Targeted epigenomic modulation of MYC enhances responses to immune checkpoint and EGFR inhibitors in preclinical models of NSCLC

Defne Yarar, Eugine Lee, Padraich Flahardy, Cameron Vergato, Graeme Hodgson, Thomas McCauley
Omega Therapeutics, Cambridge, MA, USA

2023 AACR-NCI-EORTC conference
Poster Session C, C104

Background

Aberrant expression of the transcription factor MYC is found in most cancers including non-small cell lung cancer (NSCLC). Despite MYC's therapeutic attractiveness, direct targeting of MYC activity has remained elusive, largely due to its high degree of downstream autoregulation and the lack of an enzymatic pocket required for effective drug binding. We have developed a NSCLC-specific programmable epigenomic mRNA therapy, termed a MYC epigenomic controller (NSCLC MYC-EC) that is designed to target the insulated genomic domain (IGD) of MYC, a discrete chromatin-looping region within which gene expression is mediated by CTCF (CCTC binding factor). We have previously shown that NSCLC MYC-EC effectively downregulates MYC expression pre-transcriptionally and have demonstrated that NSCLC MYC-EC inhibits NSCLC patient-derived organoid viability in vitro and abrogates xenograft tumor growth in vivo.

Here, we use in silico, in vitro, and in vivo methods to demonstrate preclinical proof-of-concept for the development of NSCLC MYC-EC in combination with immune checkpoint inhibitors or EGFR inhibitors in NSCLC patients.

Figure 1. Structure and mechanism of action of ECs

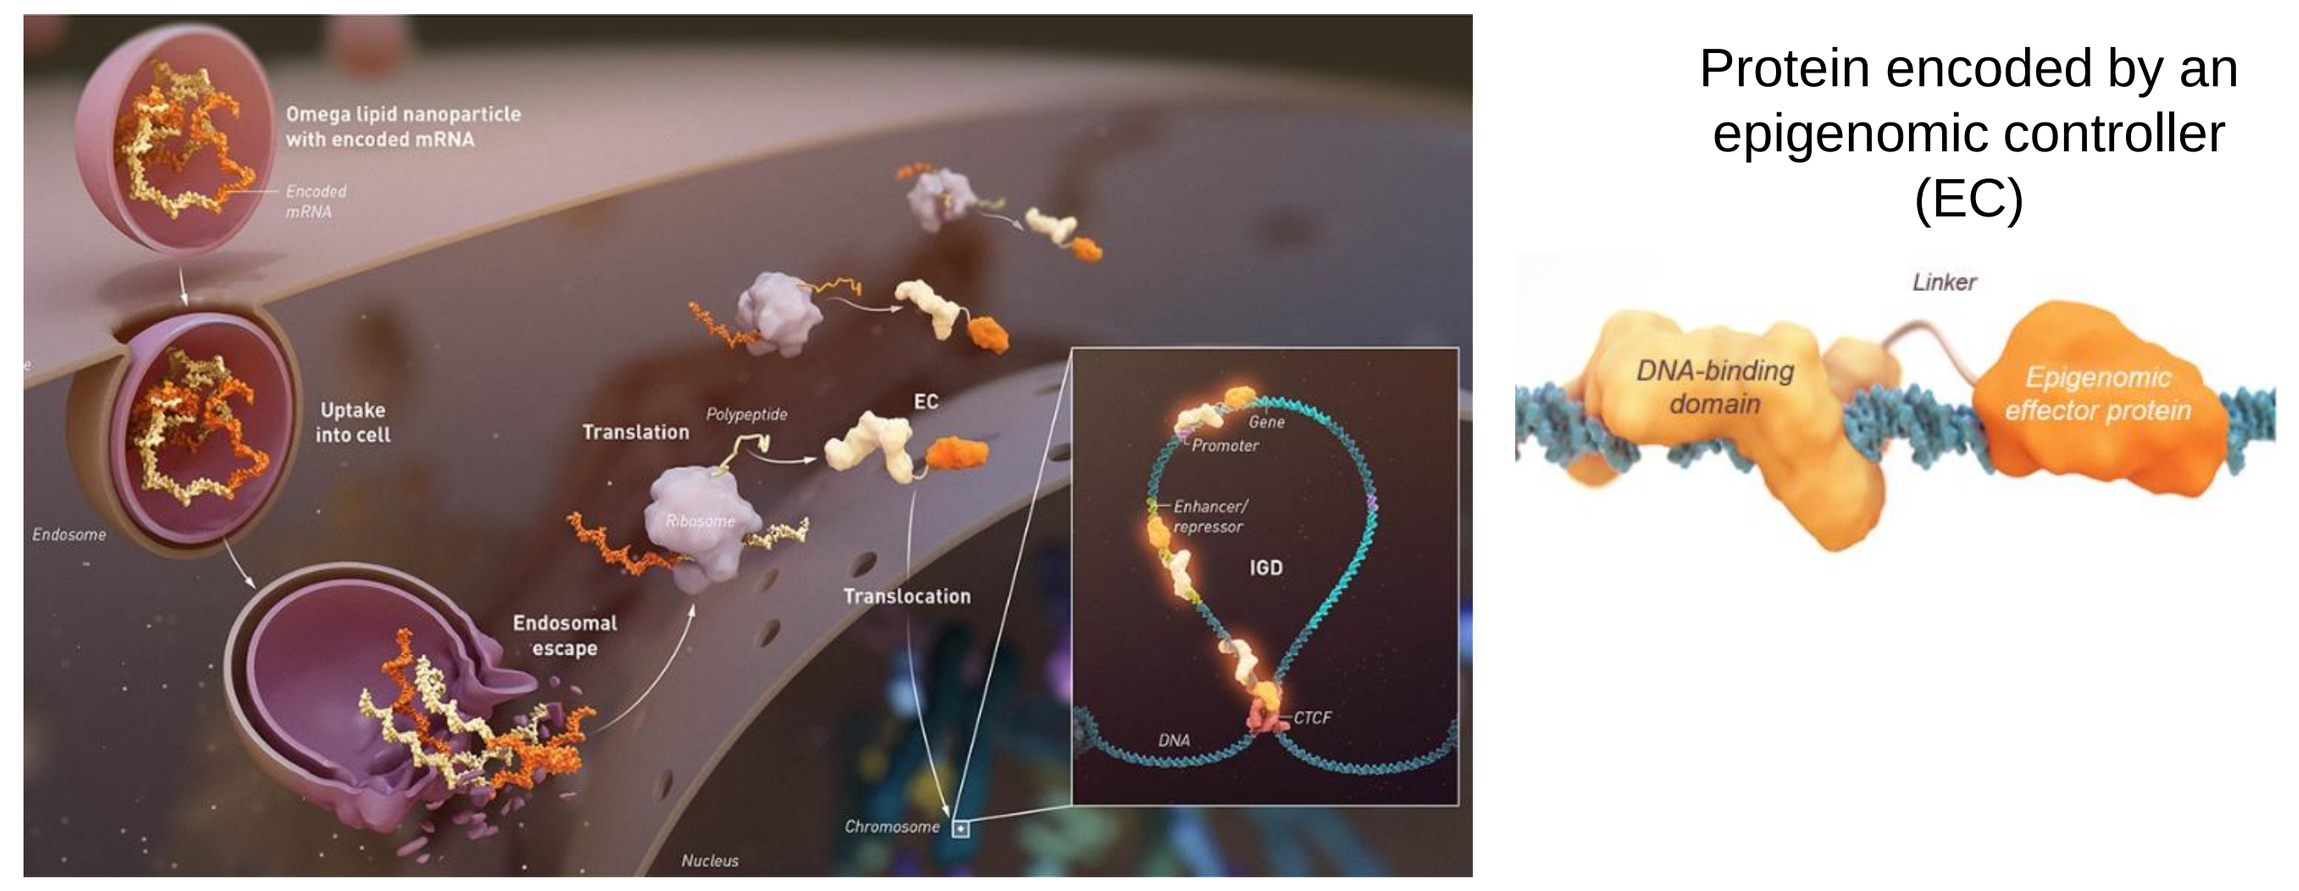
Omega lipid nanoparticle with encoded mRNA
Uptake into cell
Translation
EC
Endosomal escape
Translocation
IGD
Protein encoded by an epigenomic controller (EC)
Linker
DNA-binding domain
Epigenomic effector protein

NSCLC MYC-EC, a bicistronic construct comprised of EC1 and EC2, binds and modifies two sites within the MYC IGD

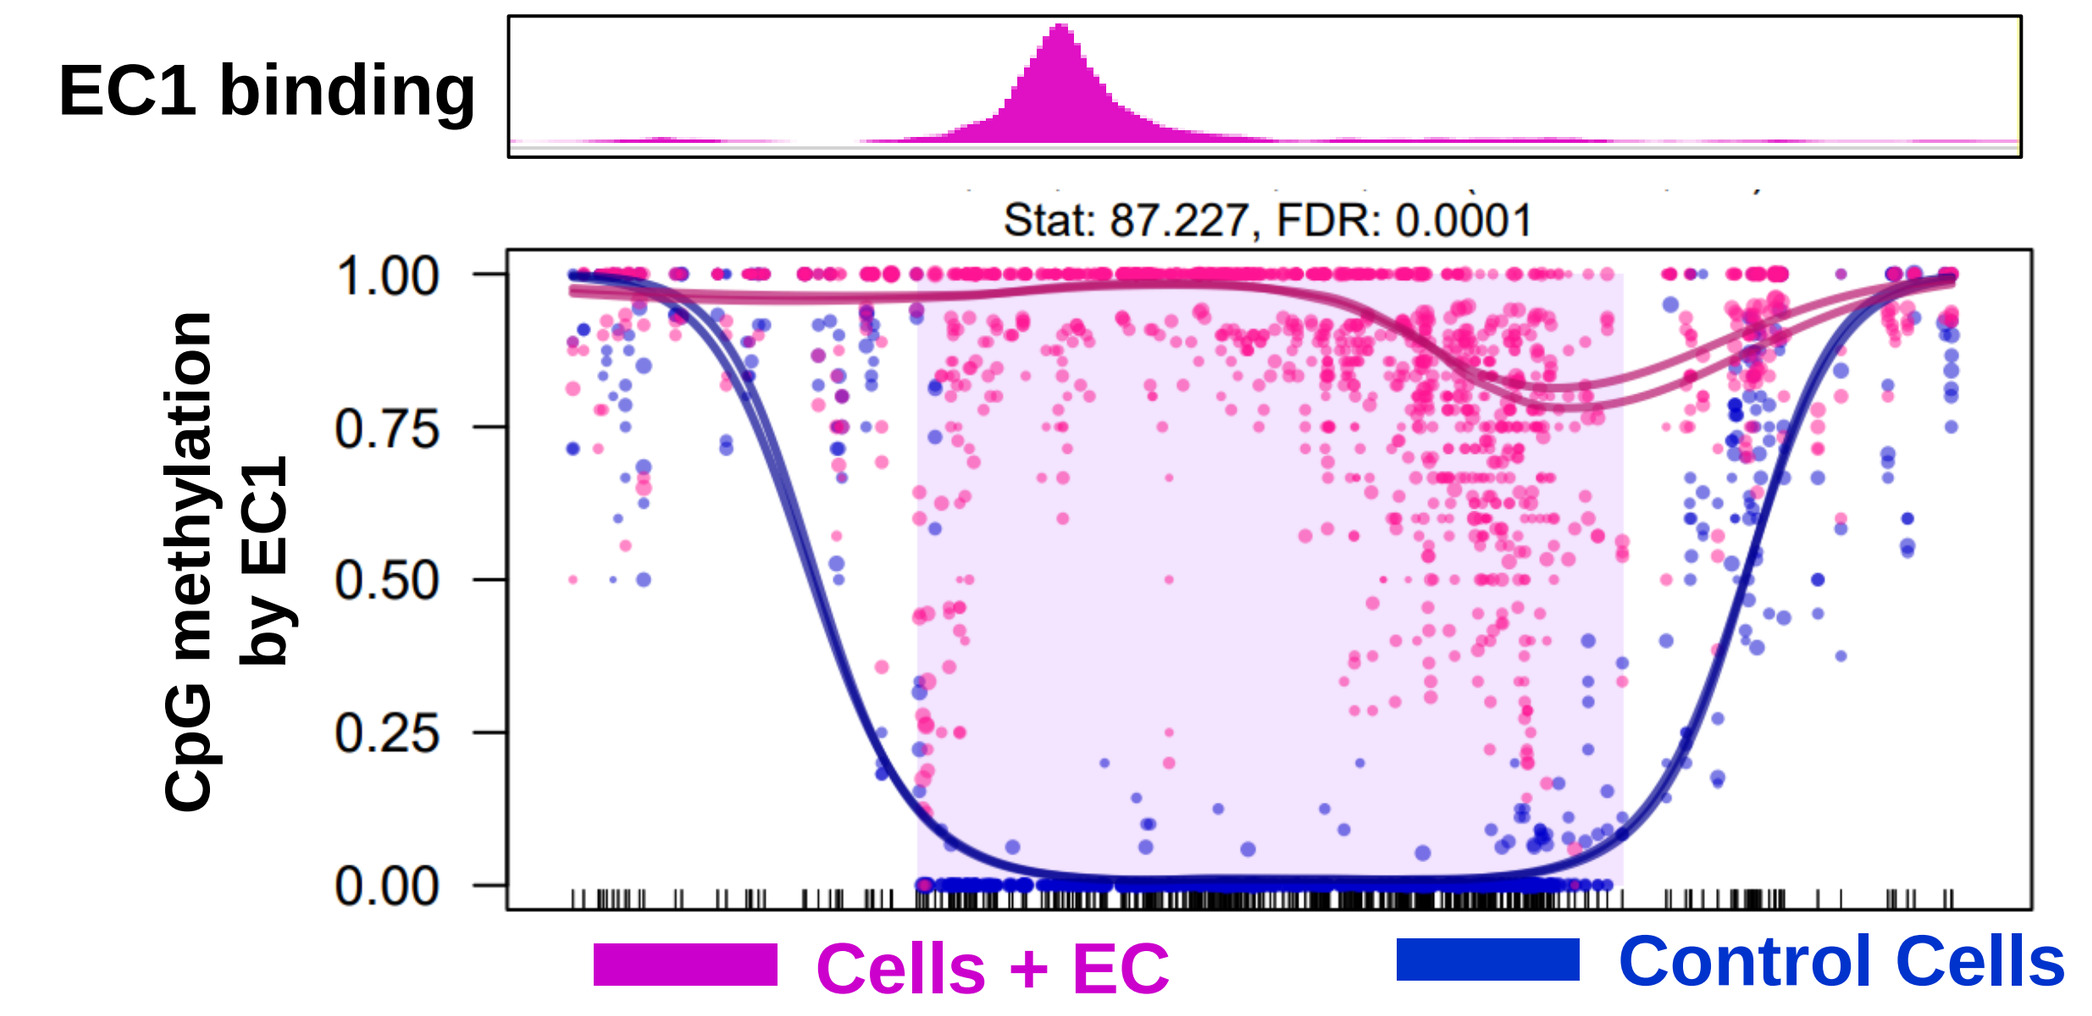
EC1 binding
Stat: 87.227, FDR: 0.0001
CpG methylation by EC1
1.00
0.75
0.50
0.25
0.00
Cells + EC
Control Cells

Figure 2. ChIP-seq shows binding of protein encoded by the NSCLC MYC-EC1 to its targeted genomic region and an increase in CpG methylation at the target site.

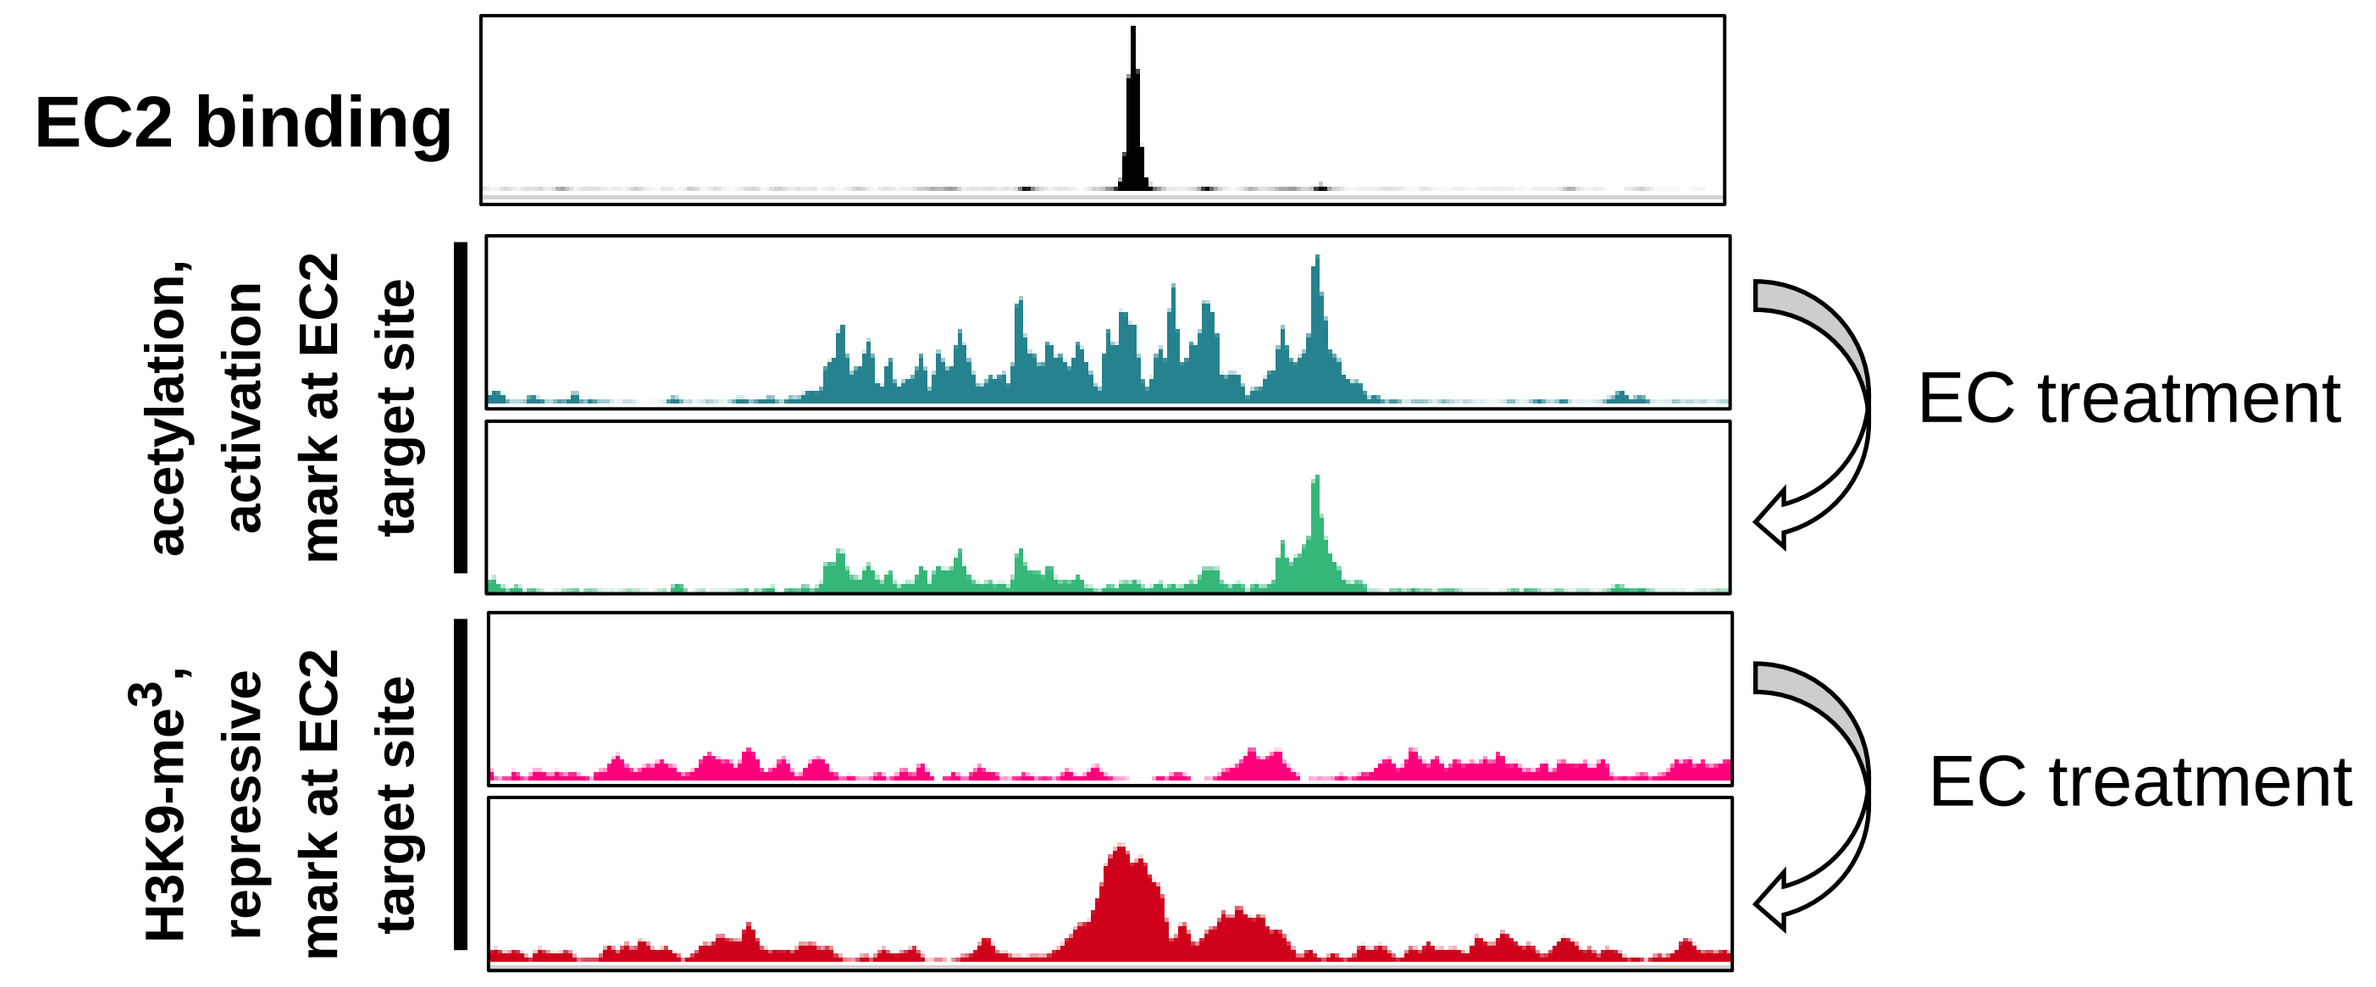
EC2 binding
acetylation, activation mark at EC2 target site
EC treatment
H3K9-me3, repressive mark at EC2 target site
EC treatment

Figure 3. ChIP-seq shows binding of the protein encoded by the NSCLC MYC-EC2 to its targeted genomic region and the corresponding changes in histone acetylation (activation mark) and methylation (repression mark).

High *MYC* mRNA levels trend towards a shorter time to progression in pembrolizumab-treated NSCLC patients

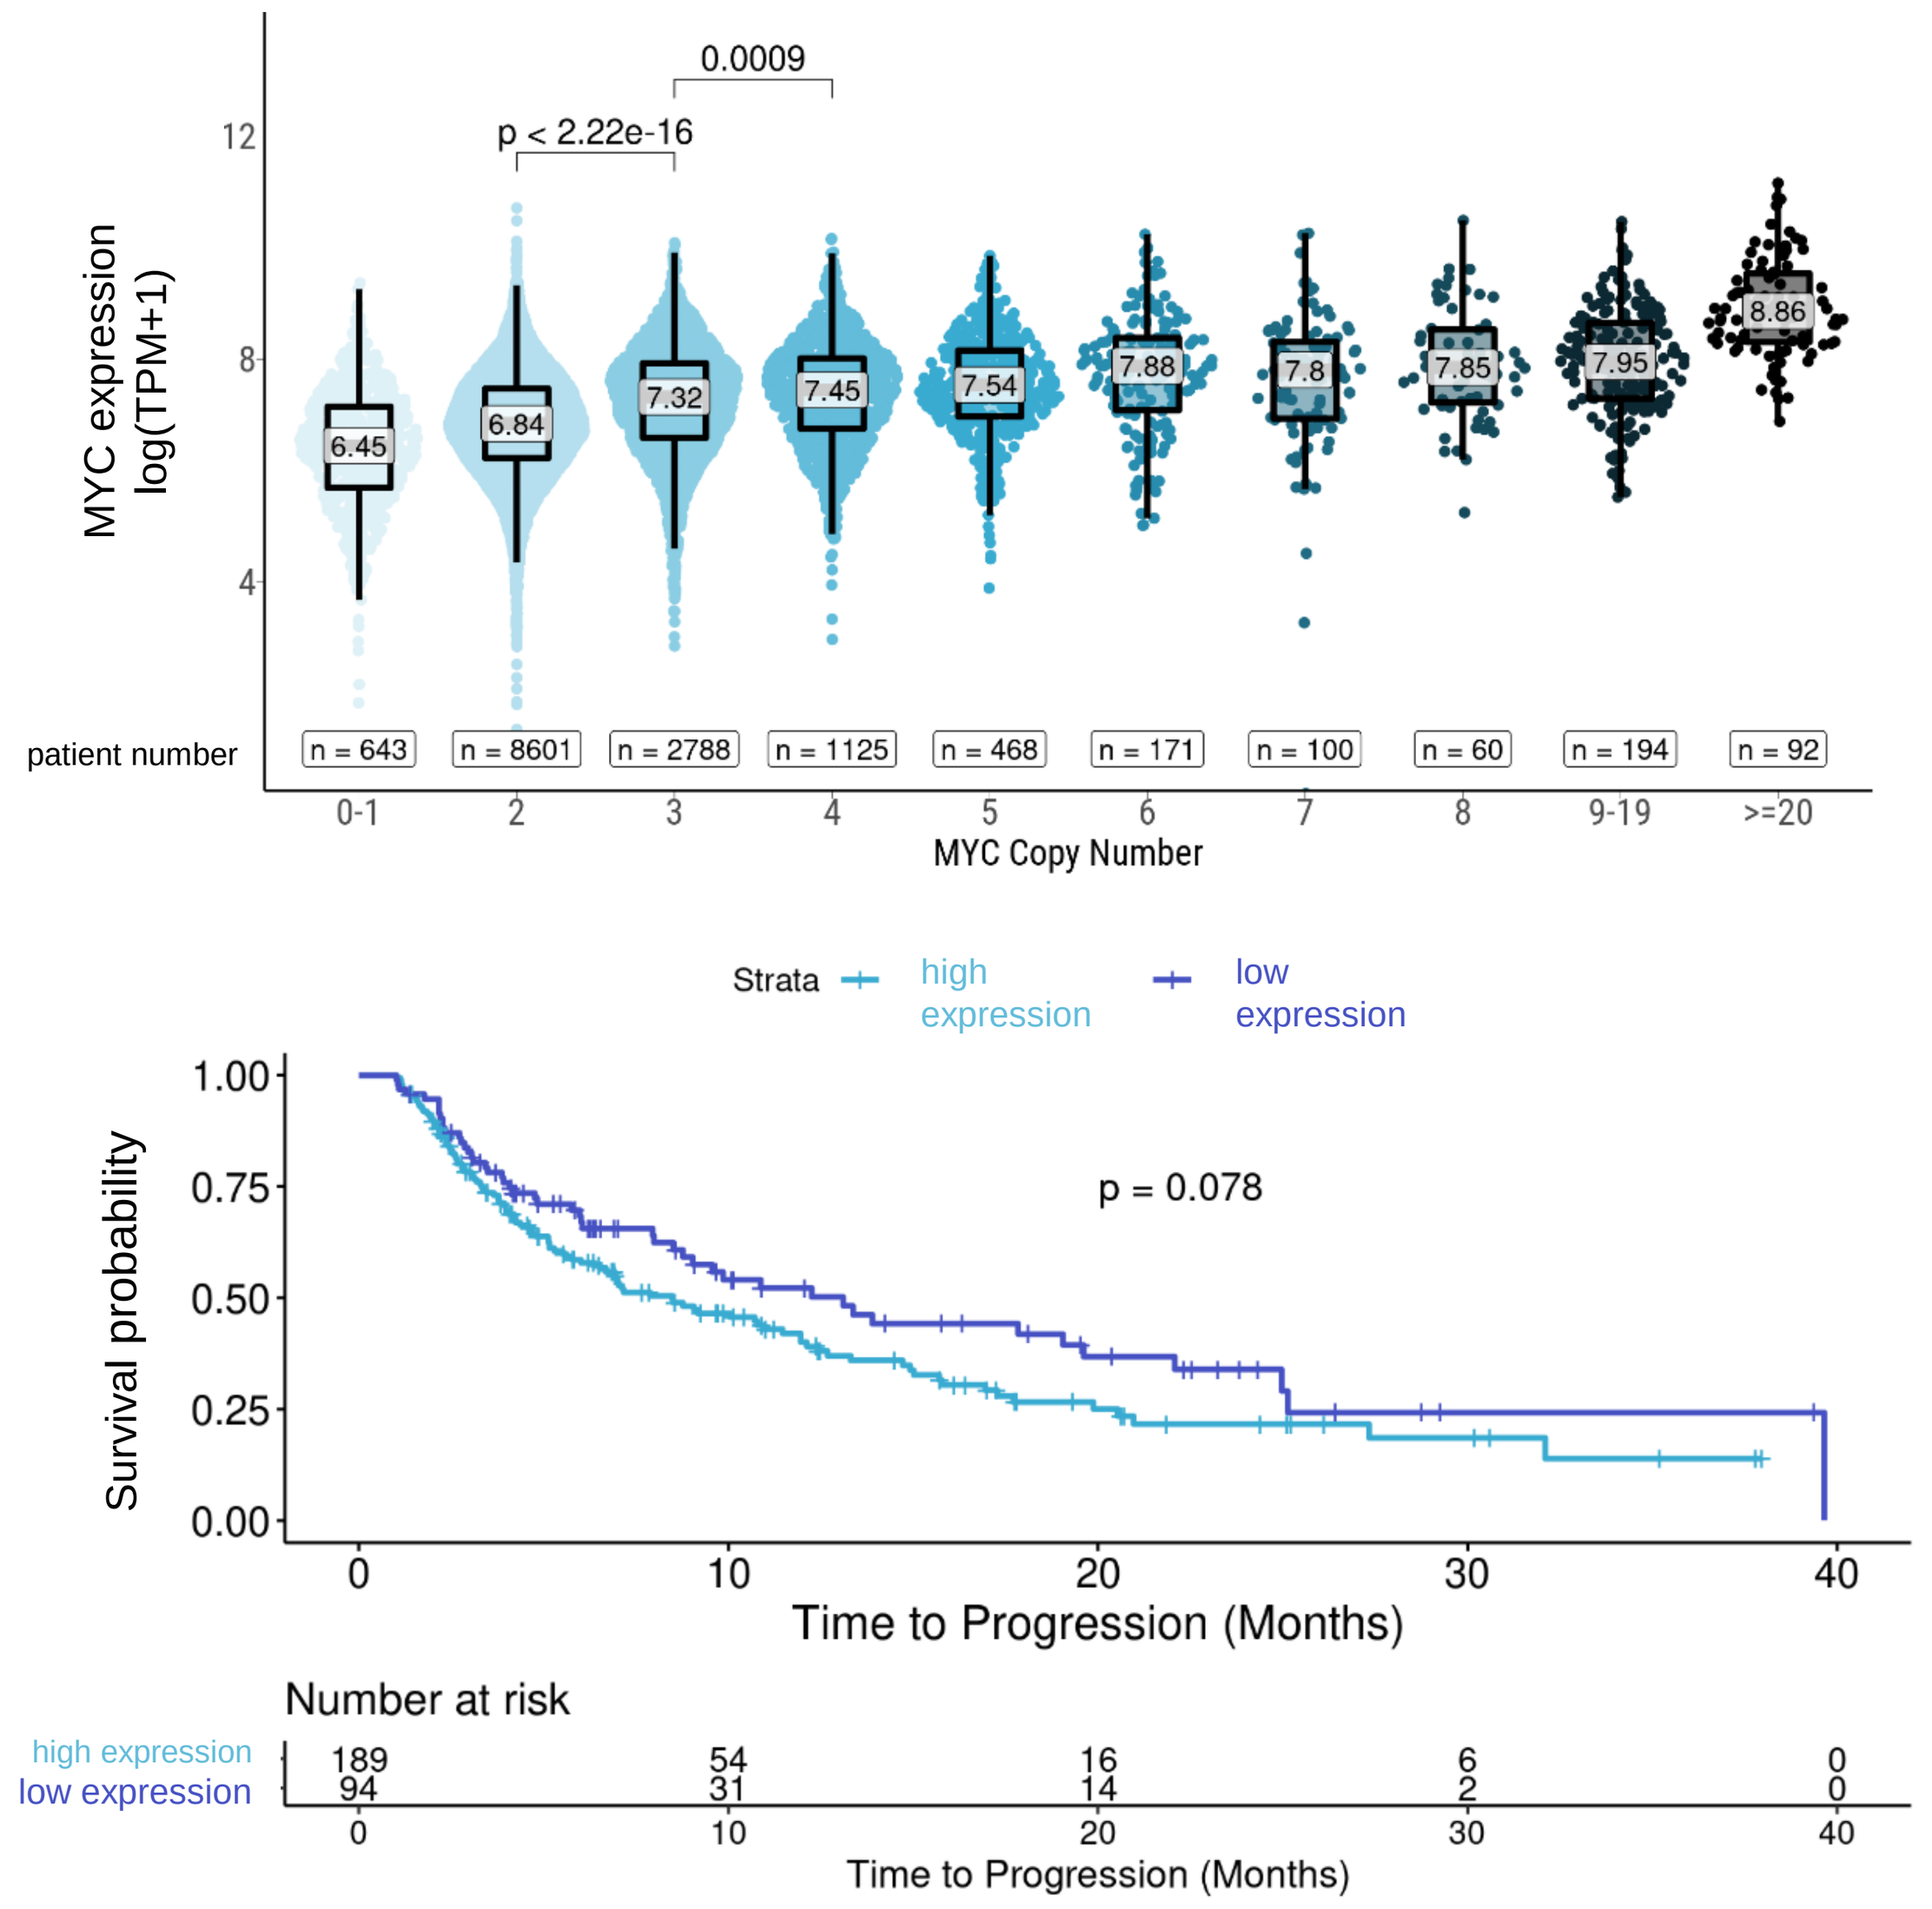
0.0009
p < 2.22e-16
MYC expression log(TPM+1)
6.45
6.84
7.32
7.45
7.54
7.88
7.8
7.85
7.95
8.86
patient number
n = 643
n = 8601
n = 2788
n = 1125
n = 468
n = 171
n = 100
n = 60
n = 194
n = 92
0-1
2
3
4
5
6
7
8
9-19
>=20
MYC Copy Number
Strata
high expression
low expression
p = 0.078
Survival probability
Time to Progression (Months)
Number at risk
high expression 189 54 16 6 0
low expression 94 31 14 2 0

Figure 4: Top - Correlation of *MYC* mRNA expression (log2(TPM+1)) and MYC copy number (CN) in NSCLC patients. P-values comparing CN are labeled. Tumors with mRNA >log2 (6.84) (i.e. median CN for diploid tumors) defined as '*MYC* high' and ≤ log2 (6.84) defined as '*MYC* low'. **Bottom** - Univariate Kaplan-Meier analysis of *MYC* expression and time to progression using real-world data from 283 patients treated with pembrolizumab. NSCLC patients with high *MYC* mRNA expression trend towards a shorter time to progression in comparison to patients with low *MYC* levels. (Analysis performed in collaboration with Tempus)

MYC mRNA expression positively correlates with PD-L1 and PD1 mRNA levels in NSCLC patients

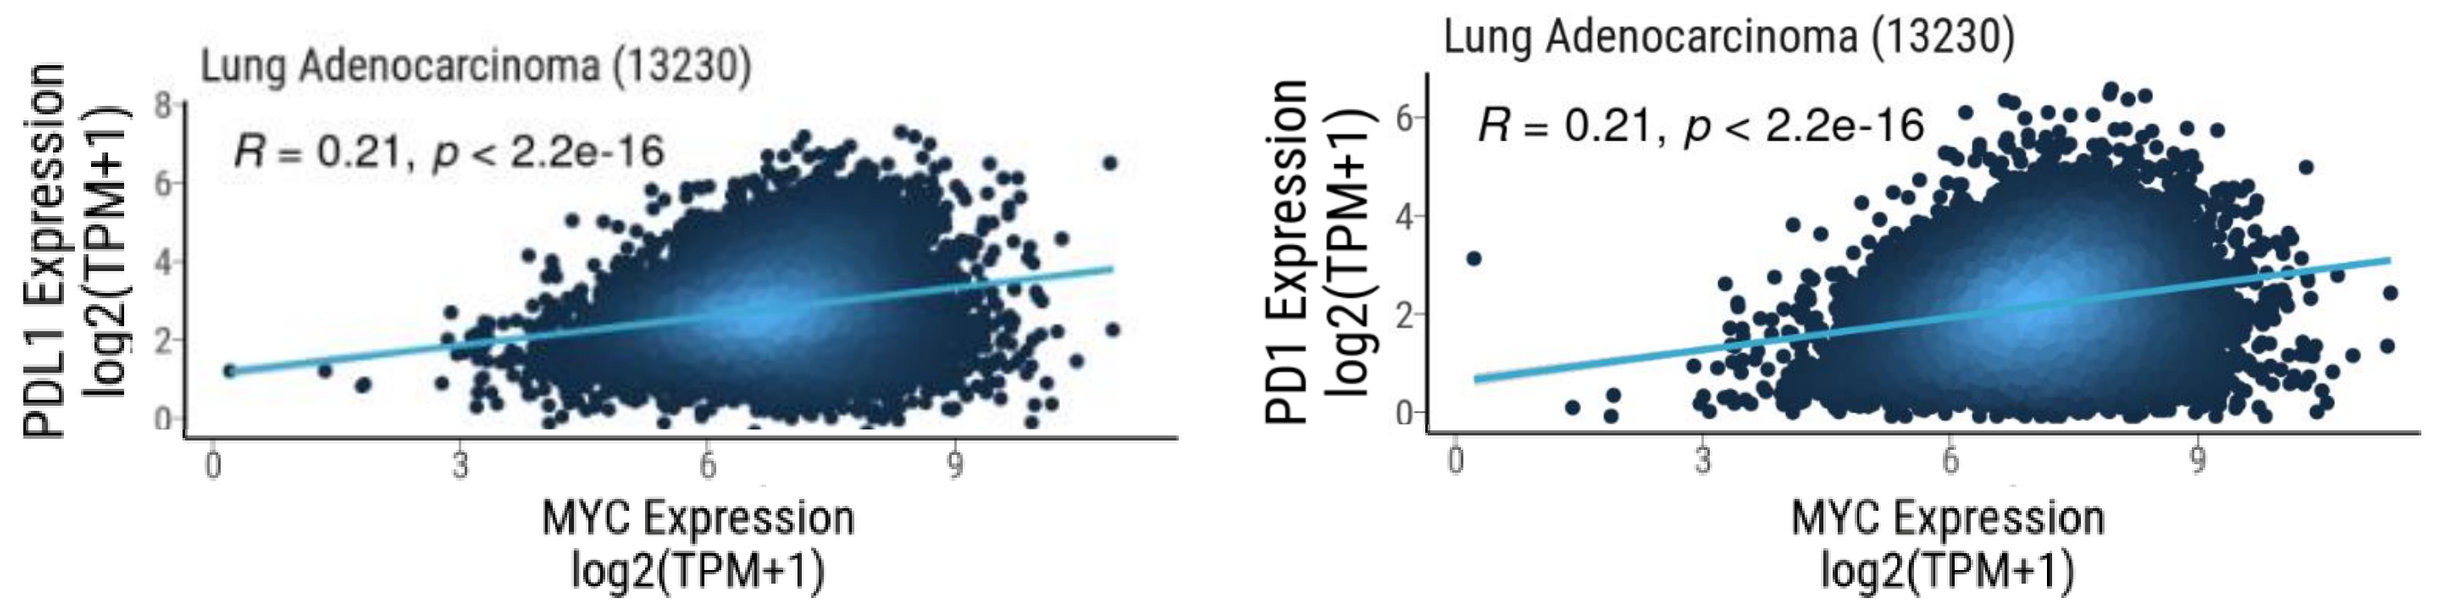
Lung Adenocarcinoma (13230)
R = 0.21, p < 2.2e-16
PDL1 Expression log2(TPM+1)
MYC Expression log2(TPM+1)
Lung Adenocarcinoma (13230)
R = 0.21, p < 2.2e-16
PD1 Expression log2(TPM+1)
MYC Expression log2(TPM+1)

Figure 5: Dot plot shows significant, positive correlation of *MYC* mRNA versus PD-L1 & PD1 expression [log2(TPM+1)] quantified by RNASeq from 13230 lung adenocarcinoma patients. (Analysis performed in collaboration with Tempus)

Combination of Mu-MYC EC with anti-PD-L1 antibody leads to more effective tumor growth inhibition in syngeneic lung model

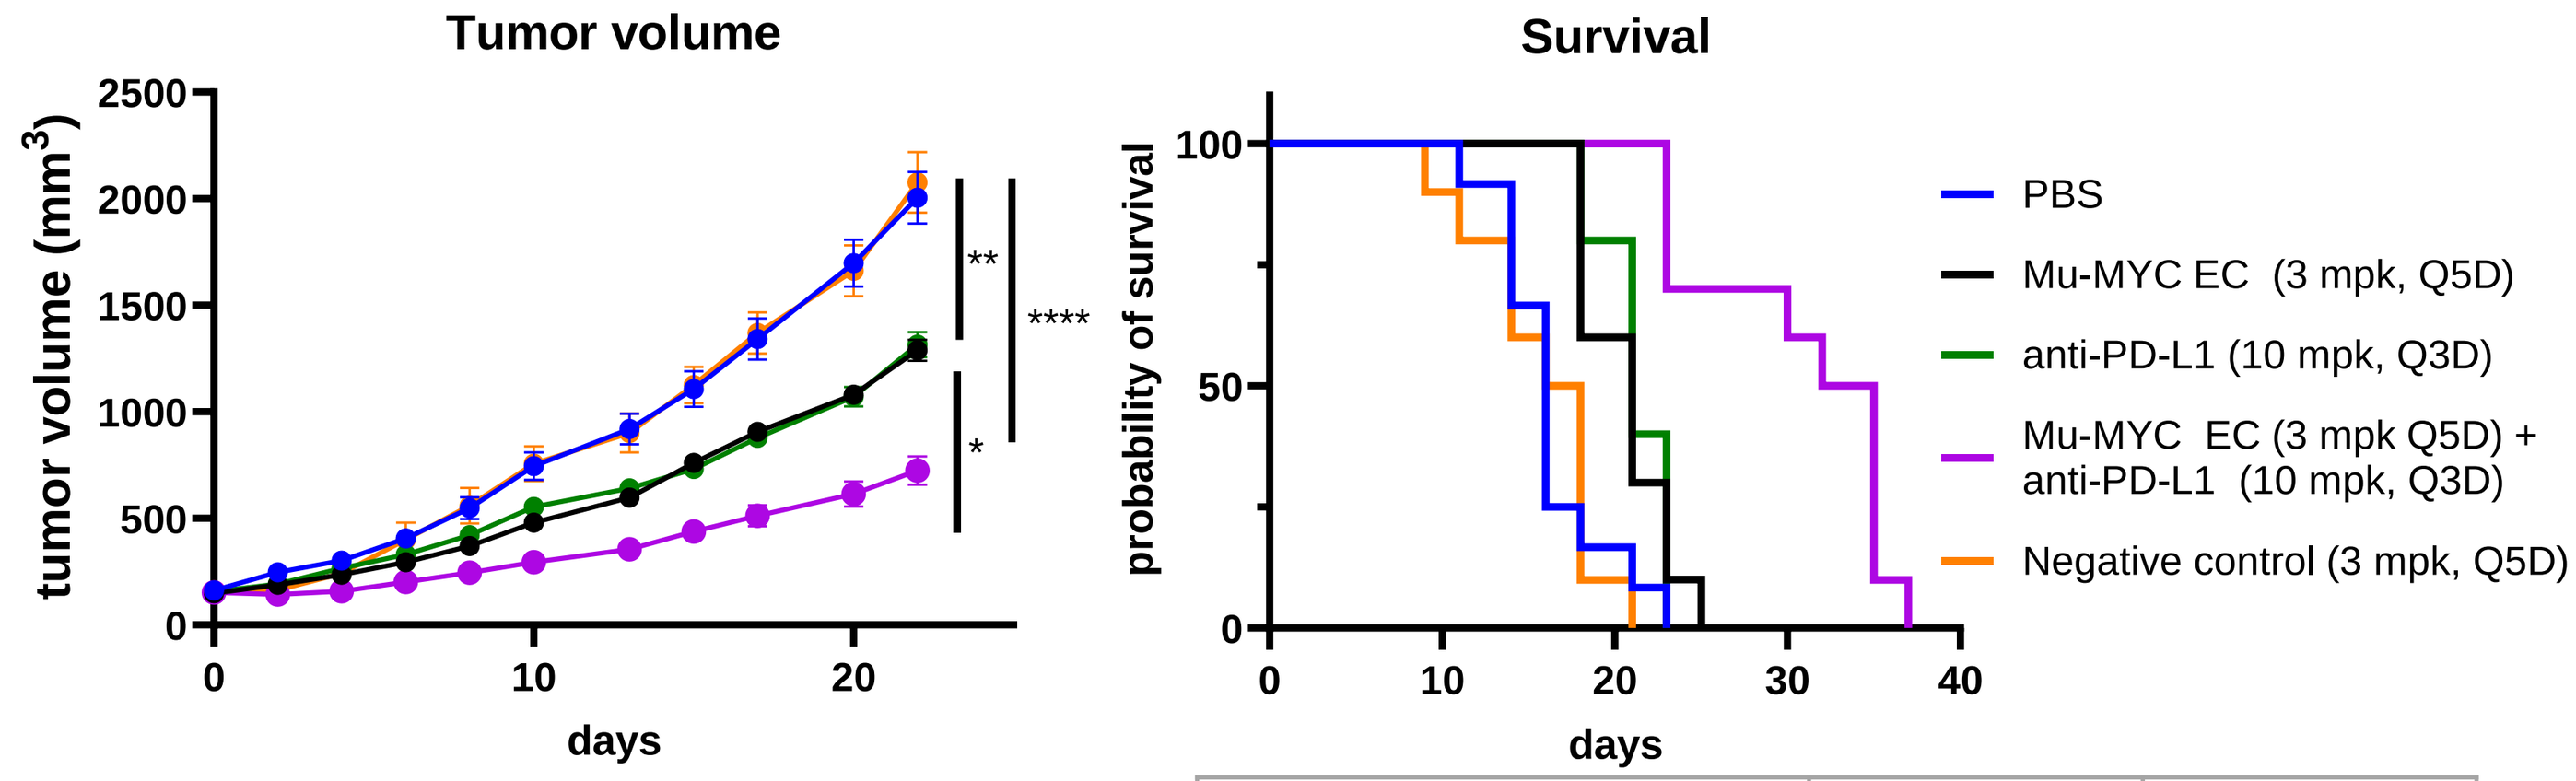
Tumor volume
tumor volume (mm3)
days
Survival
probability of survival
days
PBS
Mu-MYC EC (3 mpk, Q5D)
anti-PD-L1 (10 mpk, Q3D)
Mu-MYC EC (3 mpk Q5D) + anti-PD-L1 (10 mpk, Q3D)
Negative control (3 mpk, Q5D)

Survival stats	muMYC-EC	anti-PD-L1
Mu-MYC-EC + anti-PD-L1 vs.	****	****

Figure 6: Mu-MYC-EC (mouse surrogate EC) treatment reduces Lewis lung carcinoma subcutaneous tumor growth in vivo (left) and extends survival to 1000 mm³ (right) more effectively in combination with anti-PD-L1 antibody (clone 10F.9G2). Significant differences in tumor size and median survival duration between selected groups are shown; "****" denotes p<0.0001, '**' denotes p<0.01, '*' denotes p<0.05.

High *MYC* mRNA levels correlate with faster progression in osimertinib-treated NSCLC patients

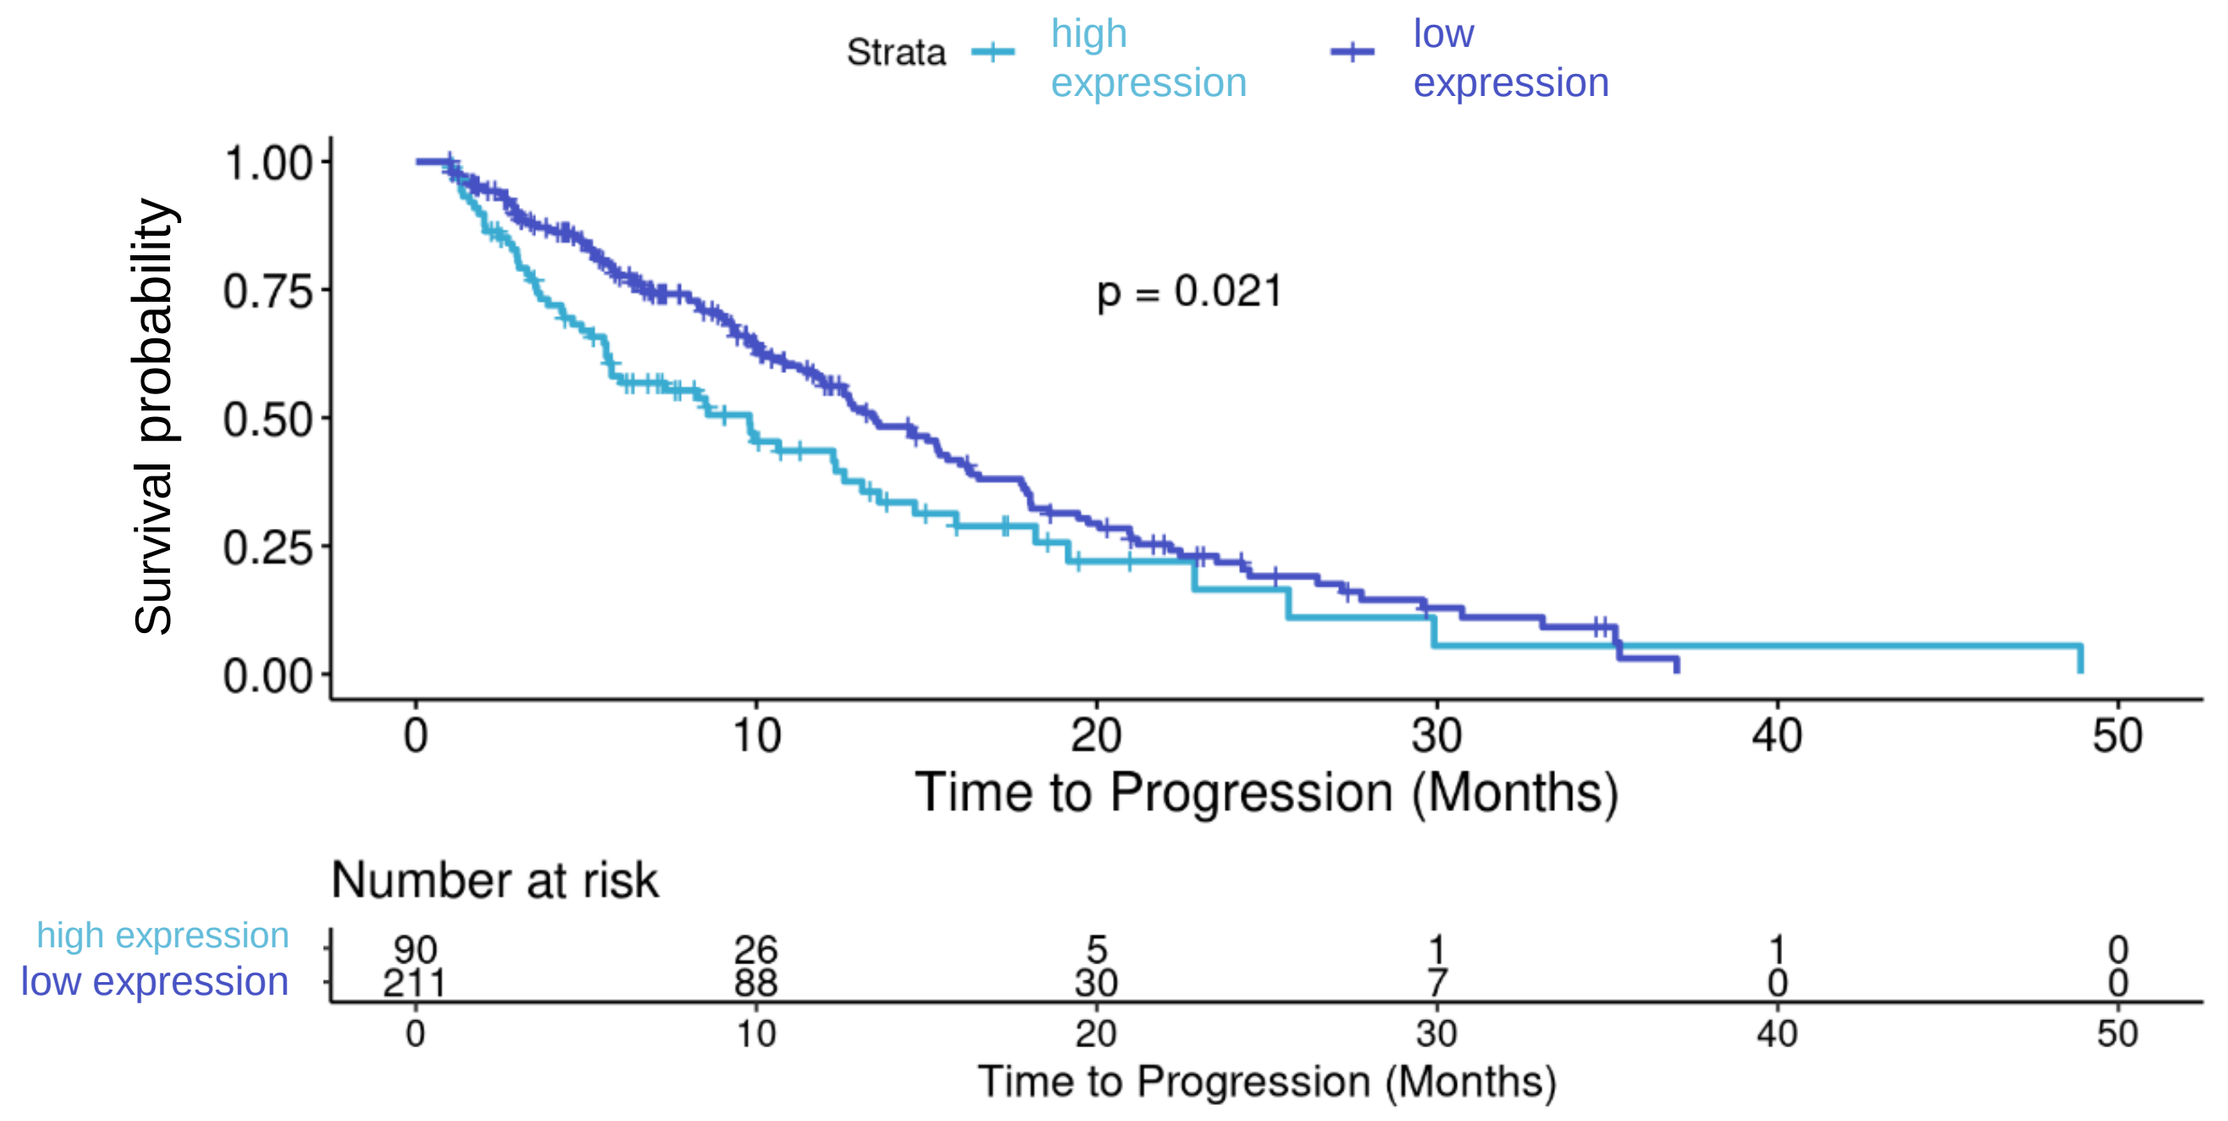
Strata
high expression
low expression
p = 0.021
Survival probability
Time to Progression (Months)
Number at risk
high expression 90 26 5 1 1 0
low expression 211 88 30 7 0 0

Figure 7: Univariate Kaplan-Meier analysis of MYC *mRNA* expression and time to progression using real-world data from 283 patients treated with osimertinib (third generation EGFR inhibitor). NSCLC patients with high *MYC* mRNA levels show significantly shorter time to progression in comparison to patients with low *MYC* levels. See figure 4 for explanation of threshold. (Analysis performed in collaboration with Tempus).

Combination of NSCLC MYC-EC with EGFR inhibitor osimertinib enhances MYC protein downregulation relative to single agent

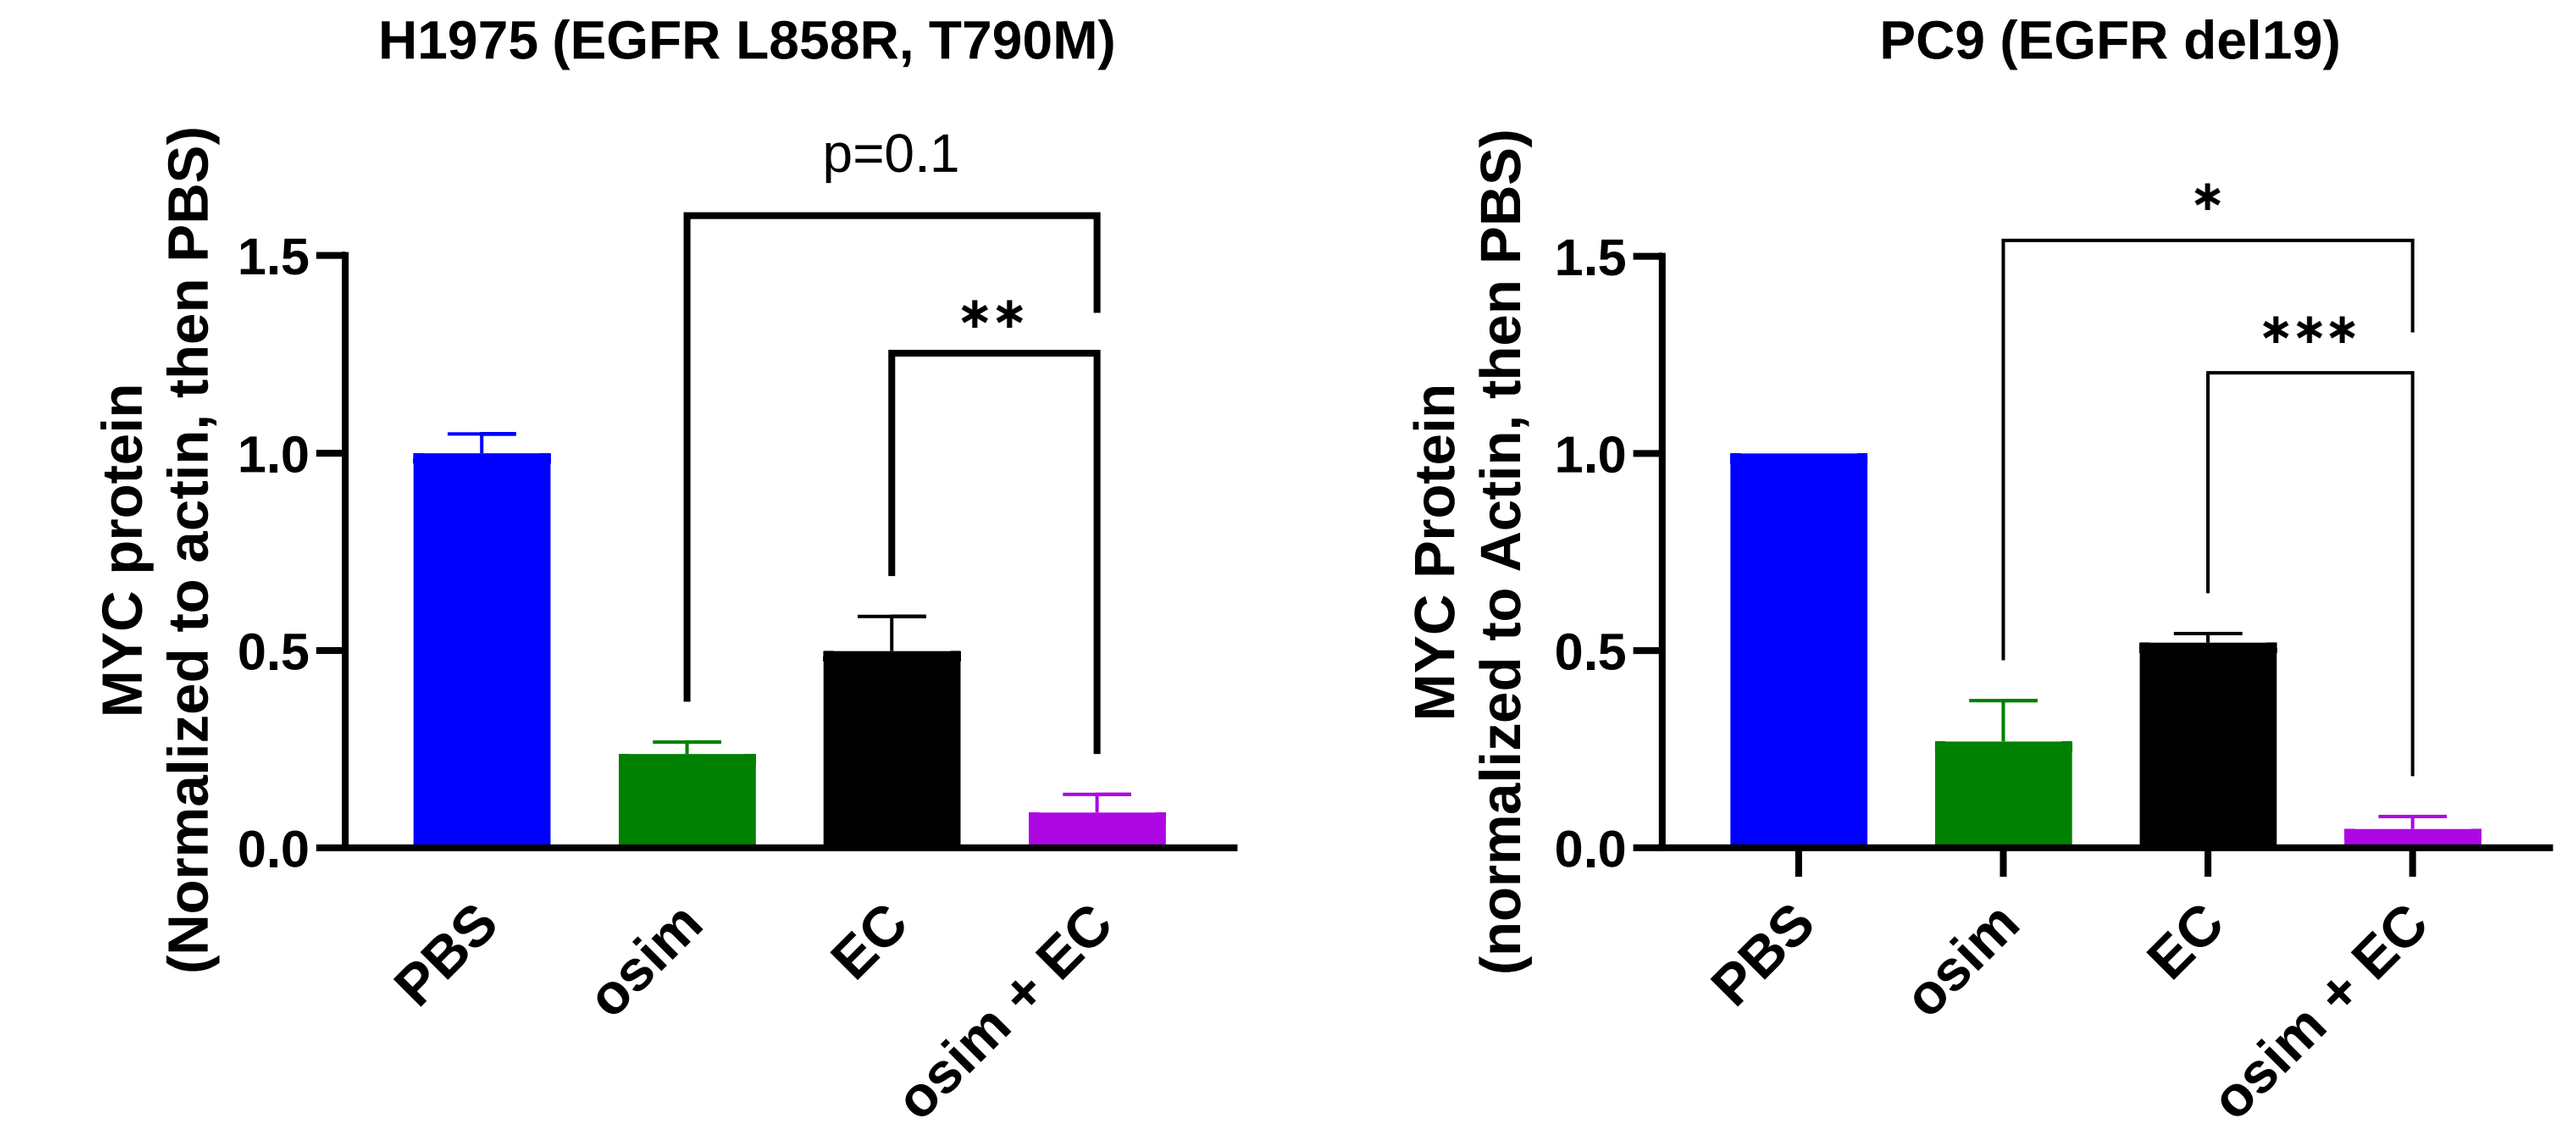
H1975 (EGFR L858R, T790M)
p=0.1
**
MYC protein (Normalized to actin, then PBS)
PBS
osim
EC
osim + EC
PC9 (EGFR del19)
*

MYC Protein (normalized to Actin, then PBS)

Figure 8: NSCLC EC combination therapy with EGFR inhibitor (osimertinib) in H1975 cells (L858R, T790M mutant EGFR, left panel) and PC9 cells (del19 mutant EGFR, right panel) enhances MYC protein downregulation as compared to monotherapy. H1975 or PC9 cells were treated with PBS, 1 uM osimertinib (osim), 0.5 ug/ml NSCLC MYC-EC (EC) or a combination of both. "***" denotes p<0.001, '**' denotes p<0.01, '*' denotes p<0.05, p=0.1 shows trend towards significance.

NSCLC MYC-EC enhances osimertinib-induced inhibition of cell viability in EGFR-mutant NSCLC cell lines

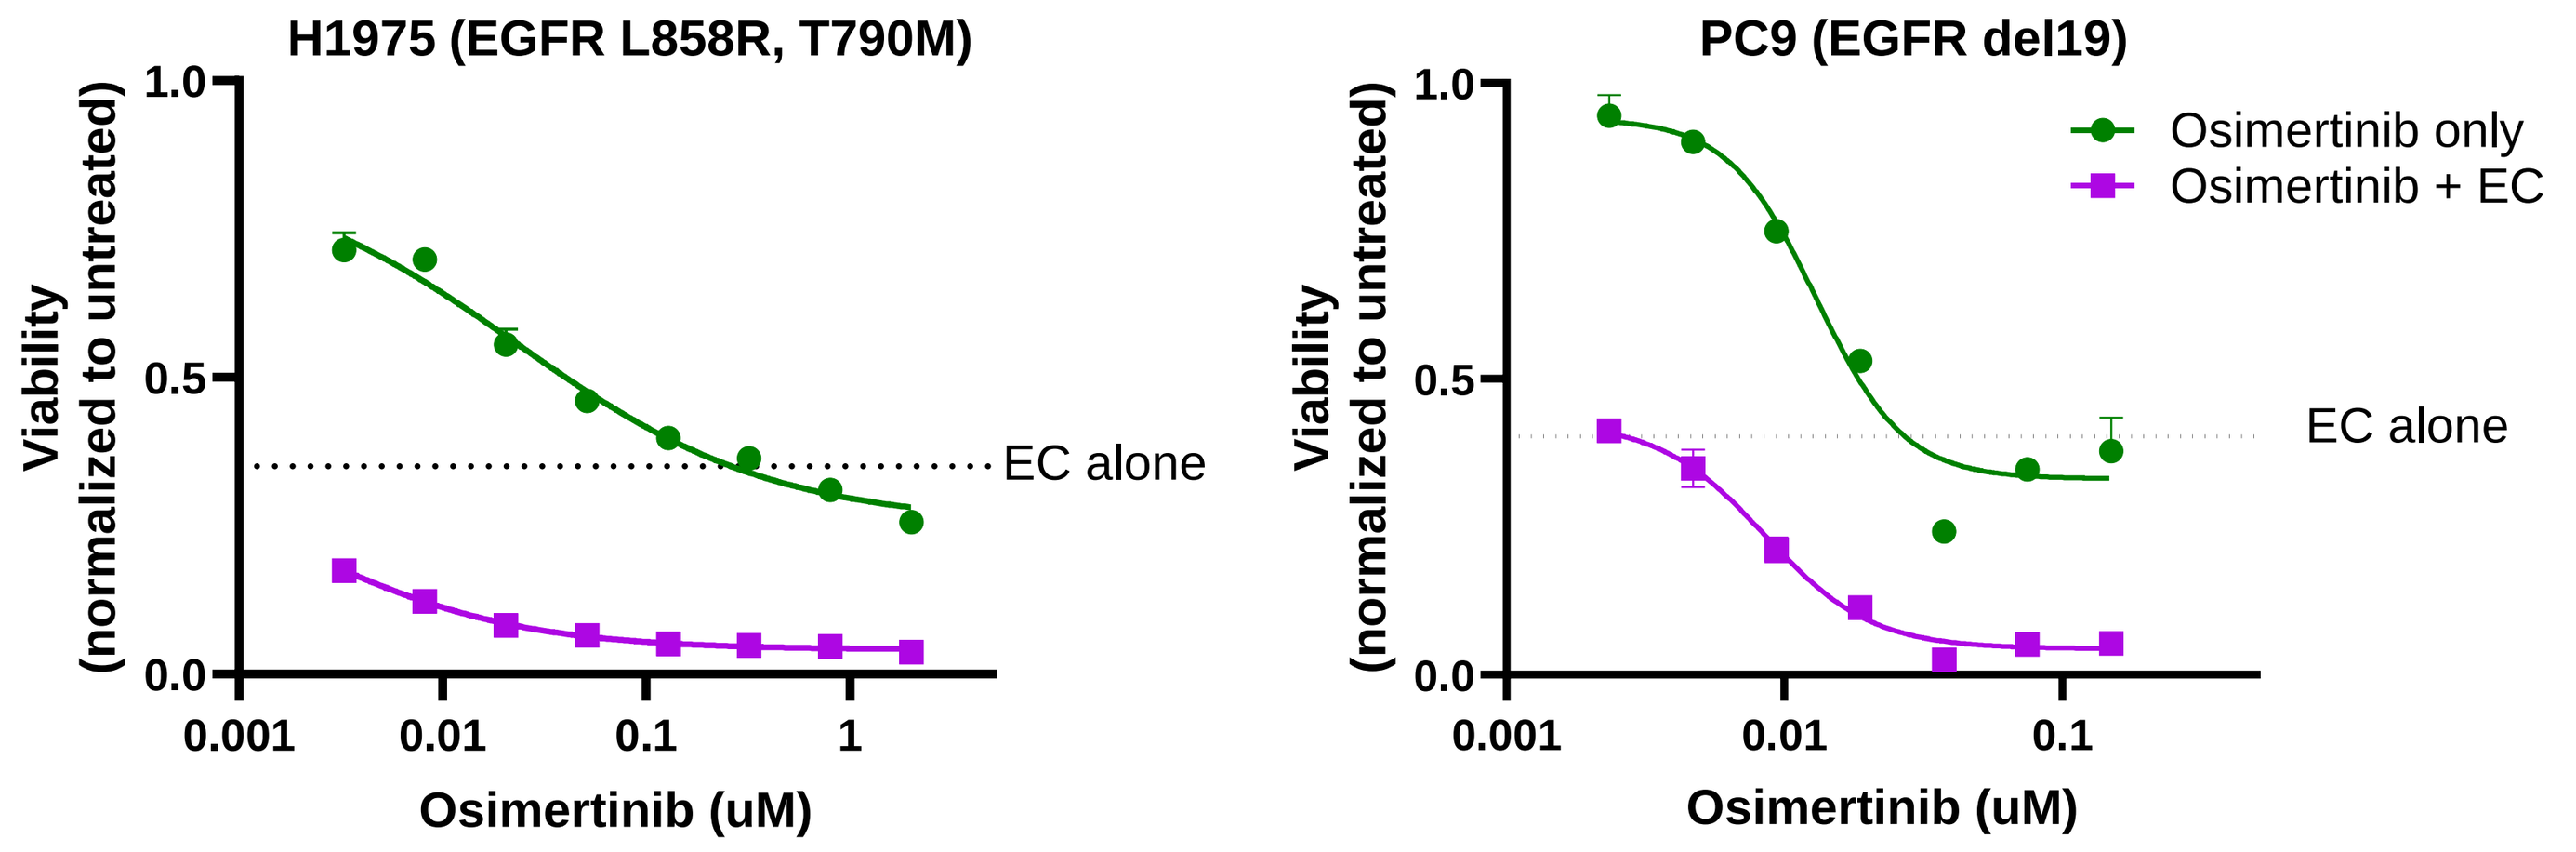
H1975 (EGFR L858R, T790M)
Viability (normalized to untreated)
EC alone
Osimertinib (uM)
PC9 (EGFR del19)
Osimertinib only
Osimertinib + EC
EC alone
Osimertinib (uM)

Figure 9: NSCLC MYC-EC (0.625 ug/mL) administered in combination with increasing concentrations of osimertinib (3 nM - 2 uM) in H1975 (left panel) cells and PC9 cells (right panel) shown.

NSCLC MYC-EC synergizes with osimertinib to reduce viability in NSCLC cells in vitro

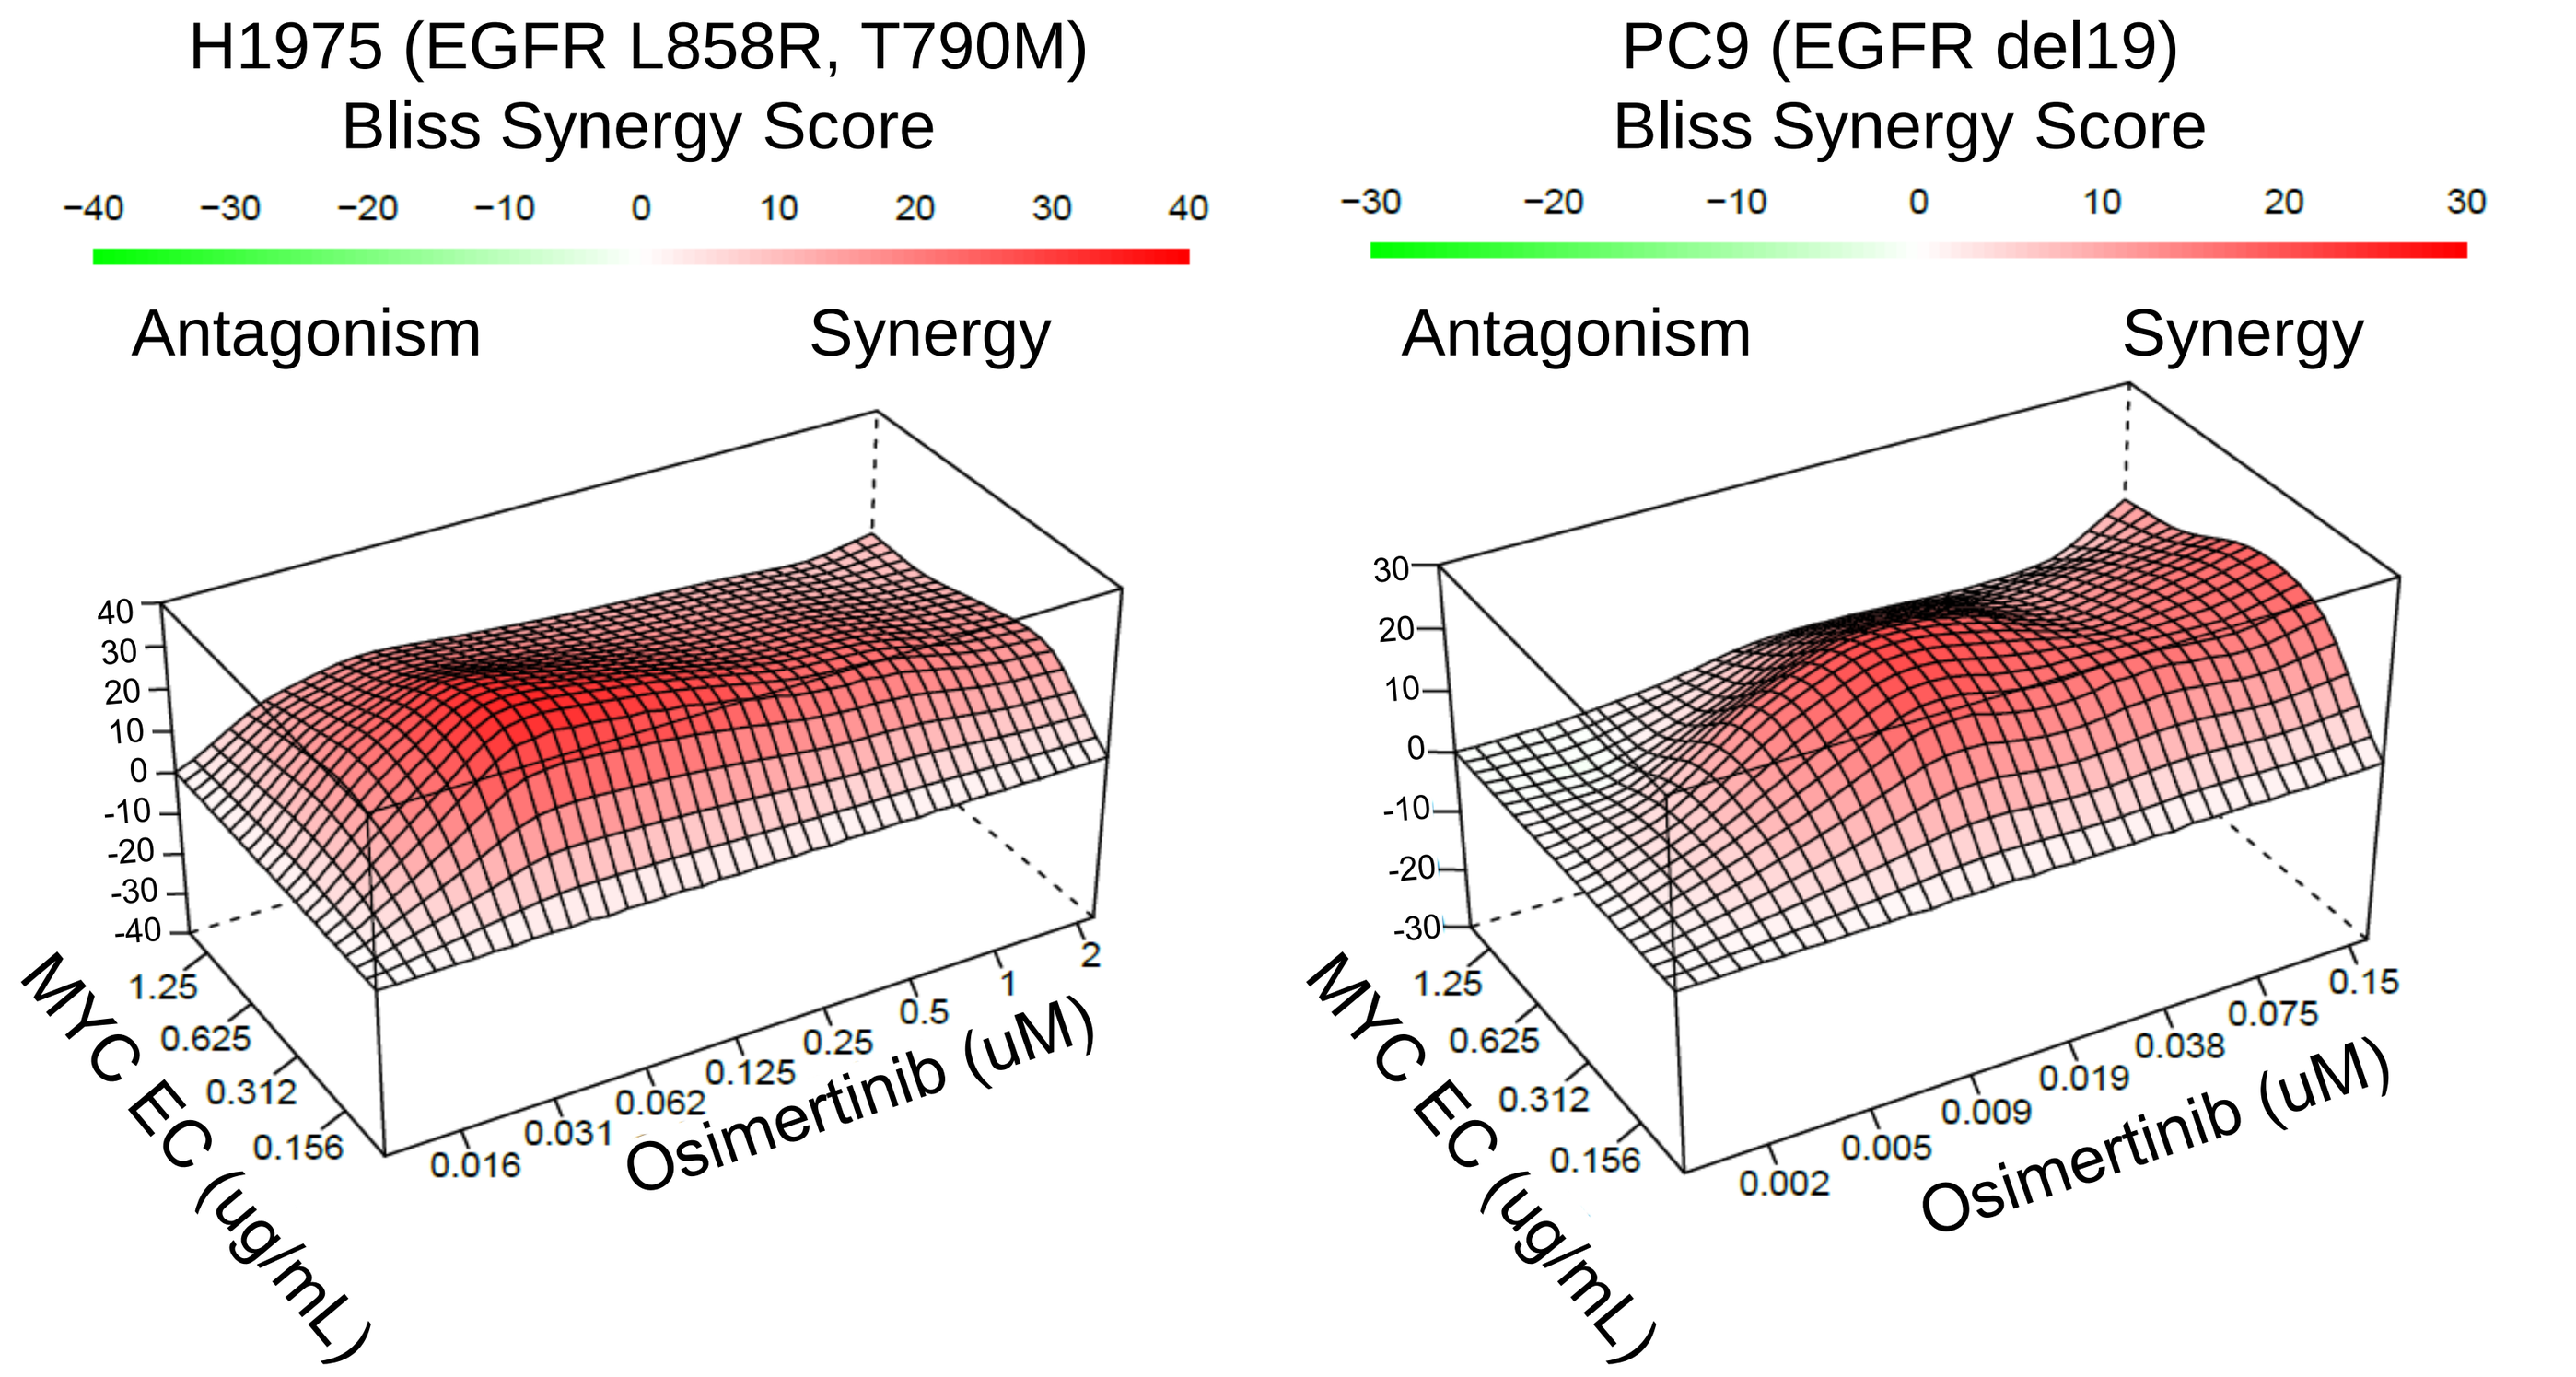
H1975 (EGFR L858R, T790M)
Bliss Synergy Score
Antagonism
Synergy
MYC EC (ug/mL)
Osimertinib (uM)
PC9 (EGFR del19)
Bliss Synergy Score
Antagonism
Synergy
MYC EC (ug/mL)
Osimertinib (uM)

Figure 10: Bliss synergy analysis of varying NSCLC MYC-EC concentrations (0.156-1.25 ug/mL) and osimertinib concentrations (0.02 – 2 uM in H1975, 2-150 nM in PC9) from data collected in H1975 (left) and PC9 (right) cells. Bliss scores from the most synergistic areas are 27 and 16 for H1975 and PC9 cells, respectively. Score ≥ 10 demonstrates synergy.

NSCLC MYC-EC plus osimertinib treatment significantly enhances tumor growth inhibition and survival in H1975 tumor-bearing mice

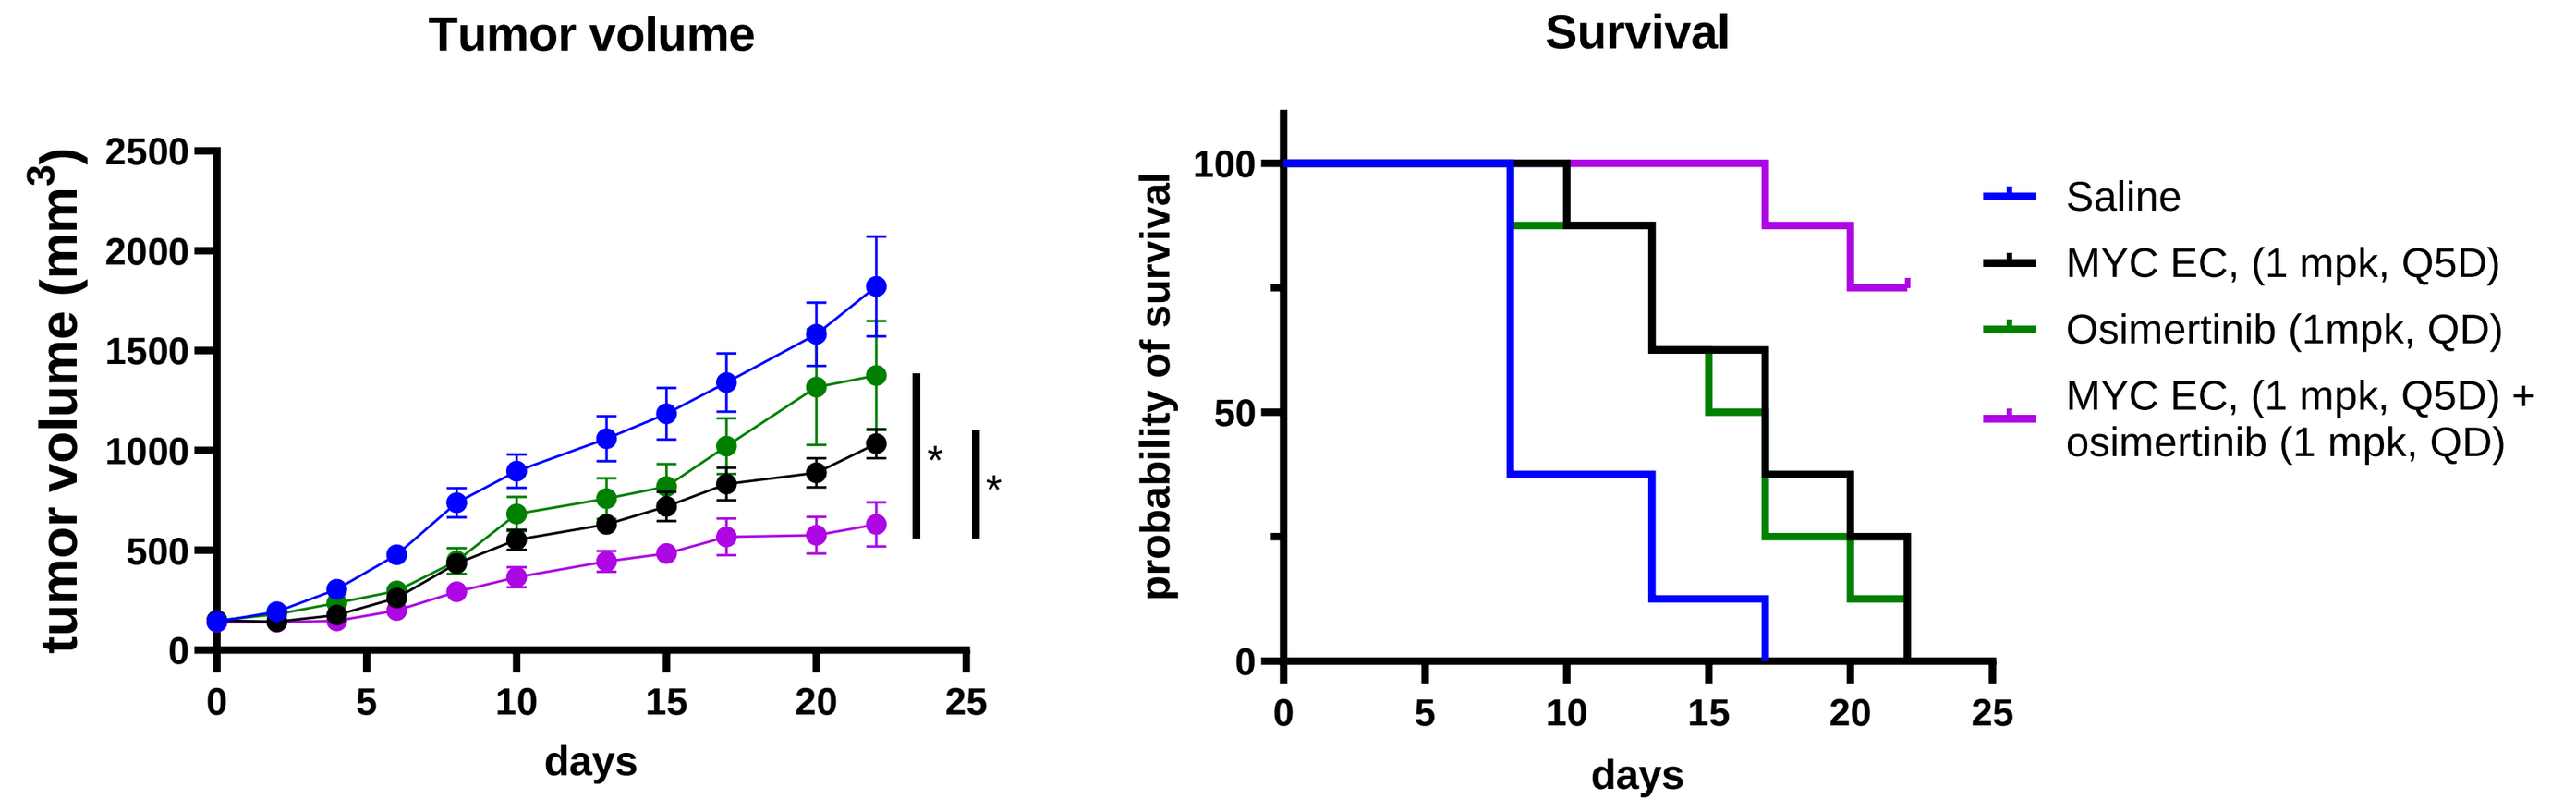
Tumor volume
tumor volume (mm3)
days
Survival
probability of survival
days
Saline
MYC EC, (1 mpk, Q5D)
Osimertinib (1mpk, QD)
MYC EC, (1 mpk, Q5D) + osimertinib (1 mpk, QD)

Survival stats	MYC-EC	osimertinib
NSCLC MYC-EC + osimertinib vs.	**	**

Figure 11: NSCLC MYC-EC treatment reduces subcutaneous tumor growth in vivo (left panel) and extends survival to 750 mm³ (right panel). H1975 subQ tumors were treated with PBS, NSCLC MYC EC LNP, osimertinib, or a combination of NSCLC MYC EC and osimertinib. LNPs were dosed Q5D and osimertinib QD. Significant differences in tumor size and median survival duration between selected groups are shown; '**' denotes p<0.01 and '*' denotes p<0.05.

Conclusions

- High *MYC* mRNA levels correlate with faster progression in pembrolizumab-treated and osimertinib-treated NSCLC patients.
- Targeted epigenomic MYC downregulation or combining NSCLC MYC-EC with immune checkpoint or EGFR inhibitors enhances responses.
 - Treatment of syngeneic lung tumor-bearing mice with mouse MYC-EC and anti-PD-L1 antibody significantly inhibits tumor growth to a greater extent than either therapy alone.
 - Combination of NSCLC MYC-EC with EGFR inhibitor (osimertinib) downregulates MYC protein to a greater extent than single agent; combination of NSCLC MYC-EC with osimertinib synergistically reduces tumor cell viability in vitro and significantly reduces tumor growth in vivo.

These results support development of NSCLC MYC-EC as a monotherapy and/or in combination with EGFR inhibitors and immune checkpoint inhibitors in NSCLC patients.

© Poster Template by Genigraphics® 1.800.790.4001 www.genigraphics.com