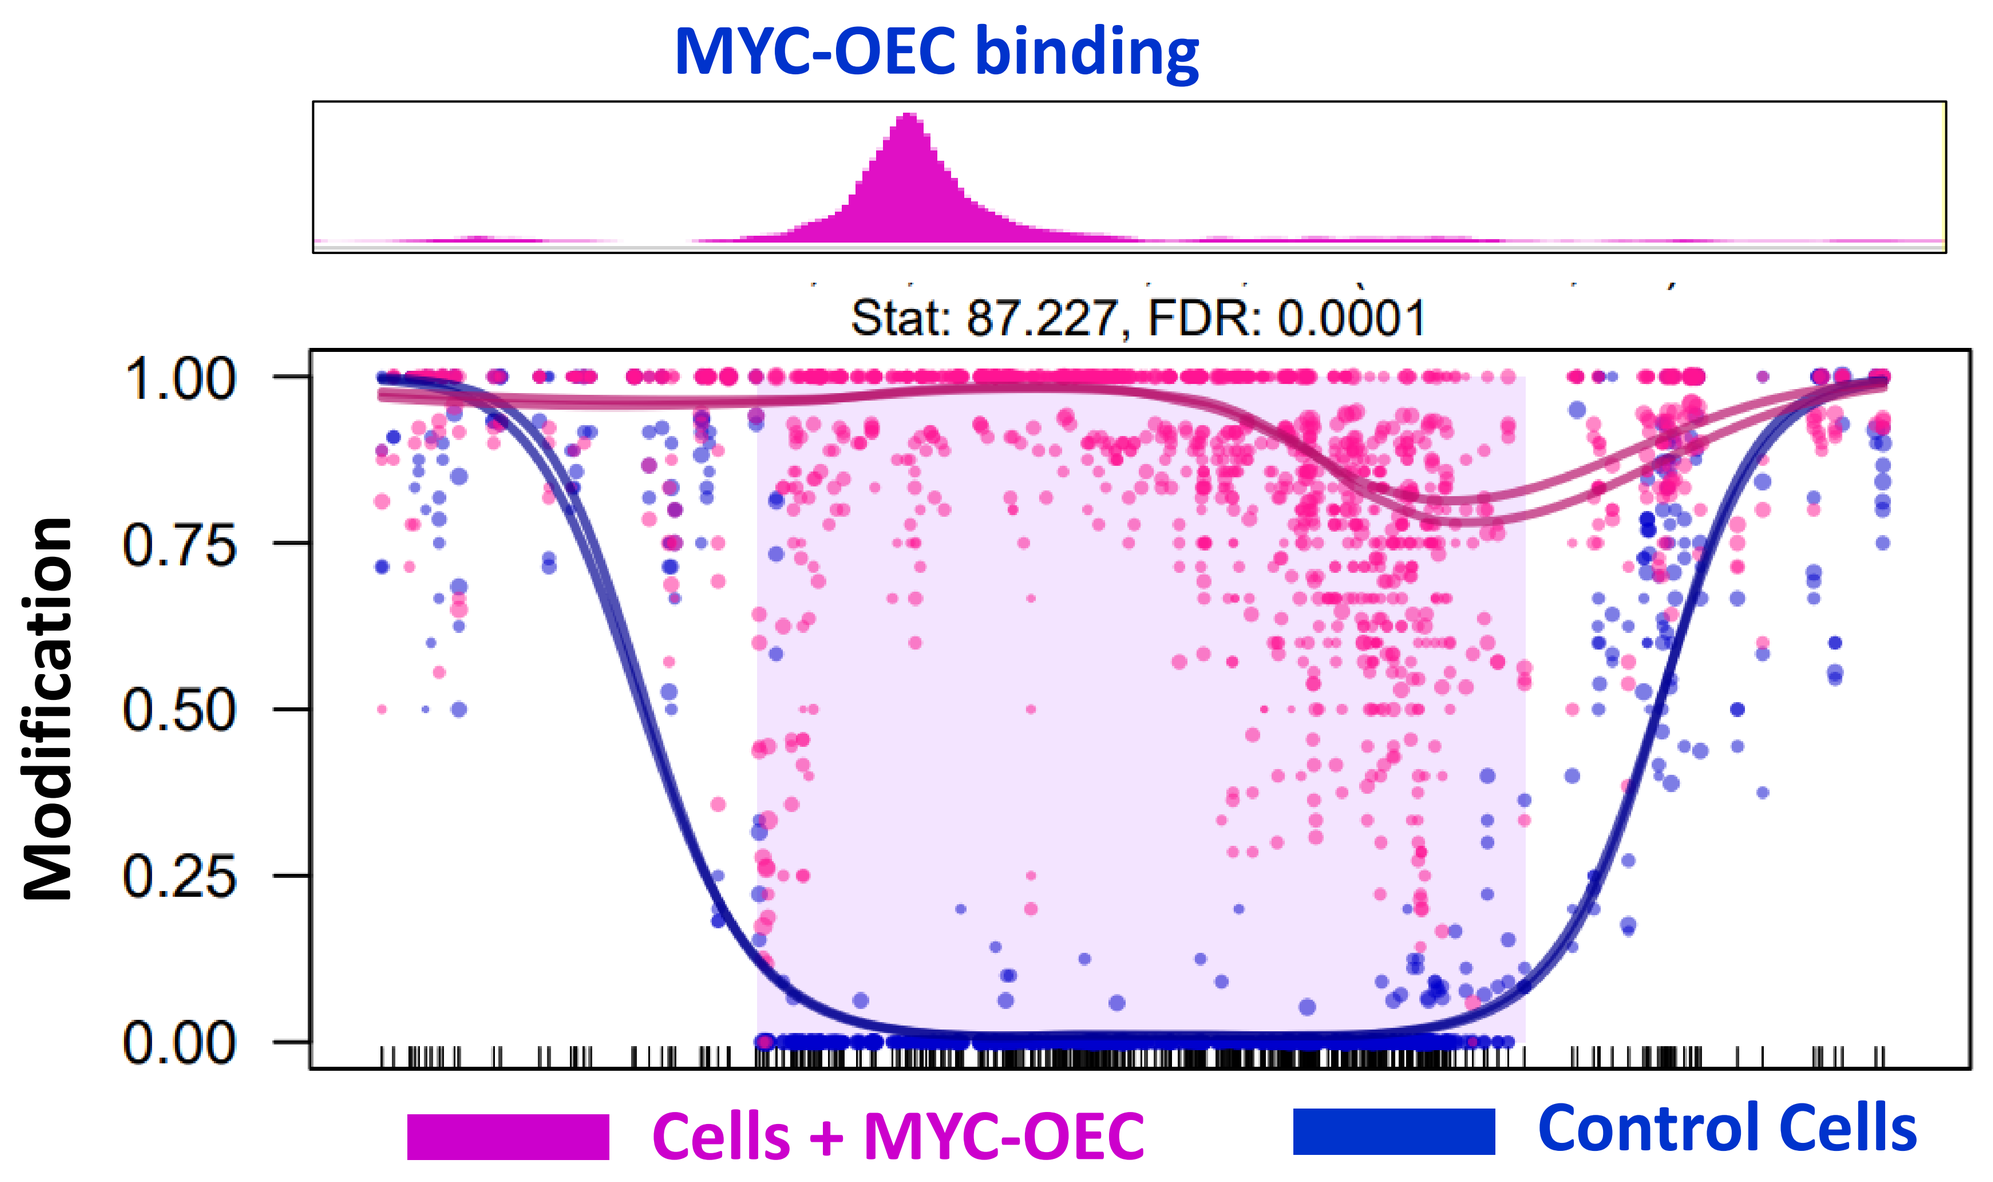
MYC-OEC binding
Stat: 87.227, FDR: 0.0001
Modification
1.00
0.75
0.50
0.25
0.00
Cells + MYC-OEC
Control Cells

Figure 2: ChIP-seq shows the binding of NSCLC MYC-OEC module to its predicted genomic region and an increase in the epigenetic marks at the target site after treatment.

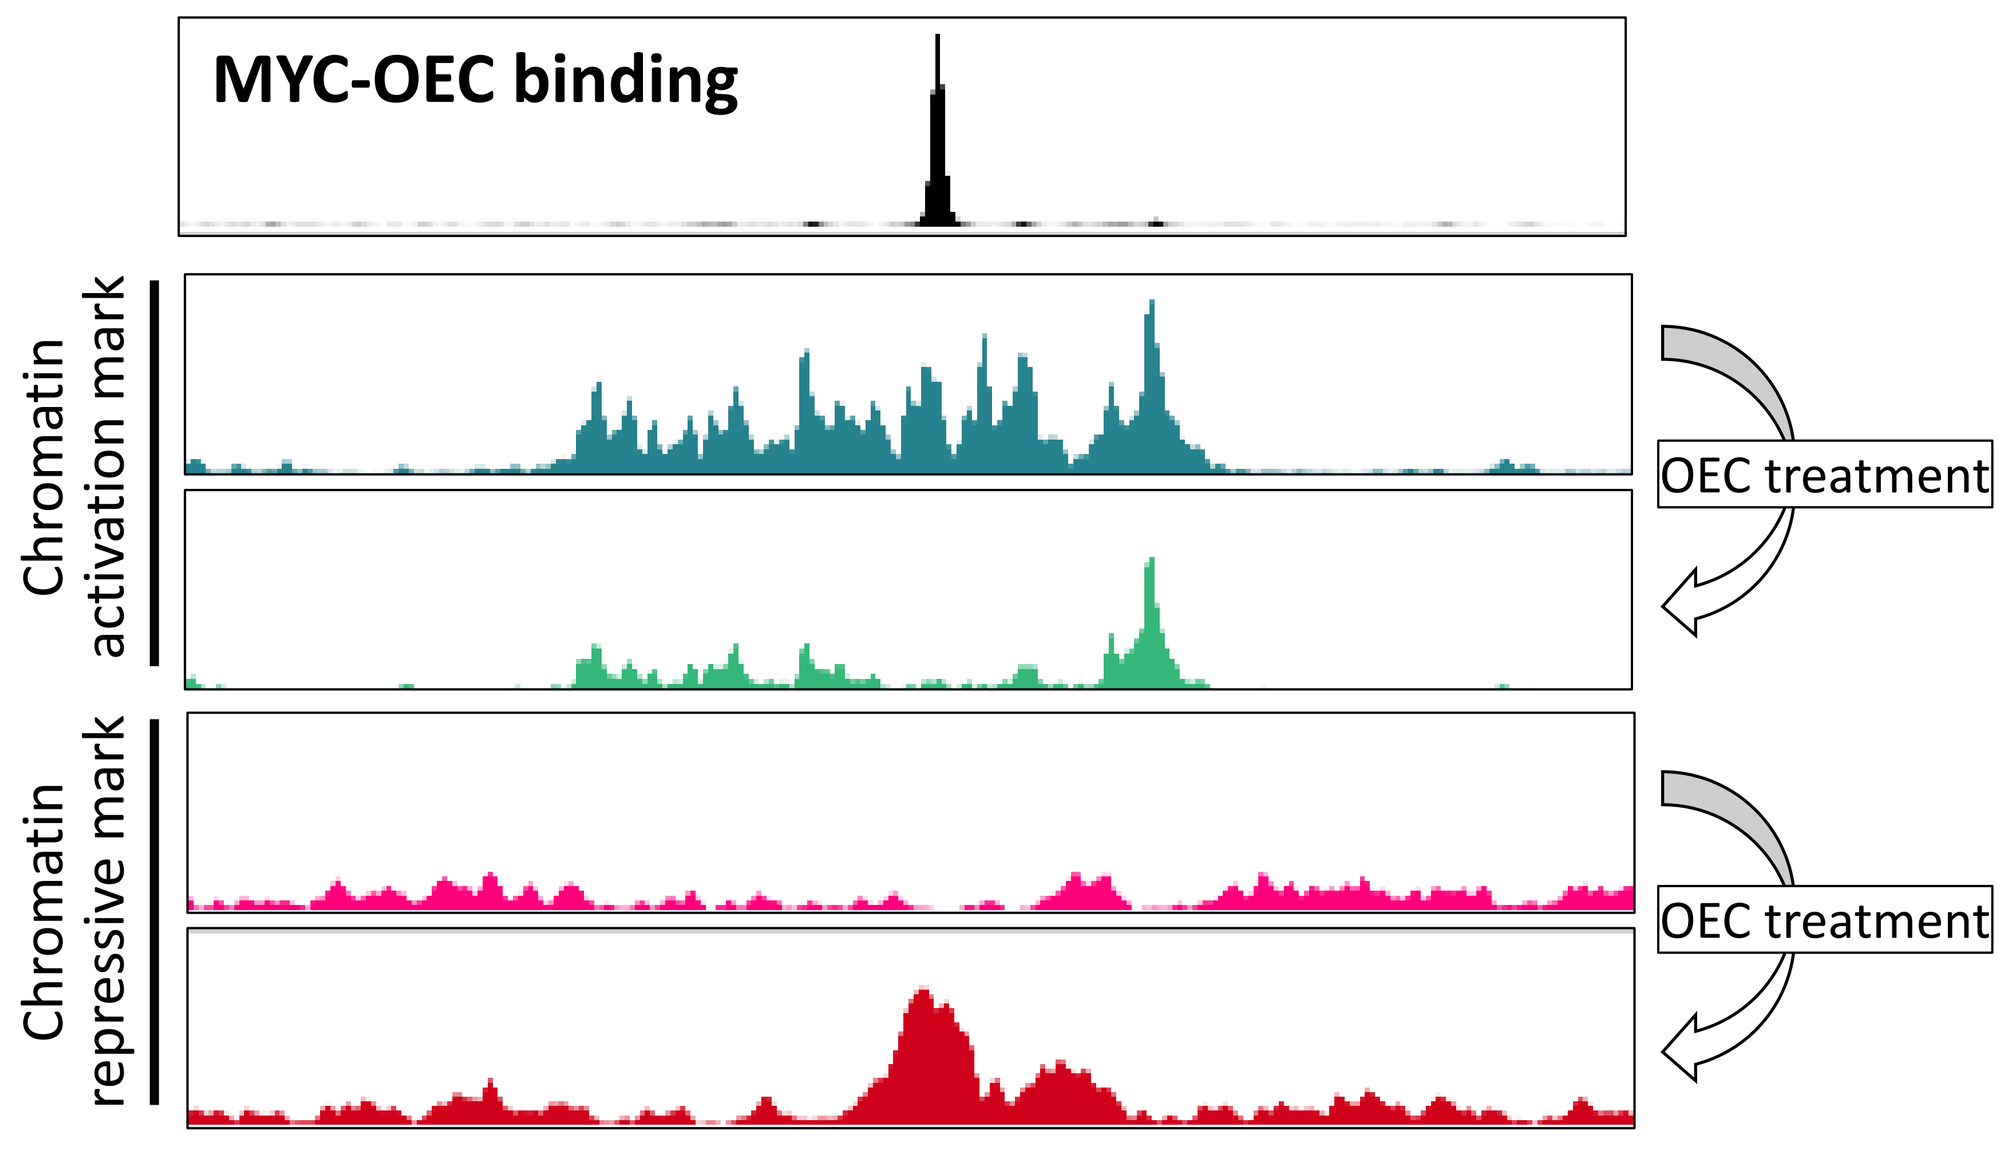
MYC-OEC binding
Chromatin activation mark
Chromatin repressive mark
OEC treatment
OEC treatment

Figure 3: ChIP-seq shows the binding of NSCLC MYC-OEC module to its predicted genomic region and the corresponding changes in epigenetic marks after treatment.

Figure 4. NSCLC MYC-OEC disrupts chromatin-looping of target sites within the MYC IGD

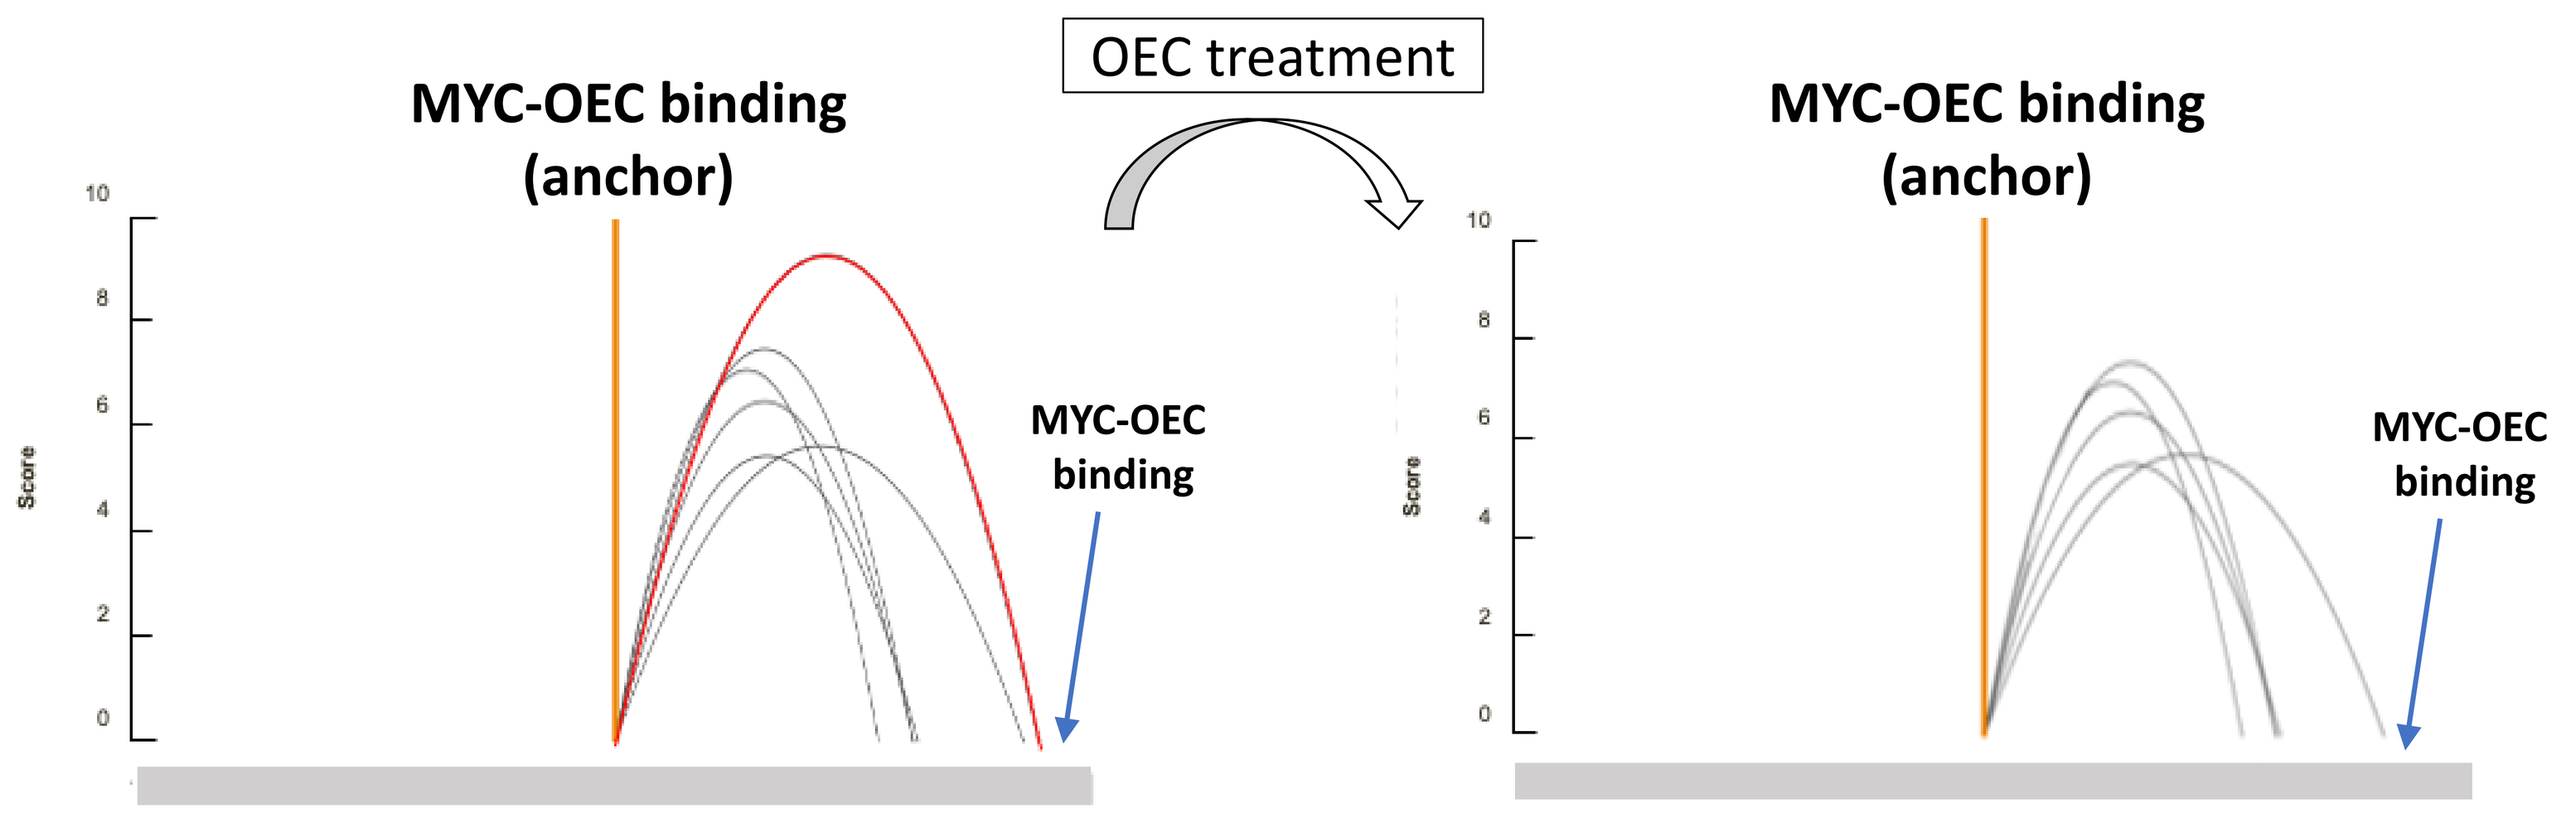
OEC treatment
MYC-OEC binding (anchor)
MYC-OEC binding
Score
MYC-OEC binding (anchor)
MYC-OEC binding
Score

Figure 4: Capture Hi-C using one of the MYC-OEC binding sites as an anchor showed that MYC-OEC treatment disrupted chromatin-looping formation between the two MYC-OEC target sites (red loop).

Figure 5. Durable MYC reduction and epigenetic modifications were observed over 13 days

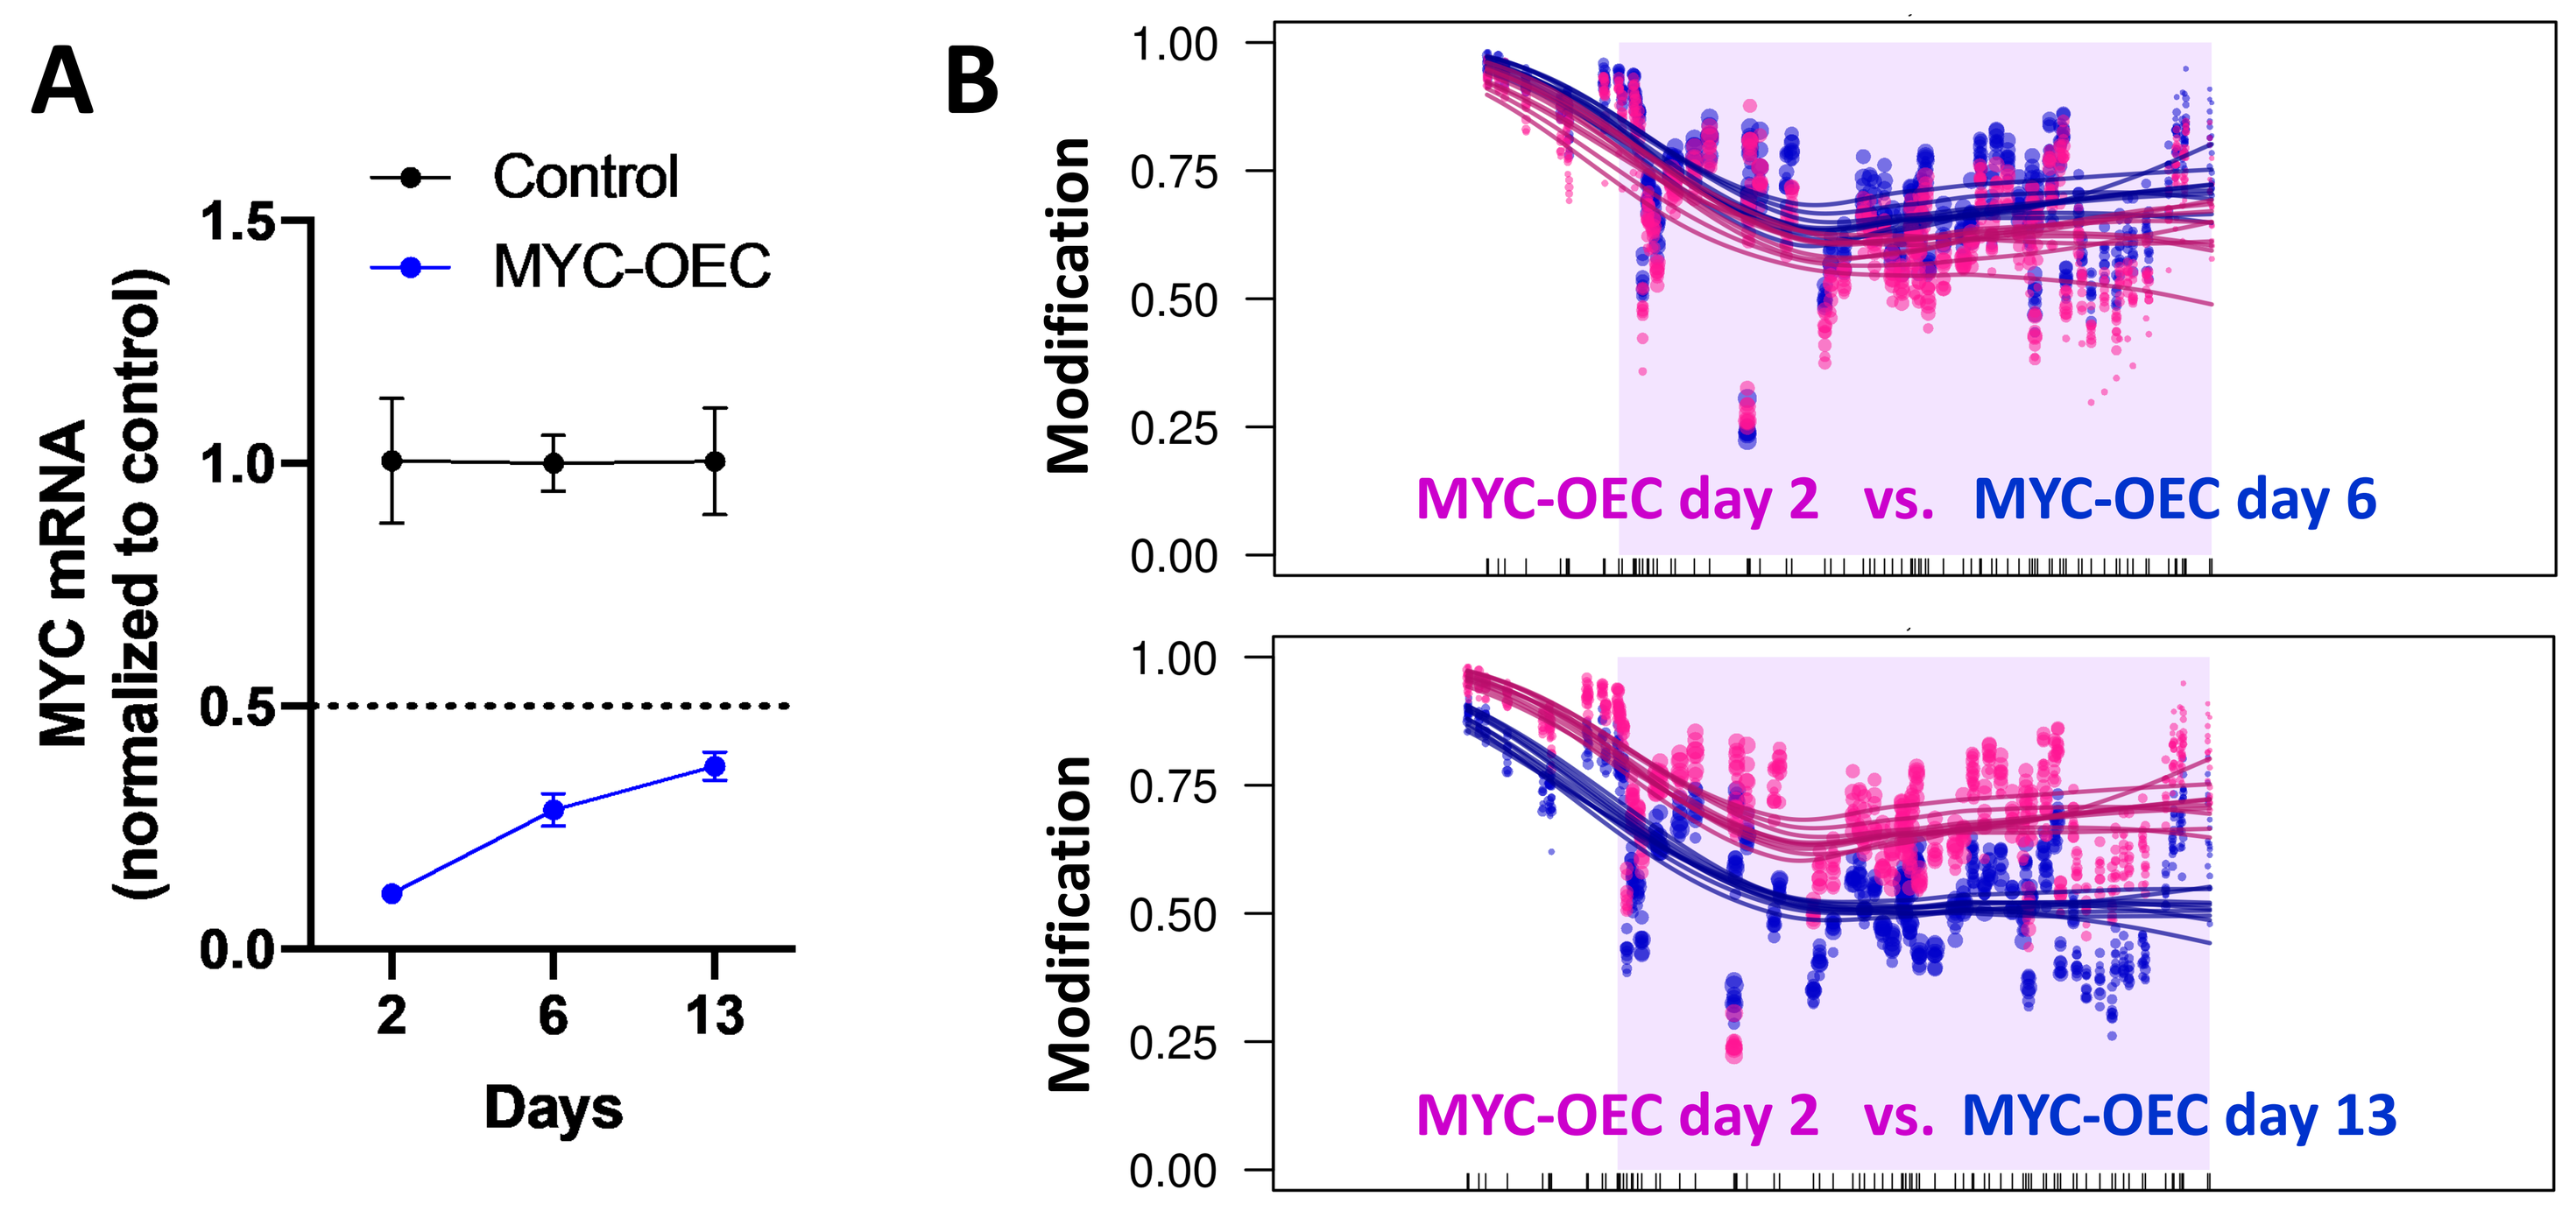
A
Control
MYC-OEC
MYC mRNA (normalized to control)
1.5
1.0
0.5
0.0
2
6
13
Days
B
Modification
MYC-OEC day 2 vs. MYC-OEC day 6
MYC-OEC day 2 vs. MYC-OEC day 13

Figure 5: Durable effects of NSCLC MYC-OEC on MYC mRNA reduction (A) and targeted epigenetic changes (B) after a single treatment on day 0. Sub-optimal concentration of MYC-OEC (0.625 ug/ml) were used to avoid gross cell death. (A) MYC-OEC treated H358 cells show durable MYC mRNA downregulation compared to control treated cells over 13 days. (B) Increased epigenetic marks observed after 2 days of MYC-OEC treatment were maintained at the same level at day 6 (top) with slightly reduction at day 13 (bottom) presumably due to the proliferation of untransfected cells.

Figure 6. NSCLC MYC-OEC treatment reduces MYC mRNA levels and viability across multiple NSCLC cell lines

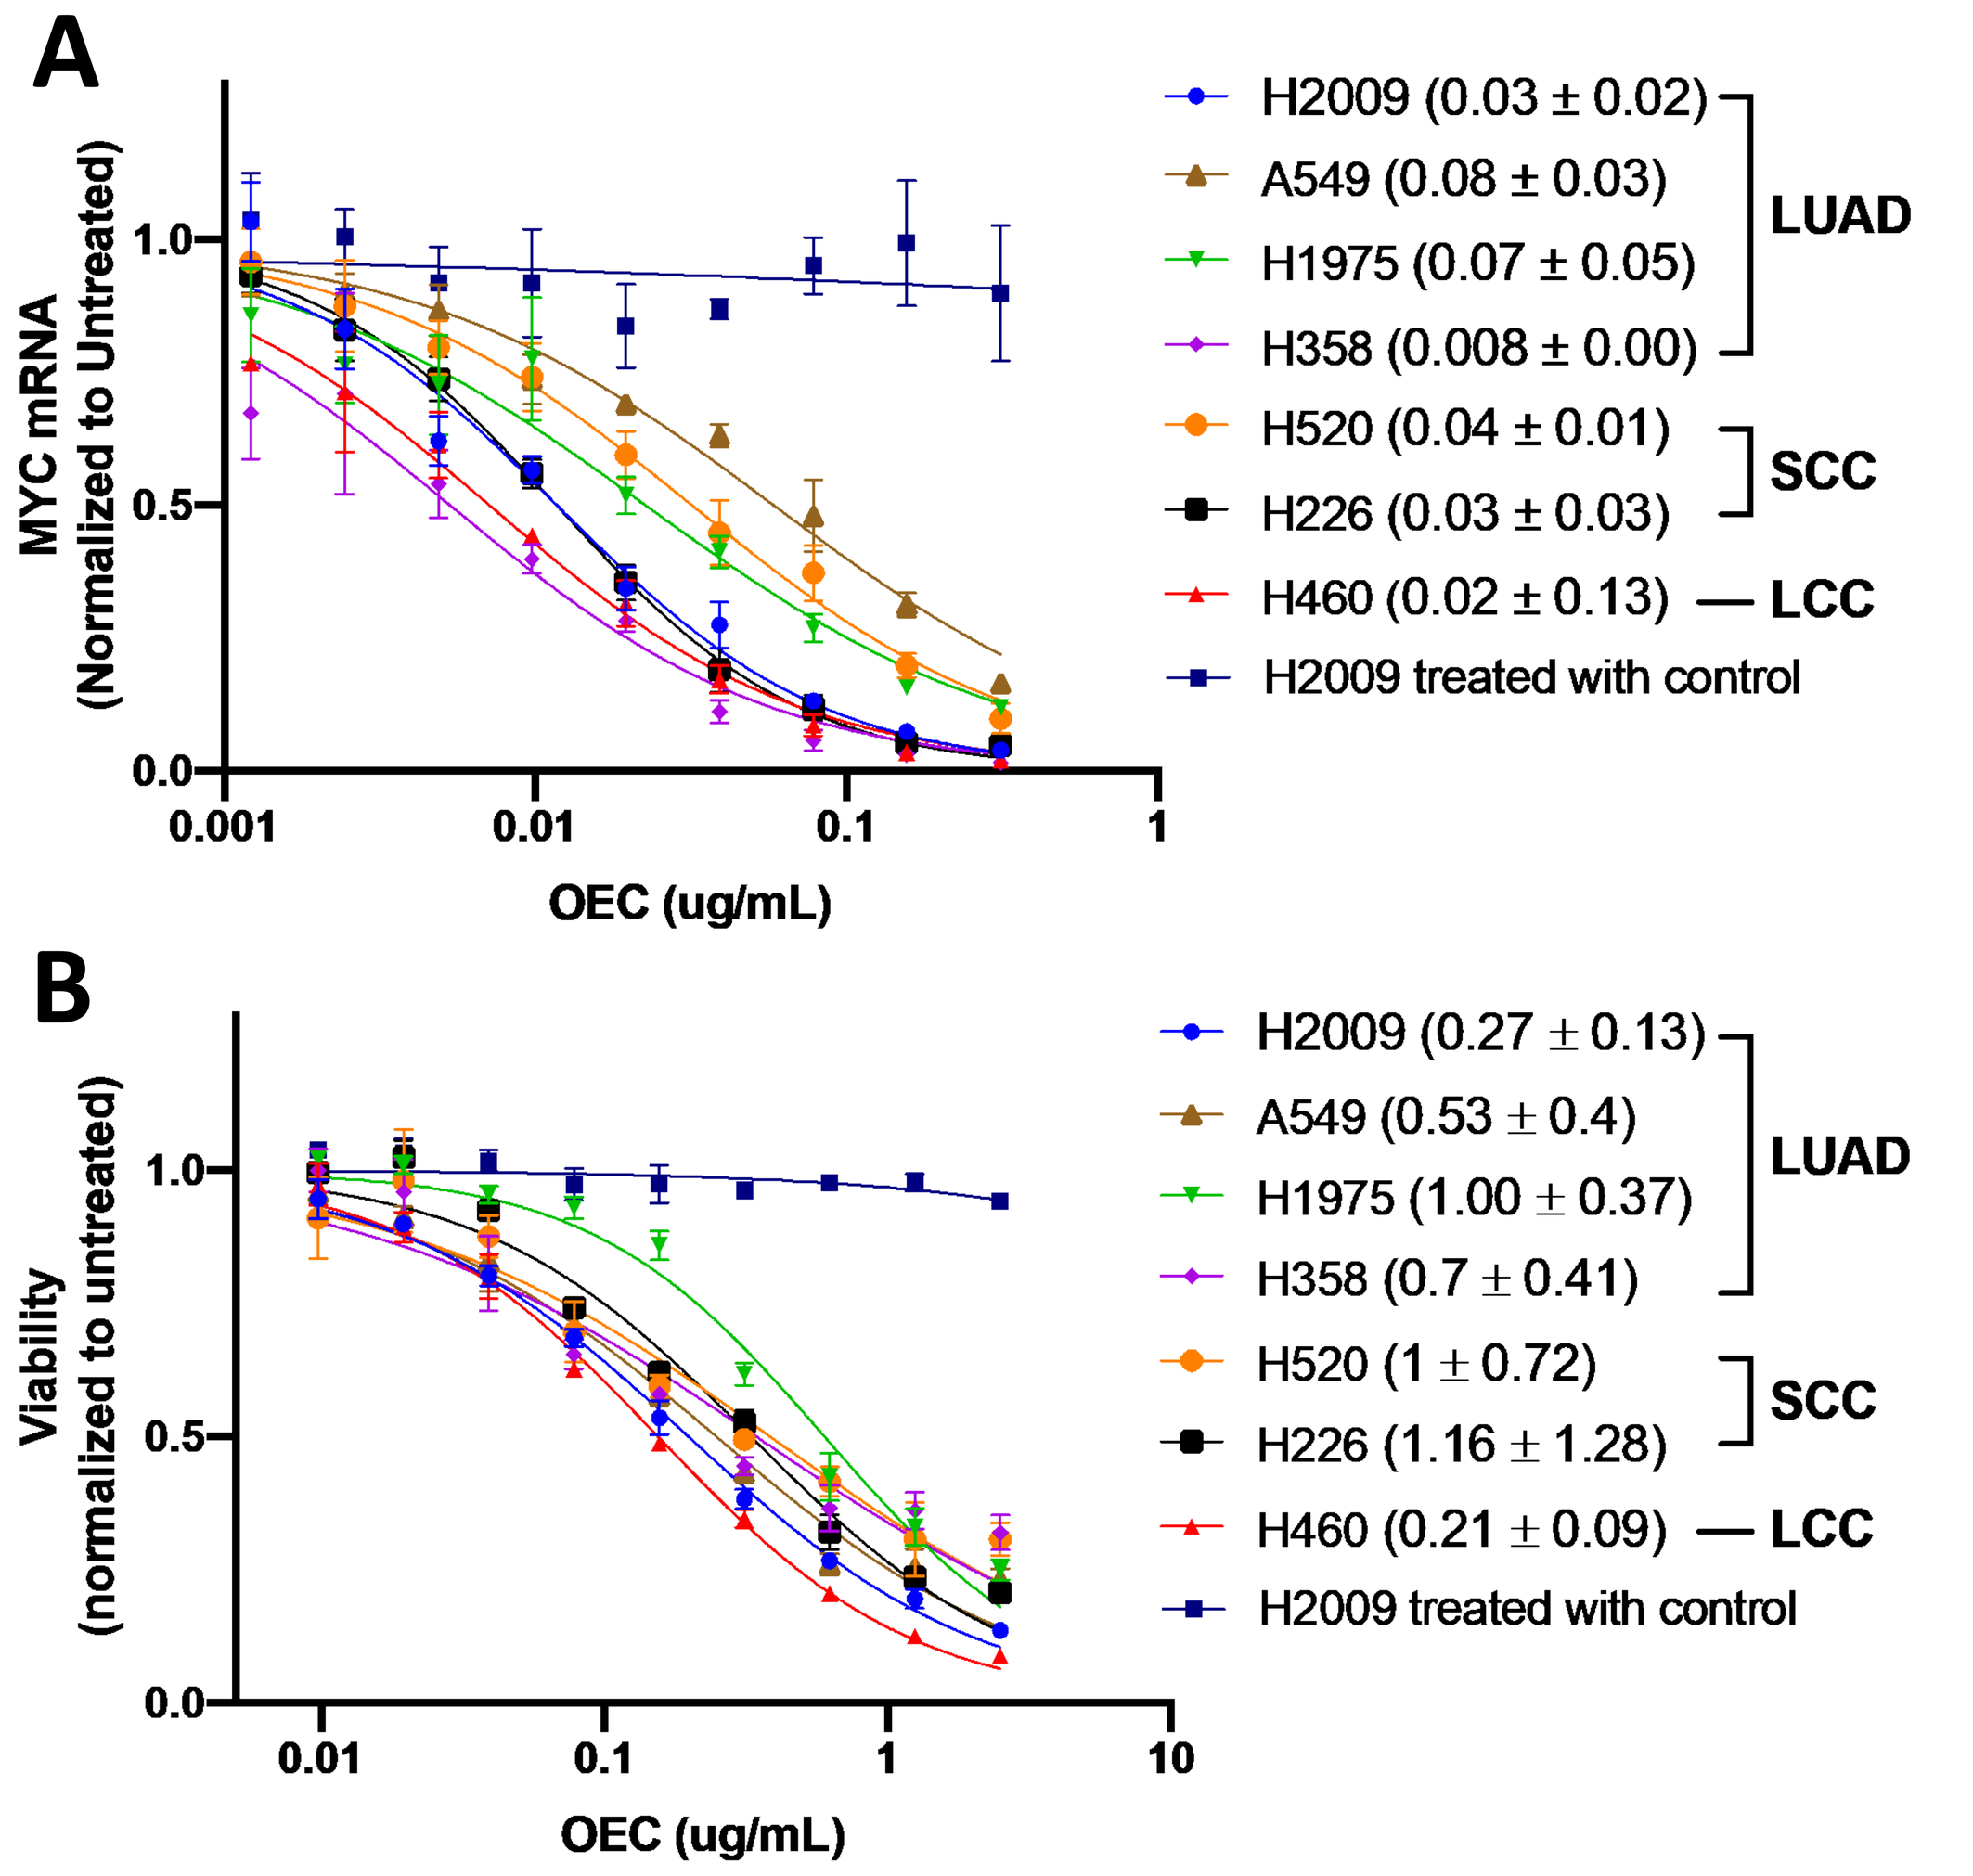
A
MYC mRNA (Normalized to Untreated)
OEC (ug/mL)
H2009 (0.03 ± 0.02)
A549 (0.08 ± 0.03)
H1975 (0.07 ± 0.05)
H358 (0.008 ± 0.00)
LUAD
H520 (0.04 ± 0.01)
H226 (0.03 ± 0.03)
SCC
H460 (0.02 ± 0.13) — LCC
H2009 treated with control
B
Viability (normalized to untreated)
OEC (ug/mL)
H2009 (0.27 ± 0.13)
A549 (0.53 ± 0.4)
H1975 (1.00 ± 0.37)
H358 (0.7 ± 0.41)
LUAD
H520 (1 ± 0.72)
H226 (1.16 ± 1.28)
SCC
H460 (0.21 ± 0.09) — LCC
H2009 treated with control

Figure 6: NSCLC MYC-OEC reduces MYC mRNA levels (A, 48 hrs. post treatment) and viability (B, 72 hrs. post treatment) in a panel of NSCLC cell lines. Representative dose-response curves shown. Average ± standard deviation of IC50 (ug/ml) shown in legend. LUAD: lung adenocarcinoma, SCC: lung squamous cell carcinoma, LCC: lung large cell carcinoma.

Figure 7 & 8. NSCLC MYC-OEC inhibits NSCLC patient-derived organoid growth and MYC mRNA levels while inducing epigenetic modifications

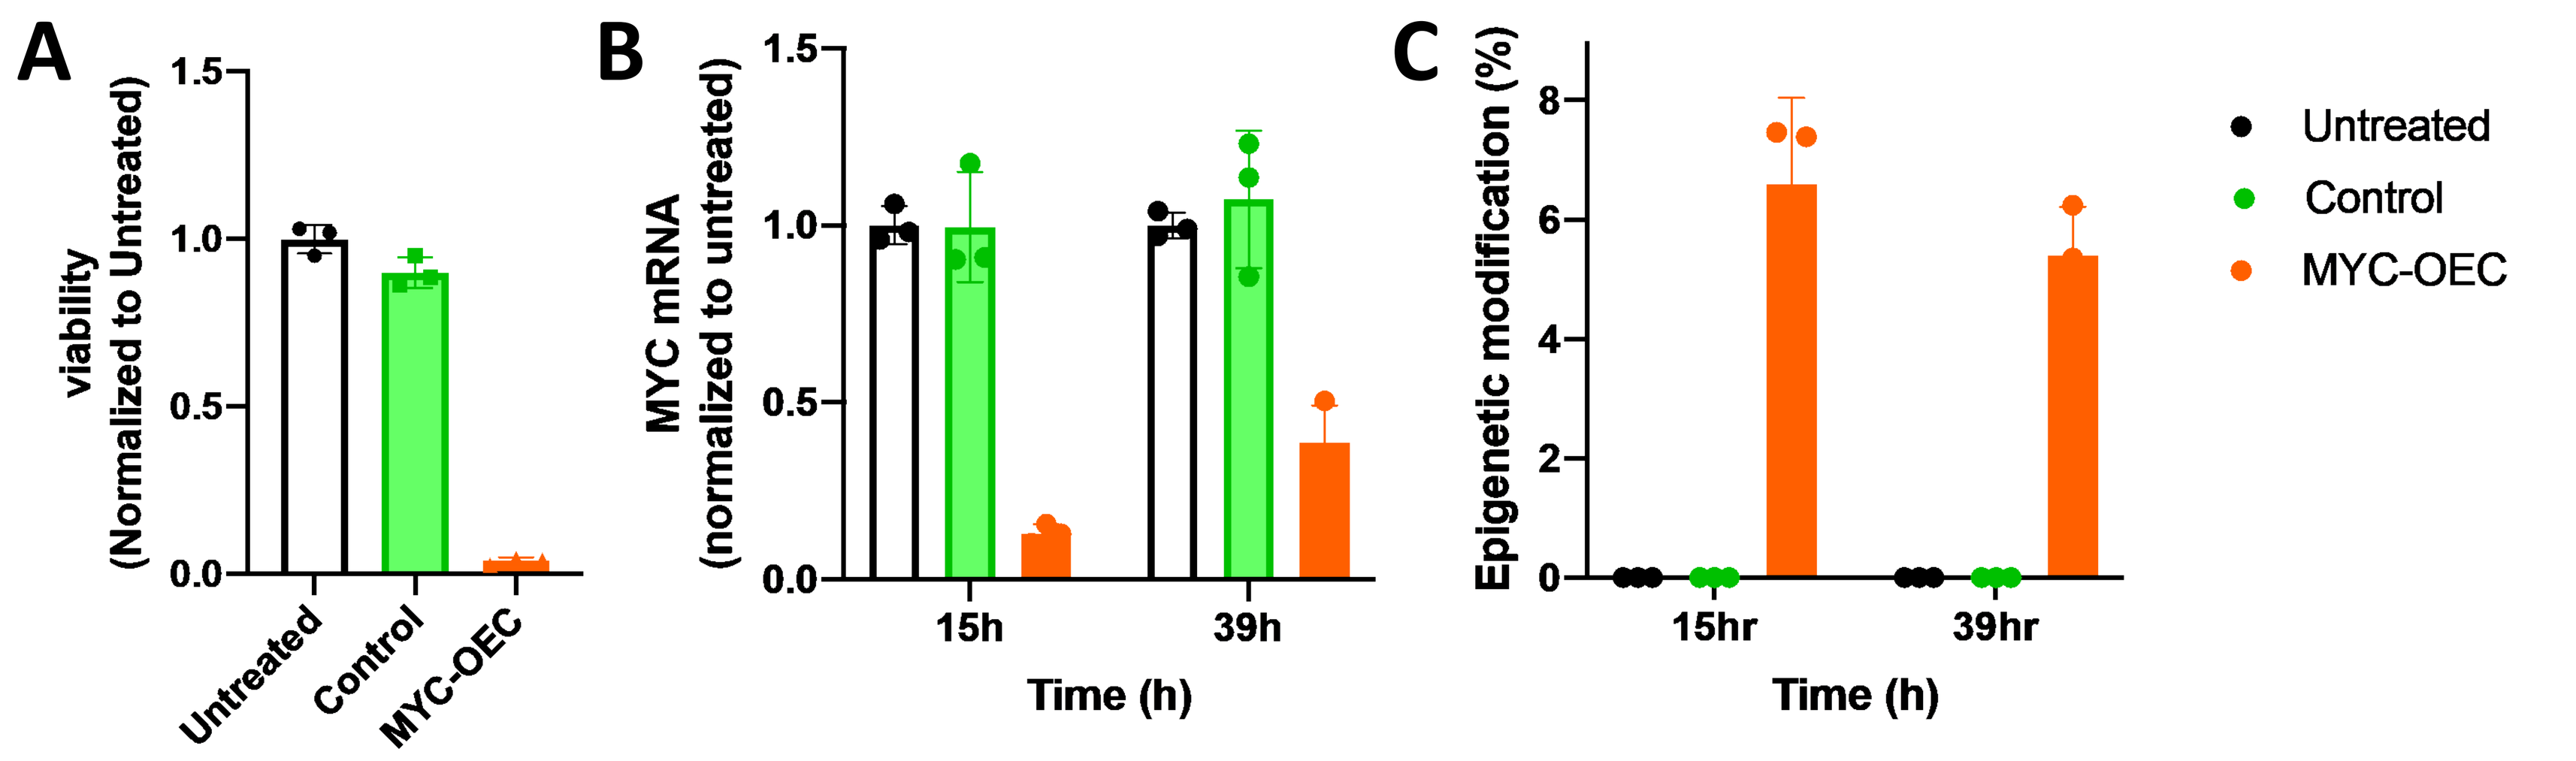
A
viability (Normalized to Untreated)
Untreated
Control
MYC-OEC
B
MYC mRNA (normalized to untreated)
15h
39h
Time (h)
C
Epigenetic modification (%)
15hr
39hr
Time (h)
Untreated
Control
MYC-OEC

Figure 7: Treatment of the NSCLC patient-derived organoid with MYC-OEC resulted in reduction in cell viability (A, >90% compared to control at day 7), and decreased MYC mRNA expression (B) as well as a corresponding increase in epigenetic modification of the target locus (C).

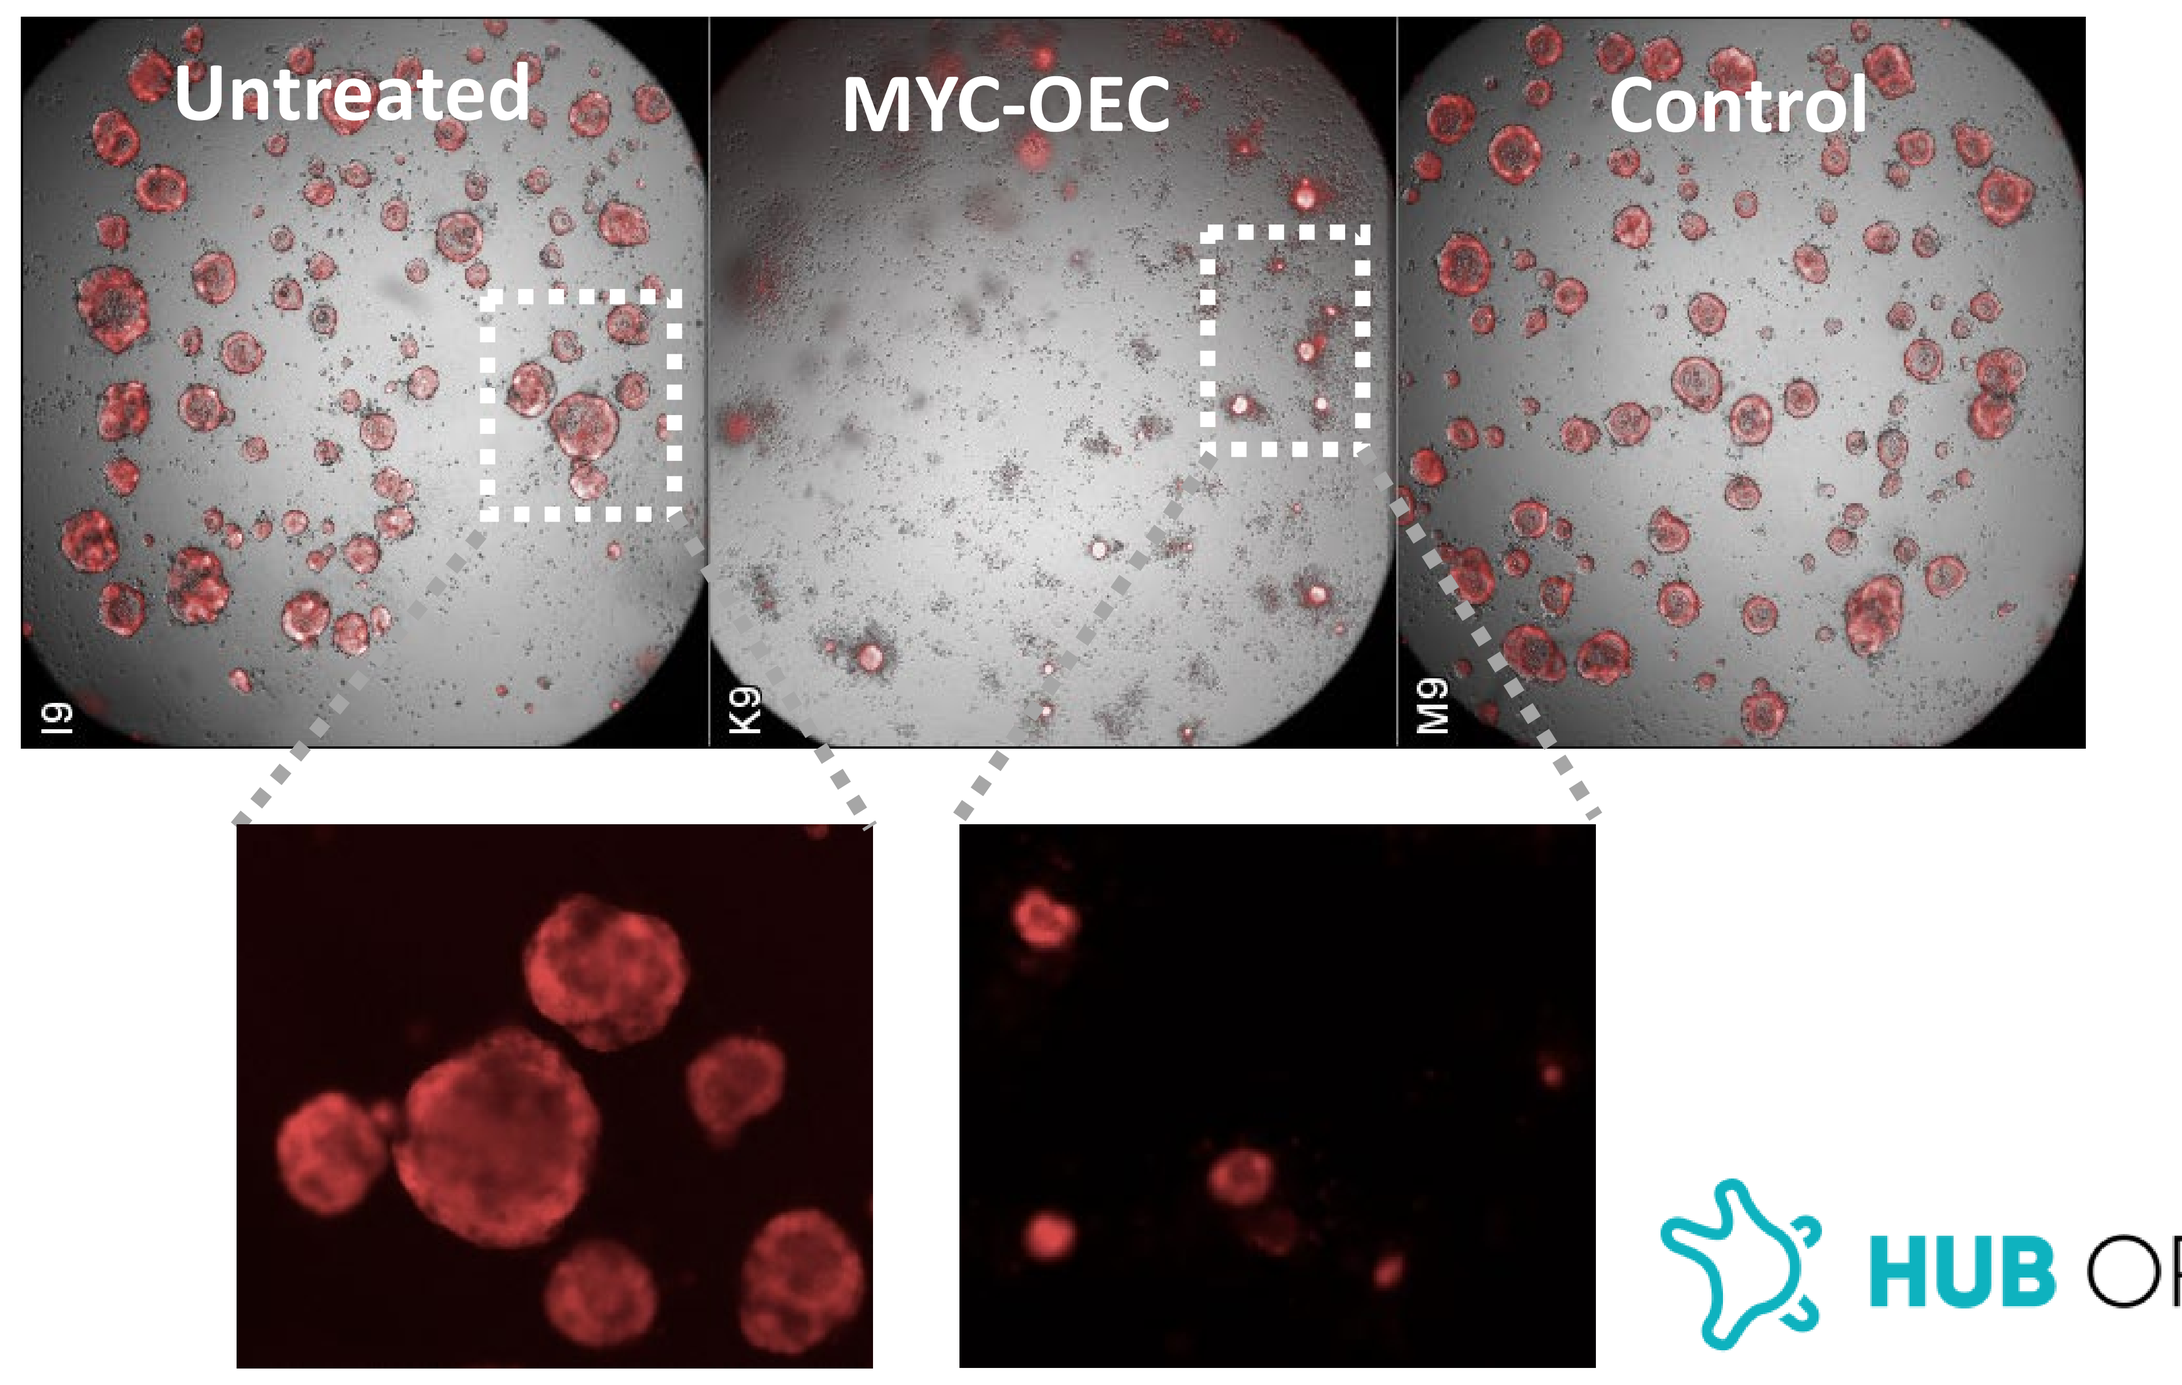
Untreated
MYC-OEC
Control

Figure 8: Representative bright-field and live cell fluorescence images of NSCLC patient-derived organoids treated with PBS (untreated), MYC-OEC, or control mRNA (Control) for 7 days.

HUB ORGANOIDS

Figure 9. Treatment of NSCLC tumor-bearing mice with NSCLC MYC-OEC significantly inhibits overall tumor growth while inducing epigenetic modifications in vivo

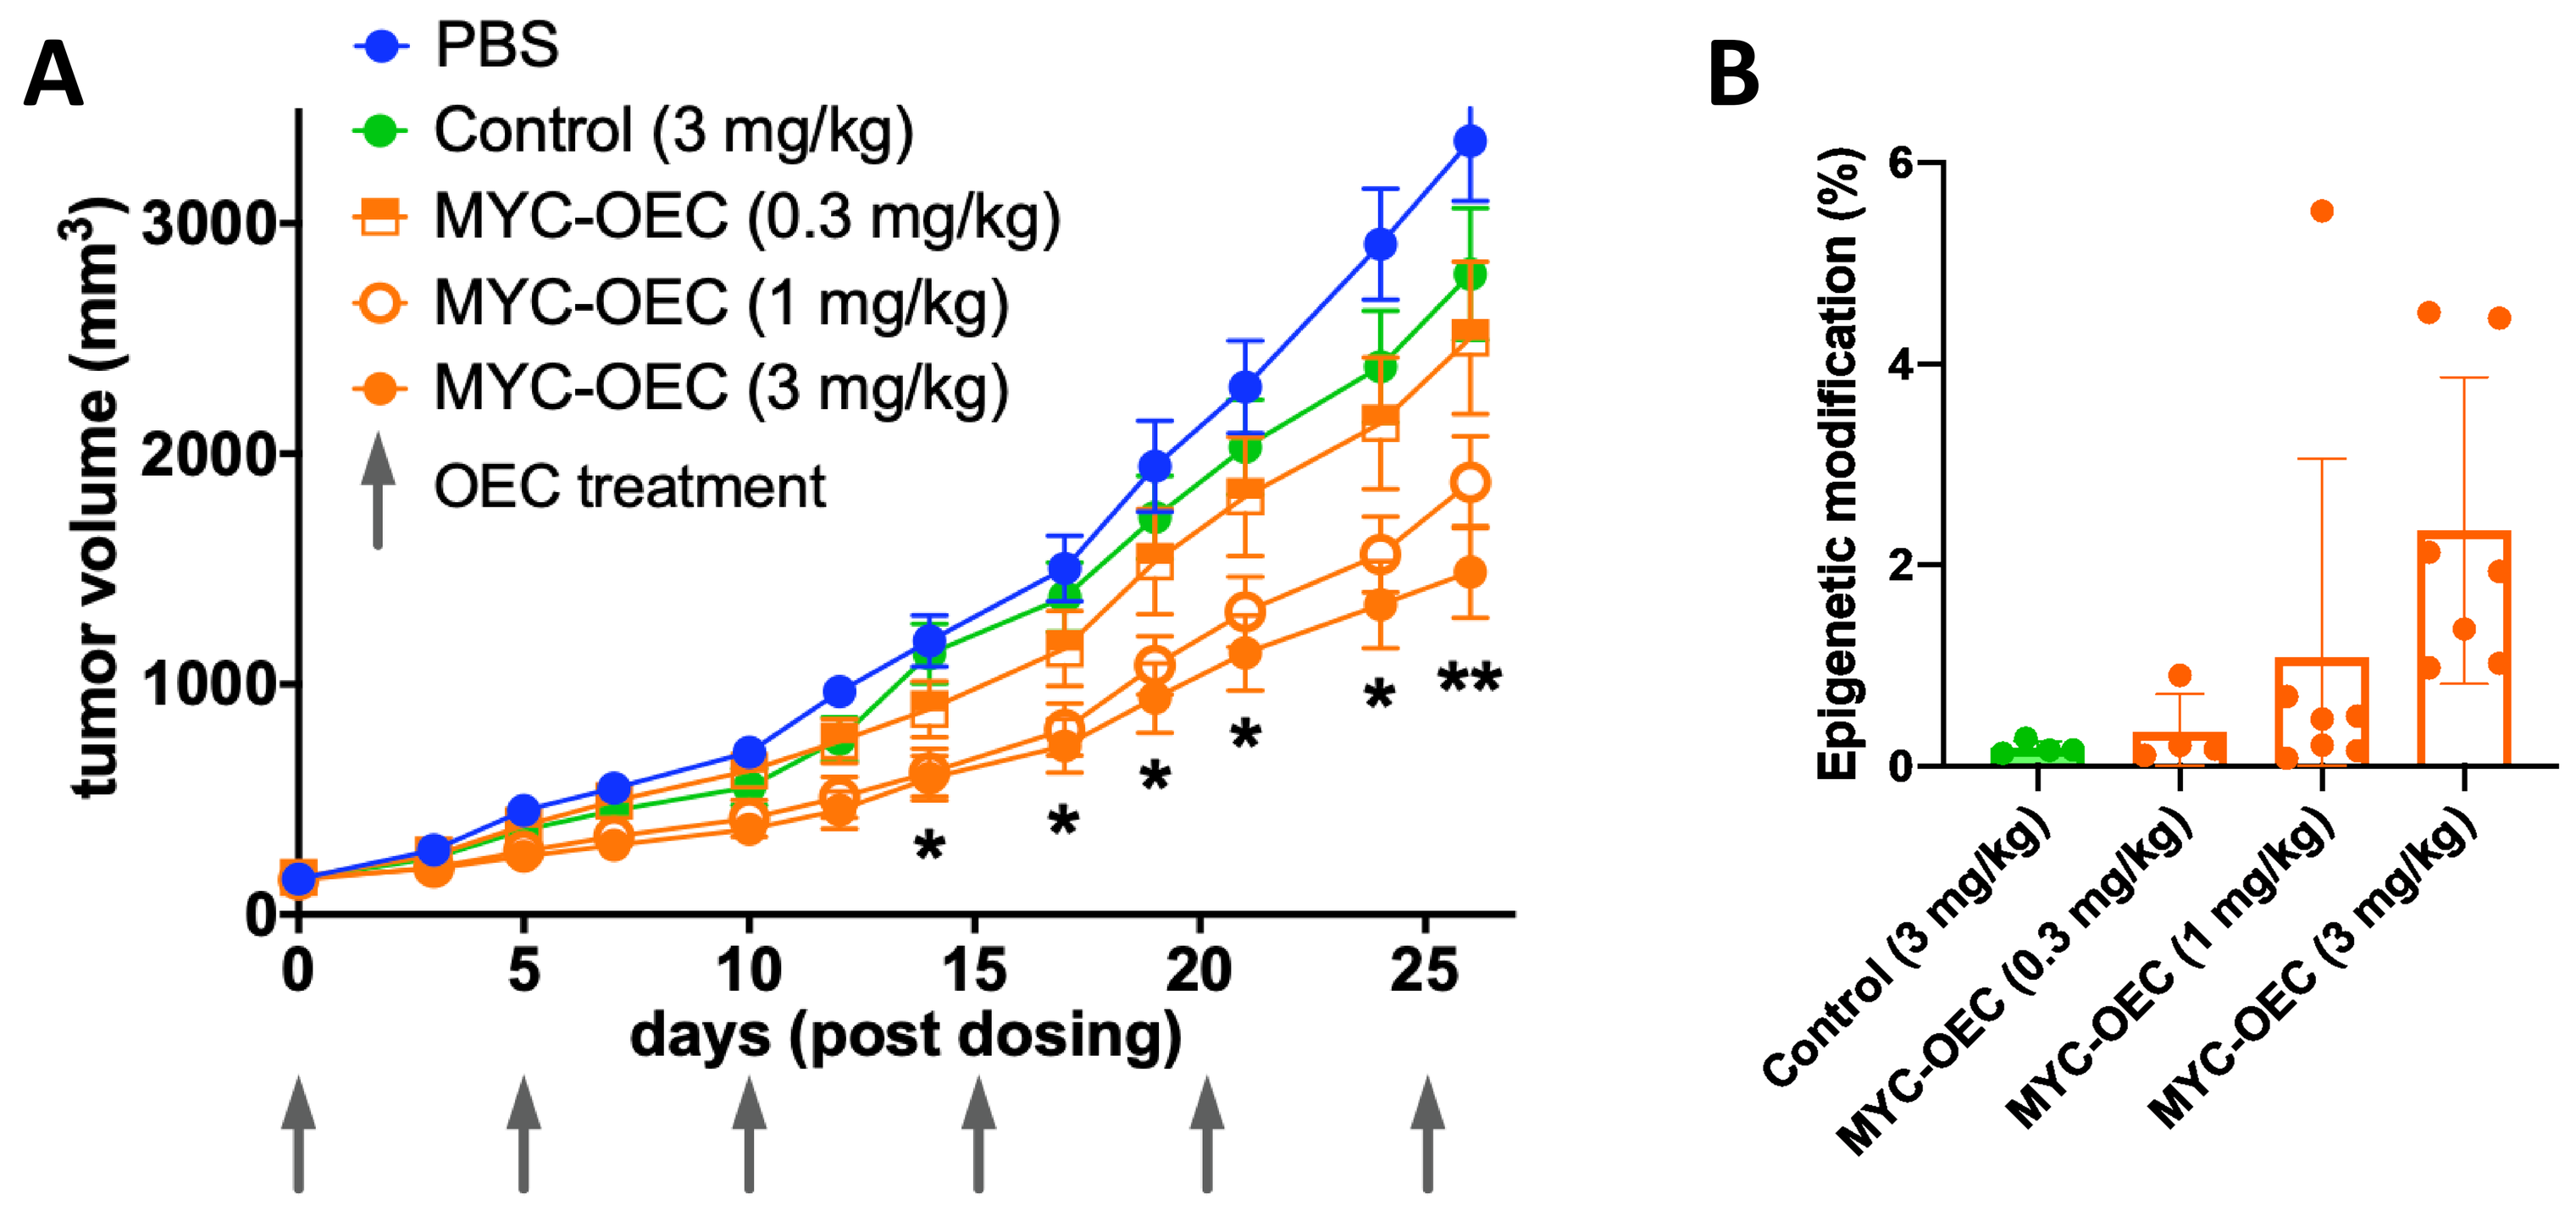
A
PBS
Control (3 mg/kg)
MYC-OEC (0.3 mg/kg)
MYC-OEC (1 mg/kg)
MYC-OEC (3 mg/kg)
OEC treatment
tumor volume (mm³)
days (post dosing)
B
Epigenetic modification (%)
Control (3 mg/kg)
MYC-OEC (0.3 mg/kg)
MYC-OEC (1 mg/kg)
MYC-OEC (3 mg/kg)

Figure 9: NSCLC MYC-OEC treatment reduces tumor growth (A) while inducing epigenetic modifications (B) *in vivo.* (A) H460 subQ tumors were treated with PBS, 3 mg/kg negative control, 3, 1, or 0.3 mg/kg NSCLC MYC-OEC every 5 days. Significant difference in tumor size between 3 mg/kg control vs. 3 mg/kg MYC-OEC treated mice shown as '**' denotes p<0.01, '*' denotes p<0.05 (2-way ANOVA). (B) Increased epigenetic modifications were observed in H460 subQ tumors treated with MYC-OEC compared to control treated tumors.

Summary

In summary, our findings have demonstrated that the OEC designed by the OMEGA platform tunably and durably modulated epigenetic regulation of MYC expression, and may offer a novel therapeutic approach to treating patients with NSCLC and other MYC-associated tumor types.

© Poster Template by Genigraphics® 1.800.790.4001 www.genigraphics.com