Induction of Hepatocyte Nuclear Factor 4 alpha (HNF4α) P1-isoforms Using a Selective Epigenomic Controller

Amy McCurley, Yoseph Kassa, Justin Chen, Wanzhu Zhao, Chris E. Pedigo, Charles W. O'Donnell, Joseph Newman, Thomas McCauley
Omega Therapeutics, Cambridge MA, USA

AASLD 2023
Poster #3444-A

Background

Hepatocyte nuclear factor 4 alpha (HNF4α) is a nuclear receptor and master regulator of liver development and function (Figure 1). HNF4α expression is dysregulated in fibrotic liver disease and upregulation of this key transcription factor has been shown to improve hepatocyte function. The HNF4α gene is driven by two promoters, with isoforms derived from the P1 promoter demonstrating enhanced transcriptional activity associated with therapeutic benefit.

The OMEGA platform develops programmable mRNA medicines called Epigenomic Controllers (ECs) that precisely tune gene expression by site-specifically modulating epigenetic state and can be delivered using tissue-specific lipid nanoparticles (LNPs; Figure 2). We designed an HNF4α-EC to induce selective upregulation of the P1 isoform of HNF4α in a liver-specific manner. This represents a novel therapeutic strategy for the treatment of fibrotic liver disease.

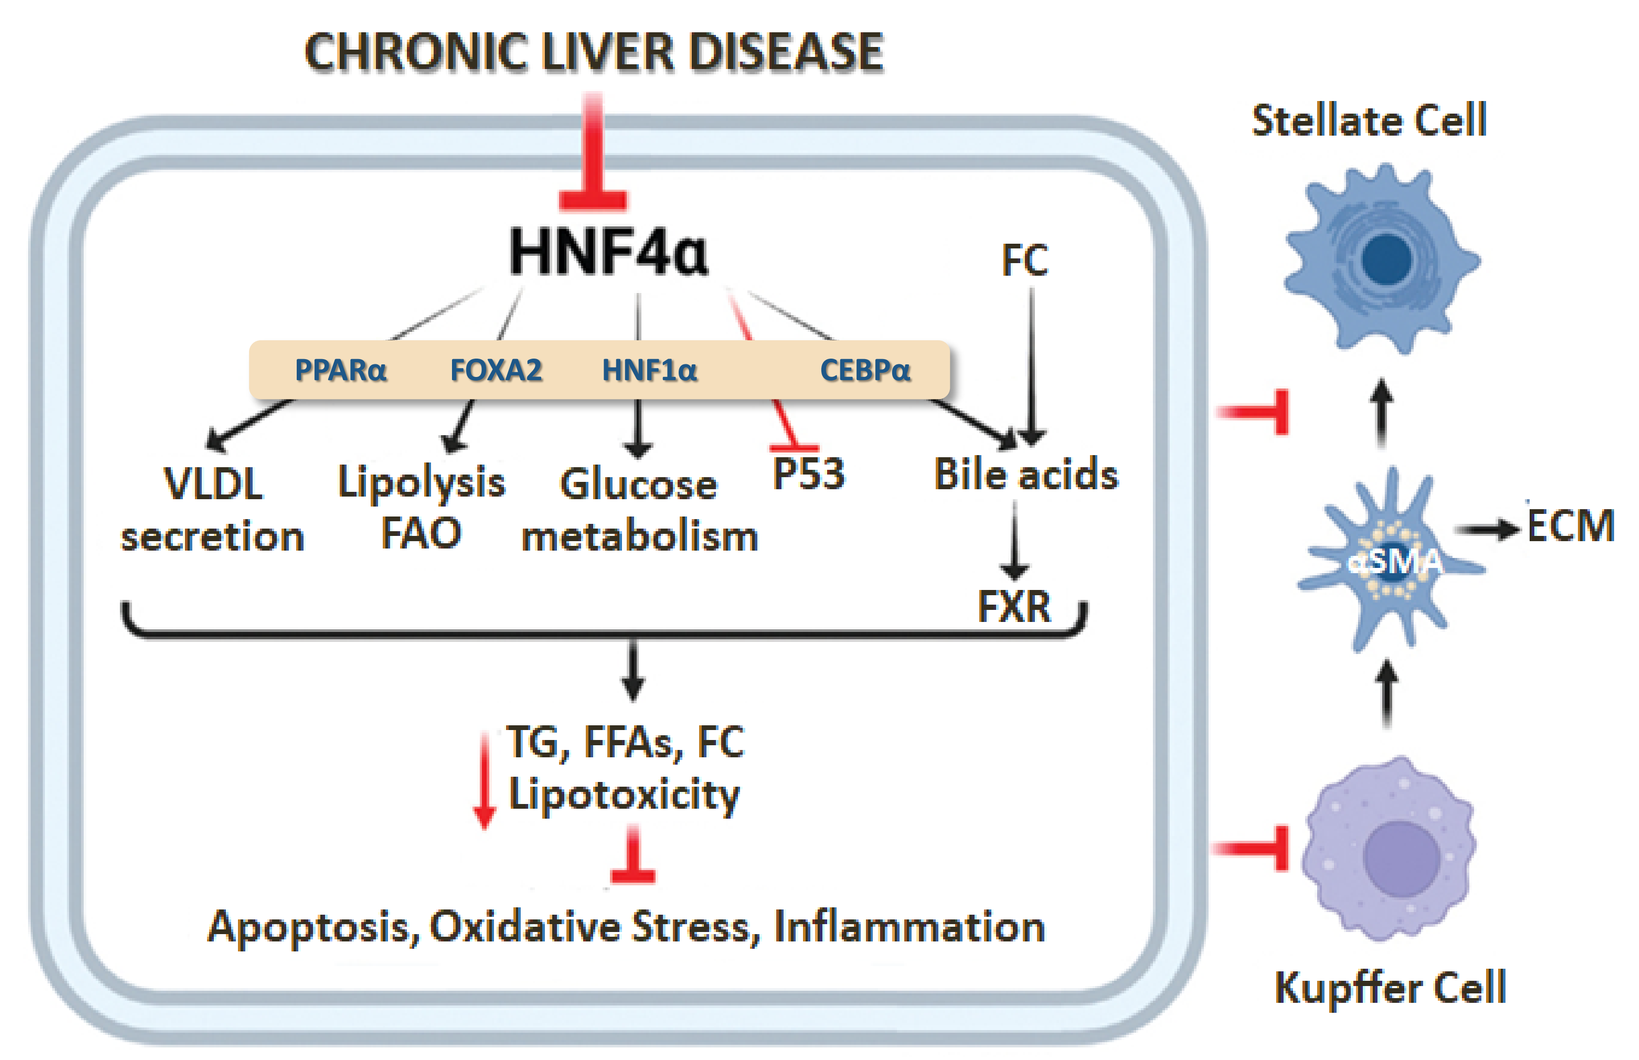
CHRONIC LIVER DISEASE
HNF4α
FC
PPARα FOXA2 HNF1α CEBPα
VLDL secretion
Lipolysis FAO
Glucose metabolism
P53
Bile acids
FXR
TG, FFAs, FC
Lipotoxicity
Apoptosis, Oxidative Stress, Inflammation
Stellate Cell
αSMA
ECM
Kupffer Cell

Modified from Pan X and Zhang Y, 2022

Figure 1. HNF4α role in liver disease. HNF4α regulates numerous genes, including a network of transcription factors, that control cholesterol, lipid, and glucose metabolism and homeostasis. Dysregulation of this signaling leads to apoptosis, oxidative stress, inflammation and fibrosis.

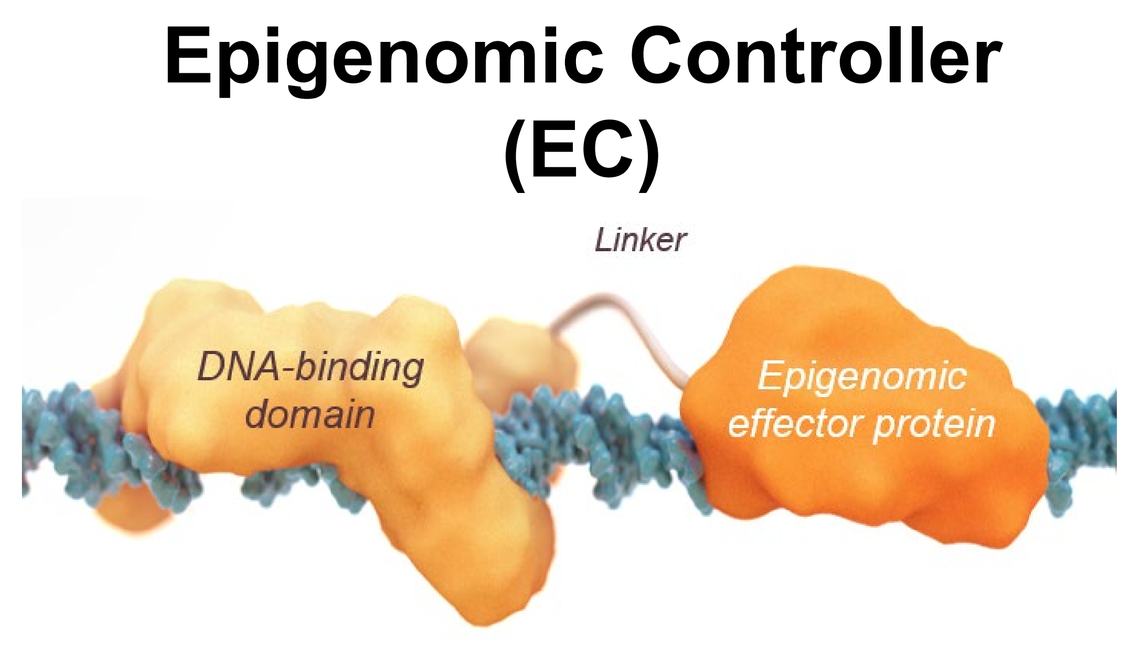
Epigenomic Controller
(EC)
Linker
DNA-binding domain
Epigenomic effector protein

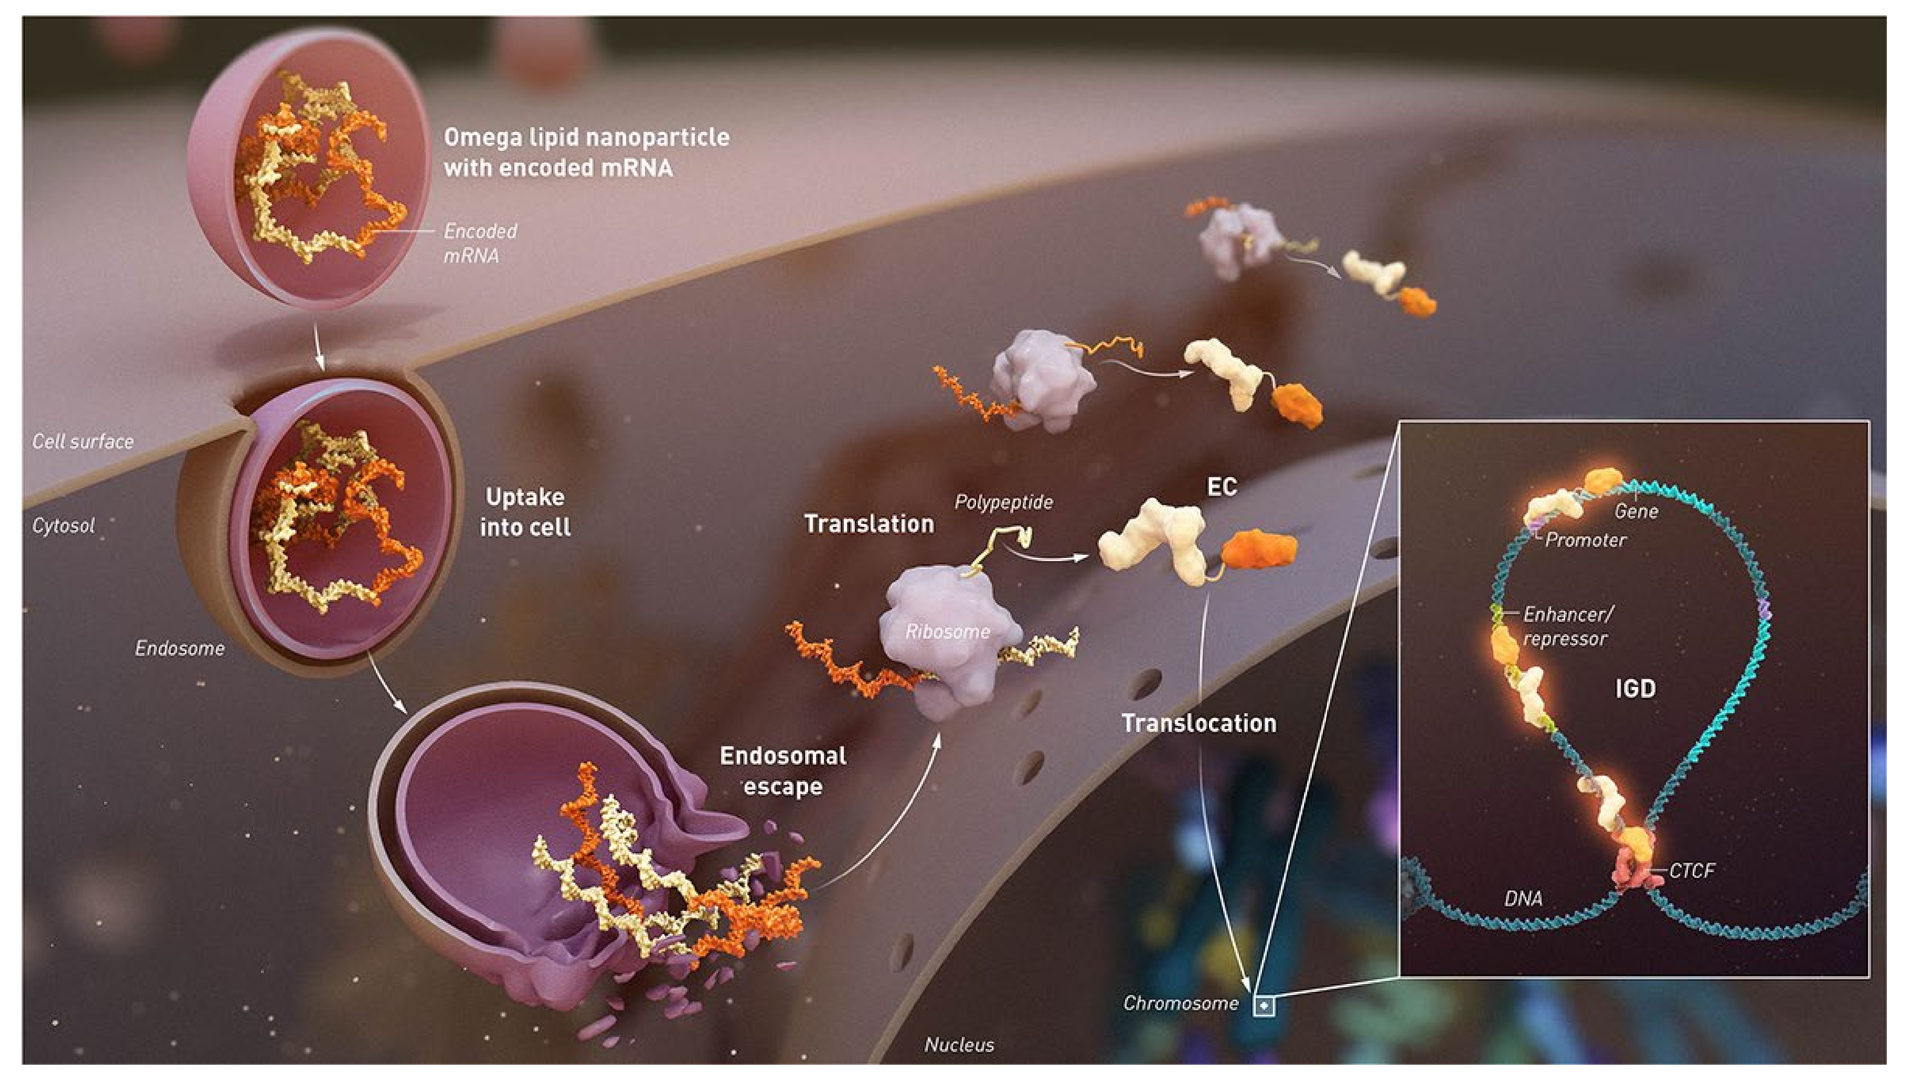
Omega lipid nanoparticle with encoded mRNA
Encoded mRNA
Cell surface
Cytosol
Uptake into cell
Endosome
Translation
Polypeptide
EC
Ribosome
Endosomal escape
Translocation
Gene
Promoter
Enhancer/ repressor
IGD
CTCF
DNA
Chromosome
Nucleus

Figure 2. Structure, Mechanism of Action and Design of ECs. Representative Insulated Genomic Domain (IGD; left) and schematic of Epigenomic Controller (EC; above) with DNA-binding domain and effector domain.

ECs induce upregulation of HNF4a expression *in vitro*

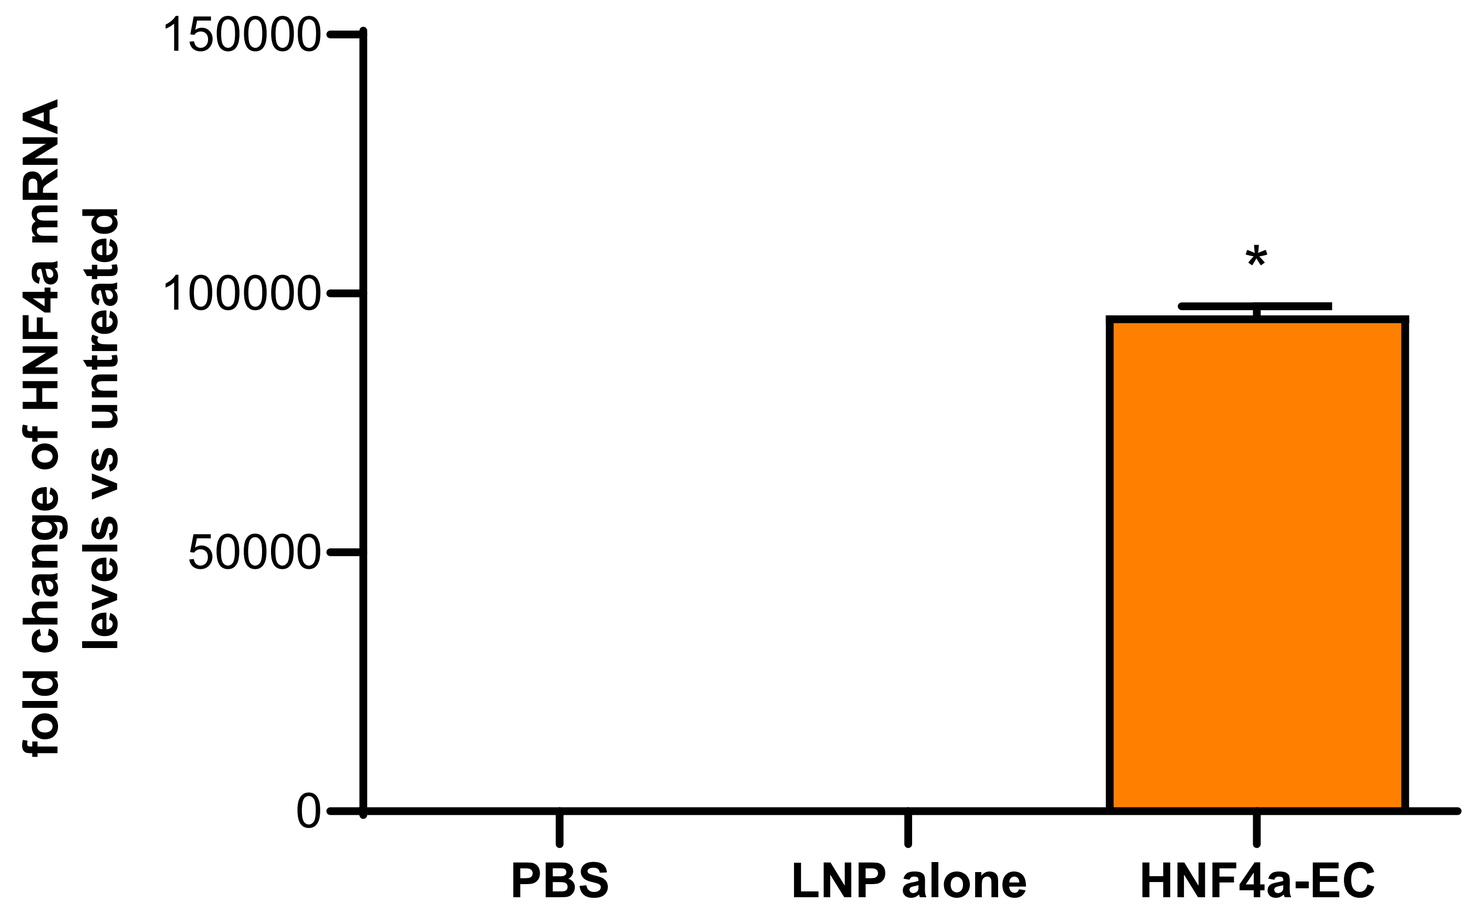
fold change of HNF4a mRNA levels vs untreated
150000
100000
50000
0
*
PBS
LNP alone
HNF4a-EC

Figure 3. HNF4α mRNA expression levels following treatment with HNF4α-EC. Hepatic stellate LX-2 cells were transfected with HNF4α-EC and HNF4α mRNA levels were measured by qPCR 24 hours later. HNF4α mRNA was nearly undetectable in LX-2 cells treated with PBS or LNP alone but was substantially upregulated with HNF4α-EC treatment. *1-way ANOVA , p<0.001

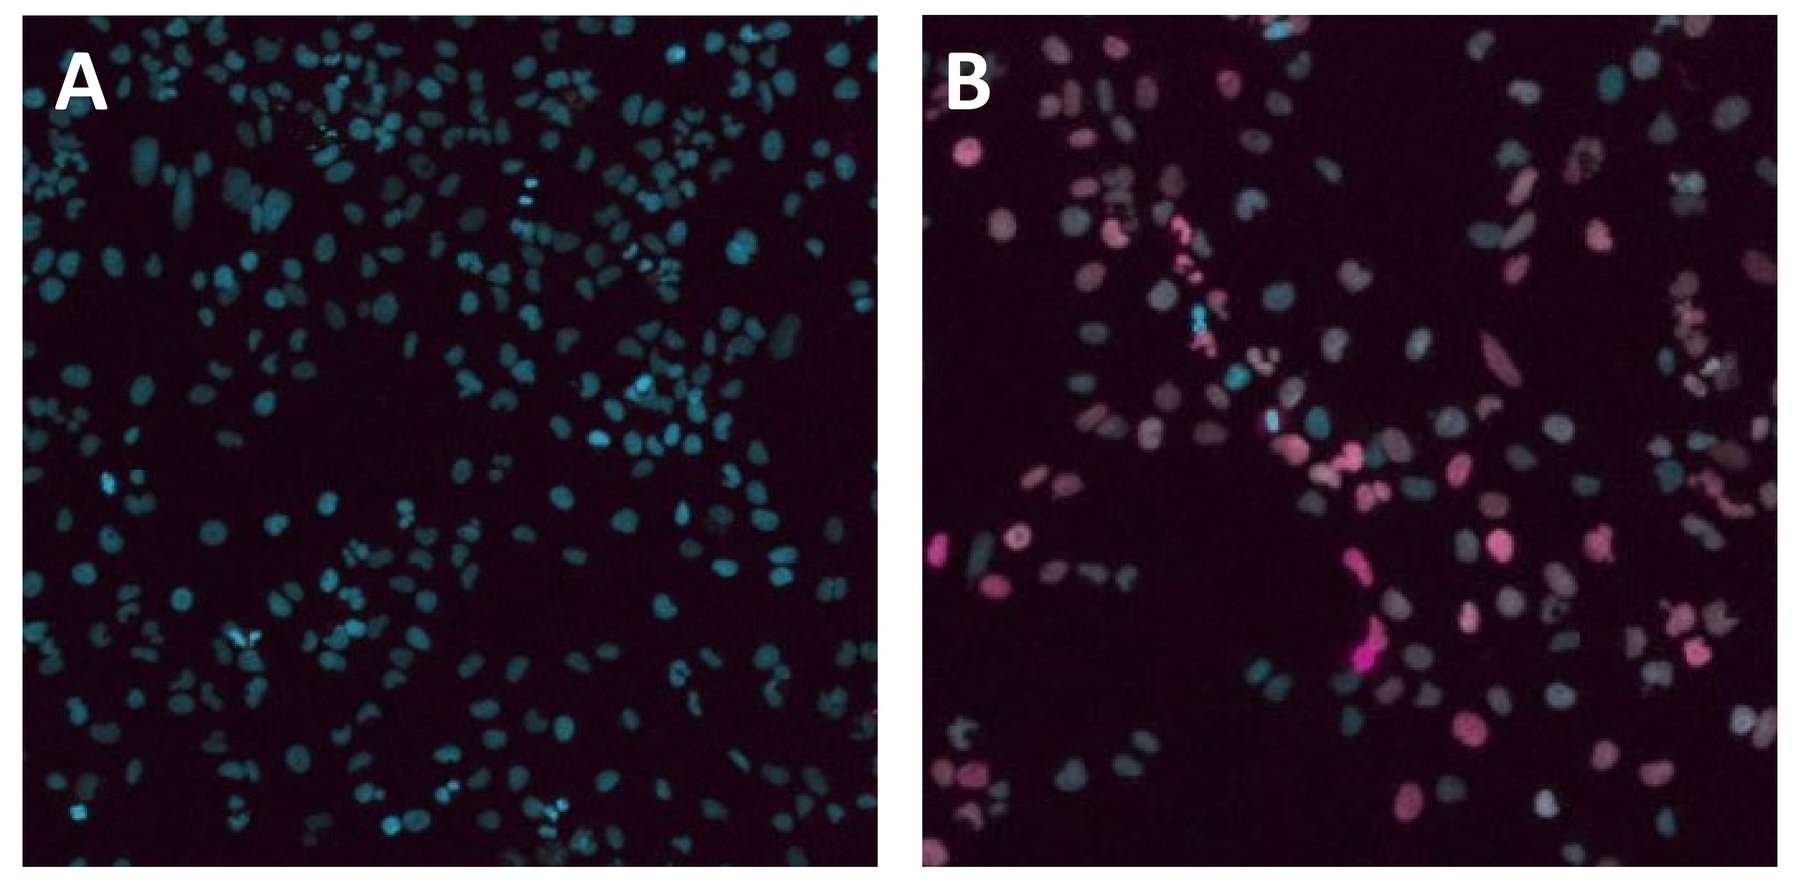
A
B

Figure 4. HNF4α protein expression in LX-2 cells following HNF4α-EC treatment. Representative fluorescence images of LX-2 cells treated with PBS (**A**) or HNF4α-EC (**B**) for 48 hours, followed by immunohistochemistry to detect HNF4α protein expression.

Durable HNF4a upregulation following single dose of EC

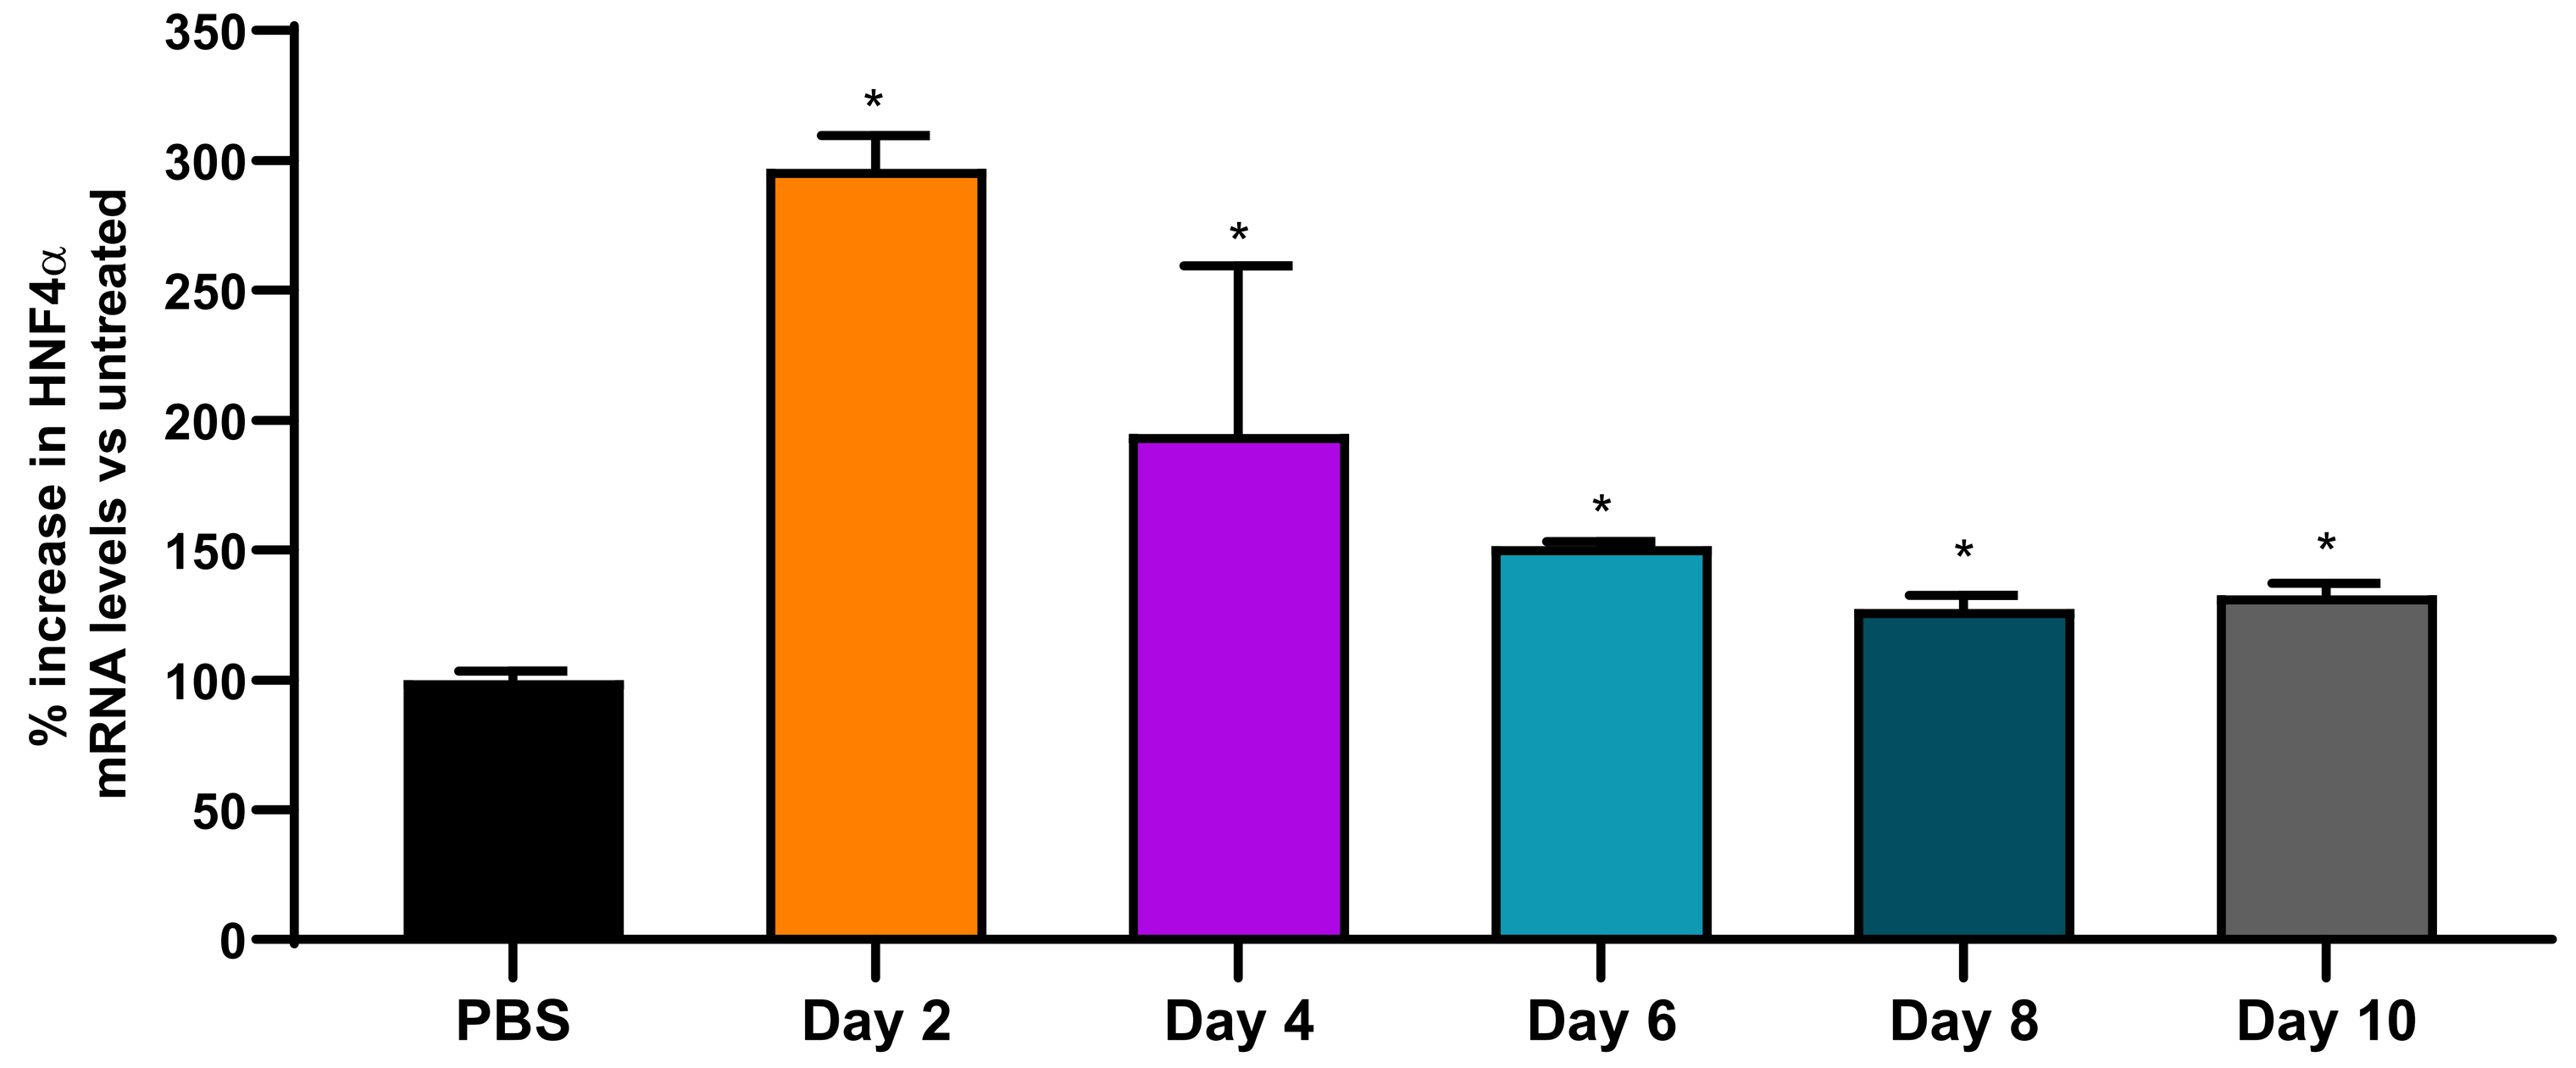
% increase in HNF4α mRNA levels vs untreated
350
300
250
200
150
100
50
0
*
PBS
Day 2
Day 4
Day 6
Day 8
Day 10

Figure 5. Time course of HNF4α mRNA expression levels following HNF4α-EC treatment. Primary human hepatocytes were transfected with HNF4α-EC. HNF4α mRNA levels were measured by qPCR at time points up to 10 days later. HNF4α expression remained above baseline for 8-10 days after transfection. Evidence suggests that inducing the HNF4α transcriptional network for as little as a week is sufficient to initiate a regenerative process, supporting the therapeutic potential for HNF4α-EC treatment. *1-way ANOVA, P<0.05

P1-targeting EC drives upregulation of P1 isoform

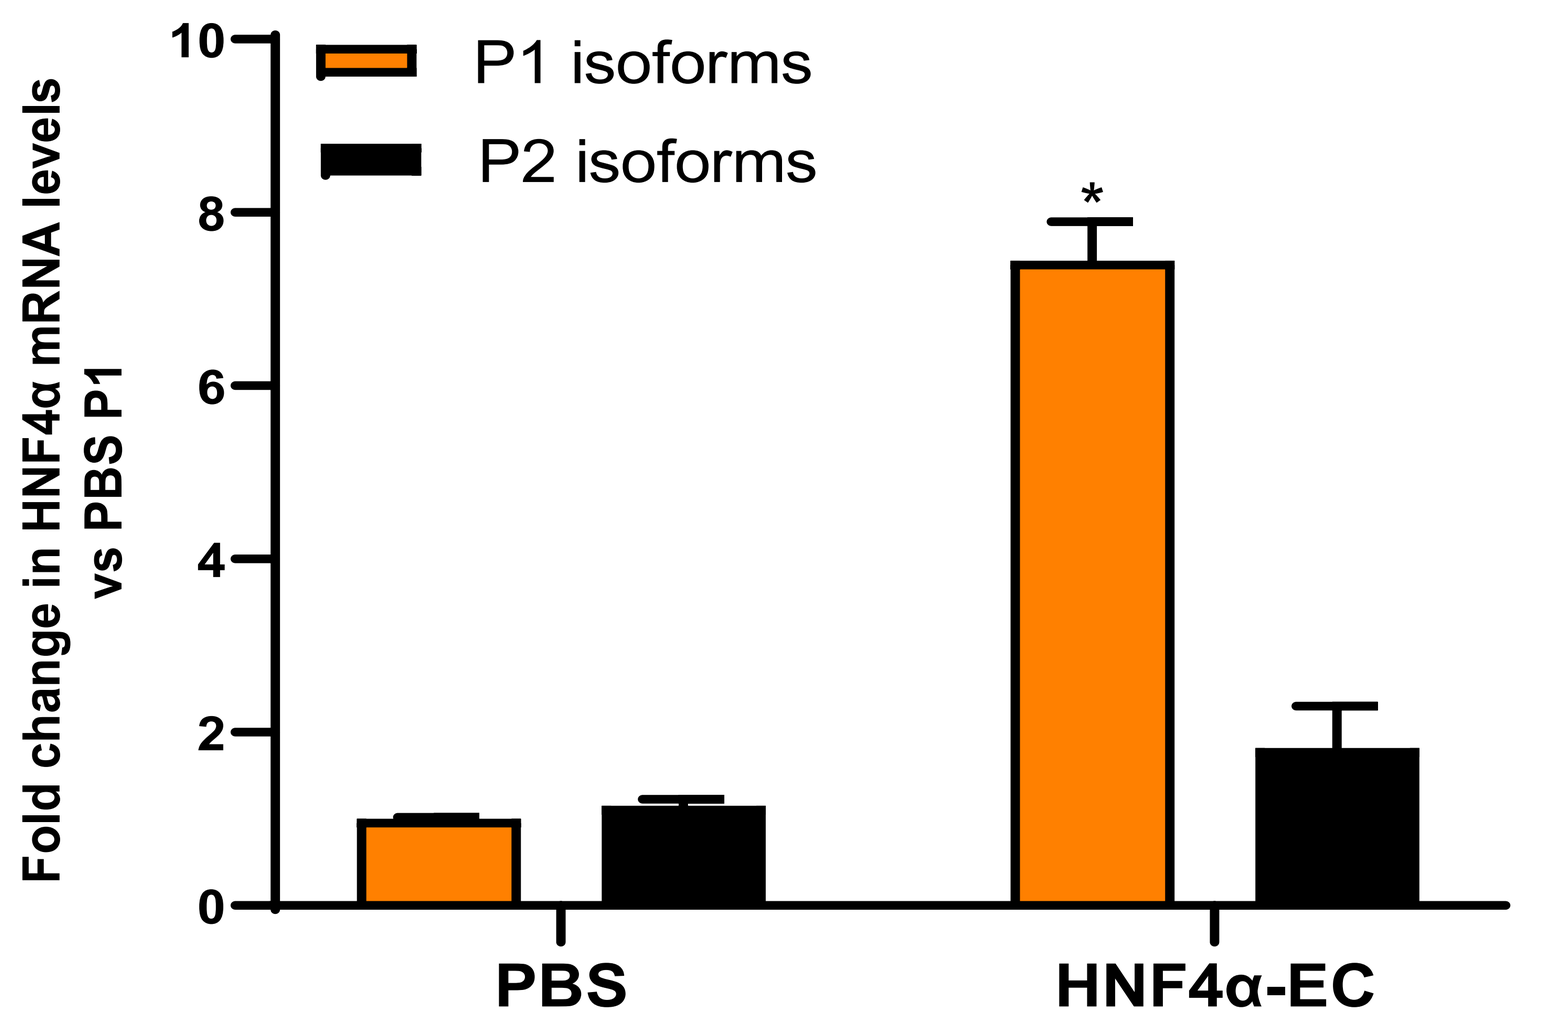
Fold change in HNF4α mRNA levels vs PBS P1
10
8
6
4
2
0
P1 isoforms
P2 isoforms
*
PBS
HNF4α-EC

Figure 6. *In vitro* expression of P1 and P2 isoforms of HNF4α after HNF4α-EC treatment. Primary human hepatocytes were treated with single dose of LNP-encapsulated EC, and qPCR was performed 24 hours later to measure HNF4α expression. *T-test vs PBS , p<0.05

In vivo induction of HNF4α with EC treatment

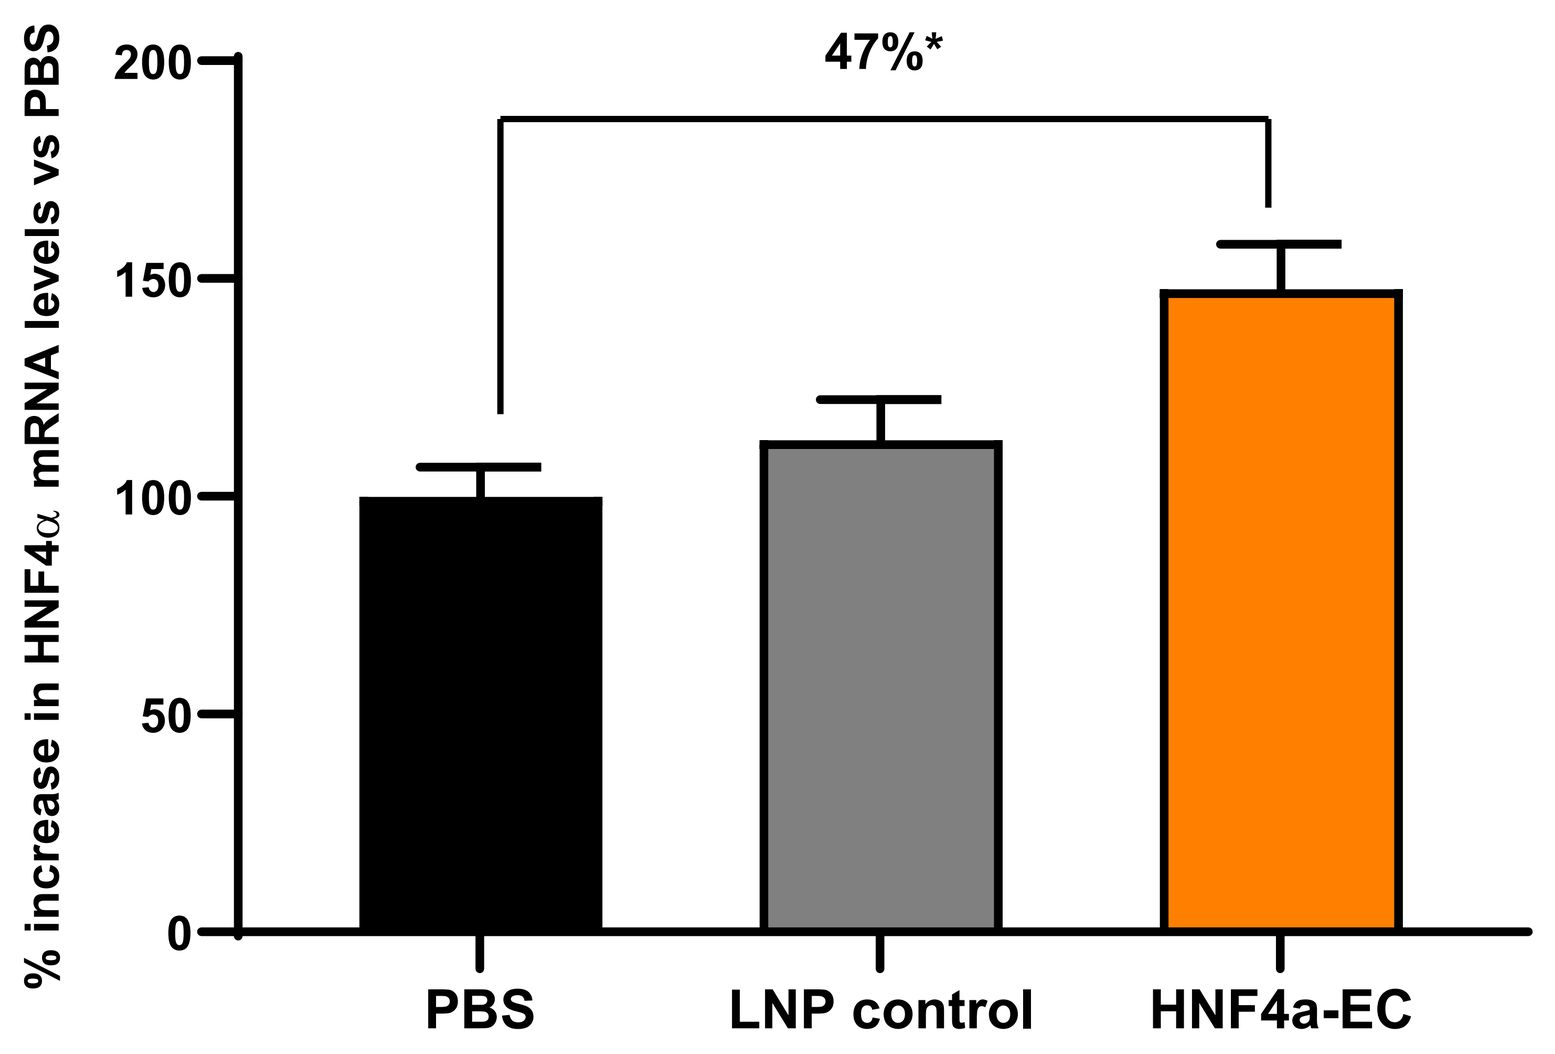
% increase in HNF4α mRNA levels vs PBS
200
150
100
50
0
47%*
PBS
LNP control
HNF4a-EC

Figure 7. HNF4α expression in humanized liver following HNF4α-EC treatment *in vivo*. To measure *in vivo* induction of HNF4α, a single dose of LNP-encapsulated EC was administered intravenously to humanized $Fah^{-/-}/Rag2^{-/-}/Il2rg^{-/-}$ (FRG) mice and qPCR was performed 24 hours later to measure HNF4α expression. *T-test vs PBS, p<0.05

HNF4a upregulation alters fibrotic gene expression *in vitro*

Figure 8. TGFβ1-induced Col1a1 and αSMA gene expression is reduced with HNF4α EC treatment. Hepatic stellate LX-2 cells were treated with 10ng/ml TGFβ1, transfected with LNP-encapsulated HNF4α ECs, and changes in expression of pro-fibrotic markers were measured by qPCR at 24 hours. EC treatment reduced Col1a1 expression by 48% and αSMA expression by 70%, compared to TGFβ1-treatment alone. * T-test vs TGFβ1 , p<0.05

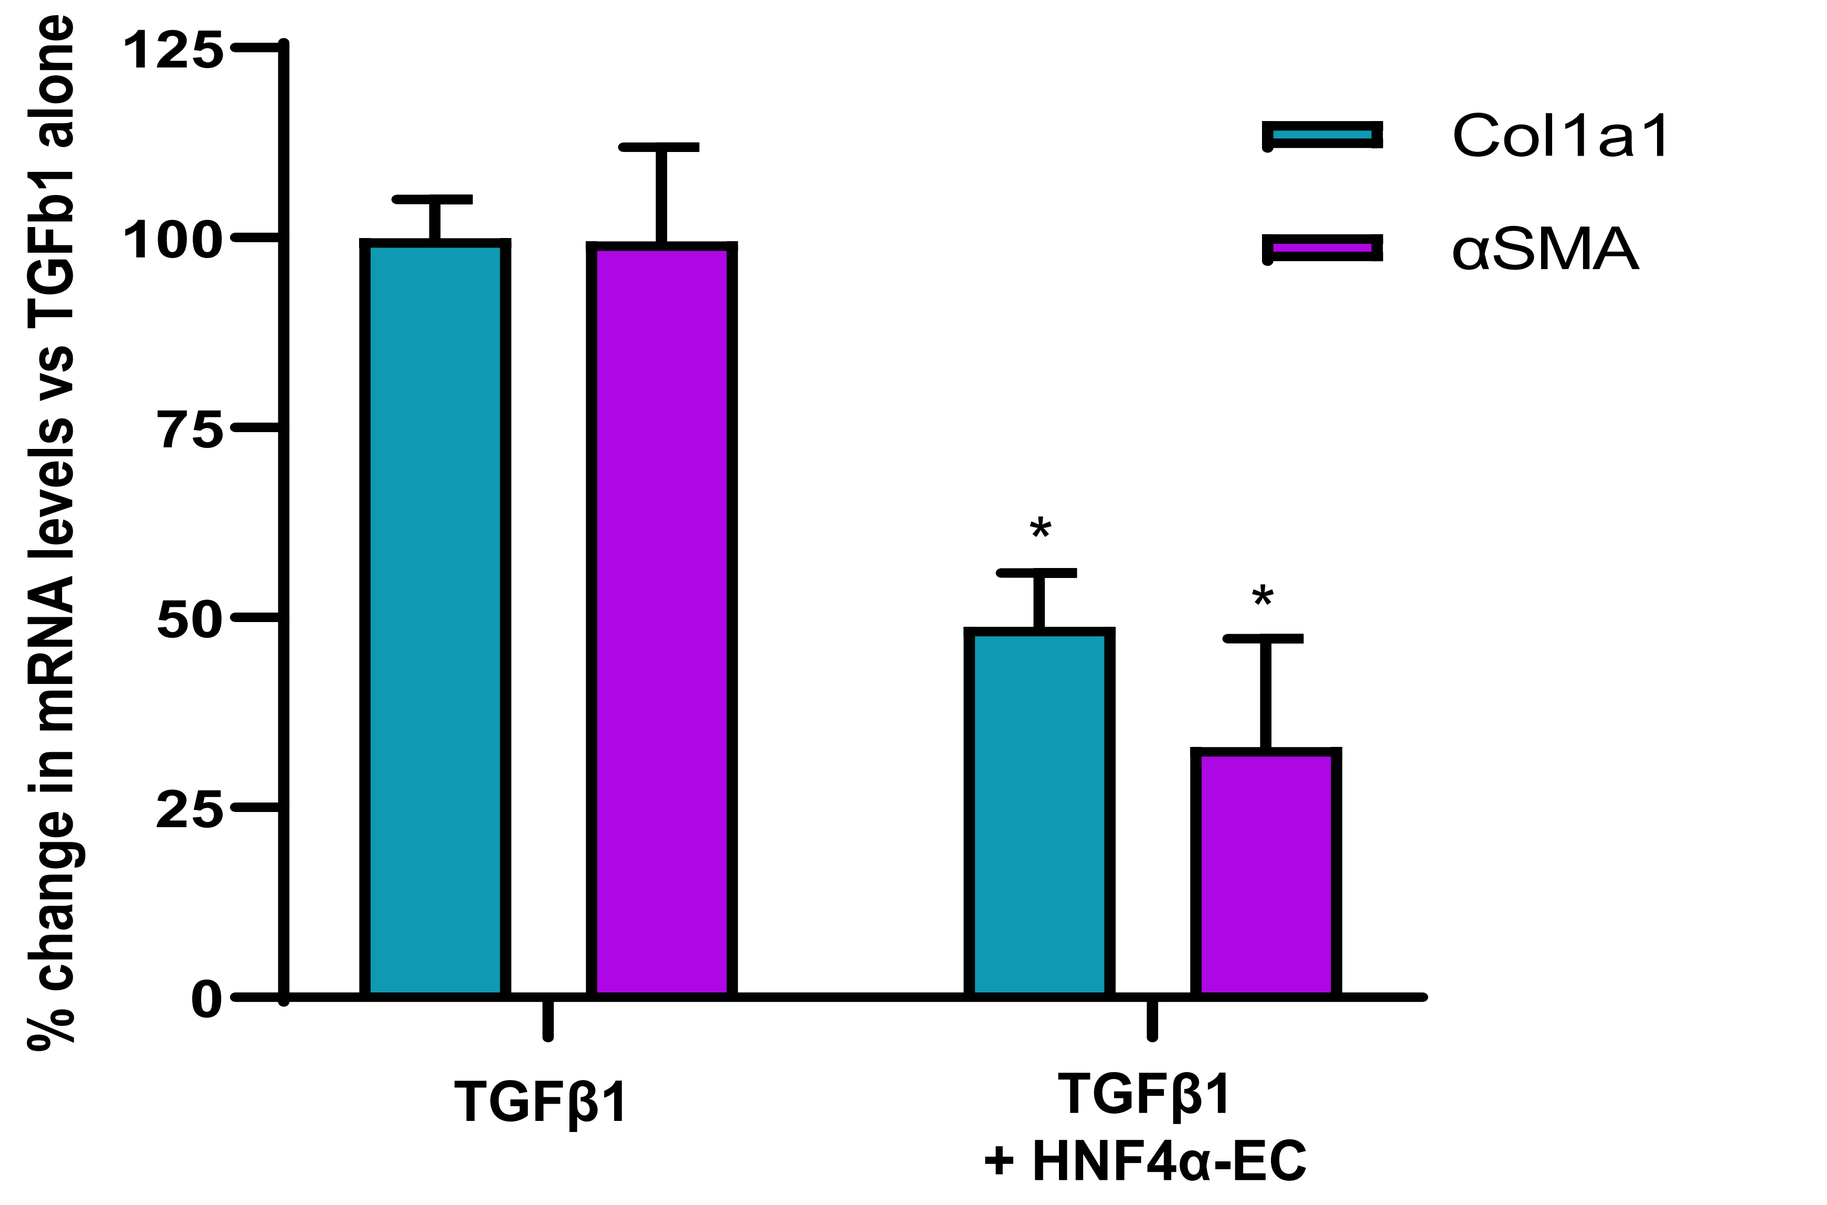
% change in mRNA levels vs TGFb1 alone
125
100
75
50
25
0
Col1a1
αSMA
*
TGFβ1
TGFβ1 + HNF4α-EC

HNF4α induction confers protection in DDC fibrosis model

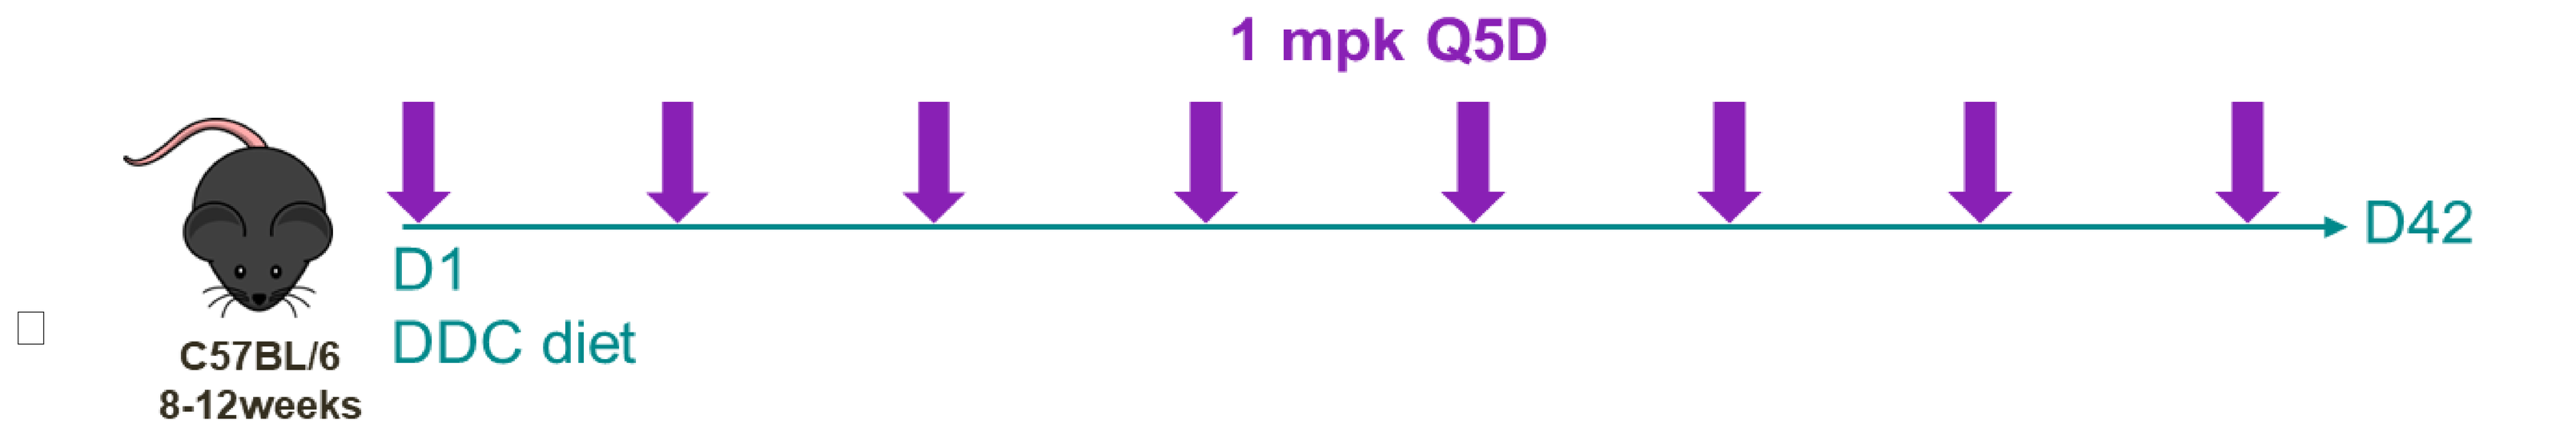
1 mpk Q5D
C57BL/6
8-12weeks
D1
DDC diet
D42

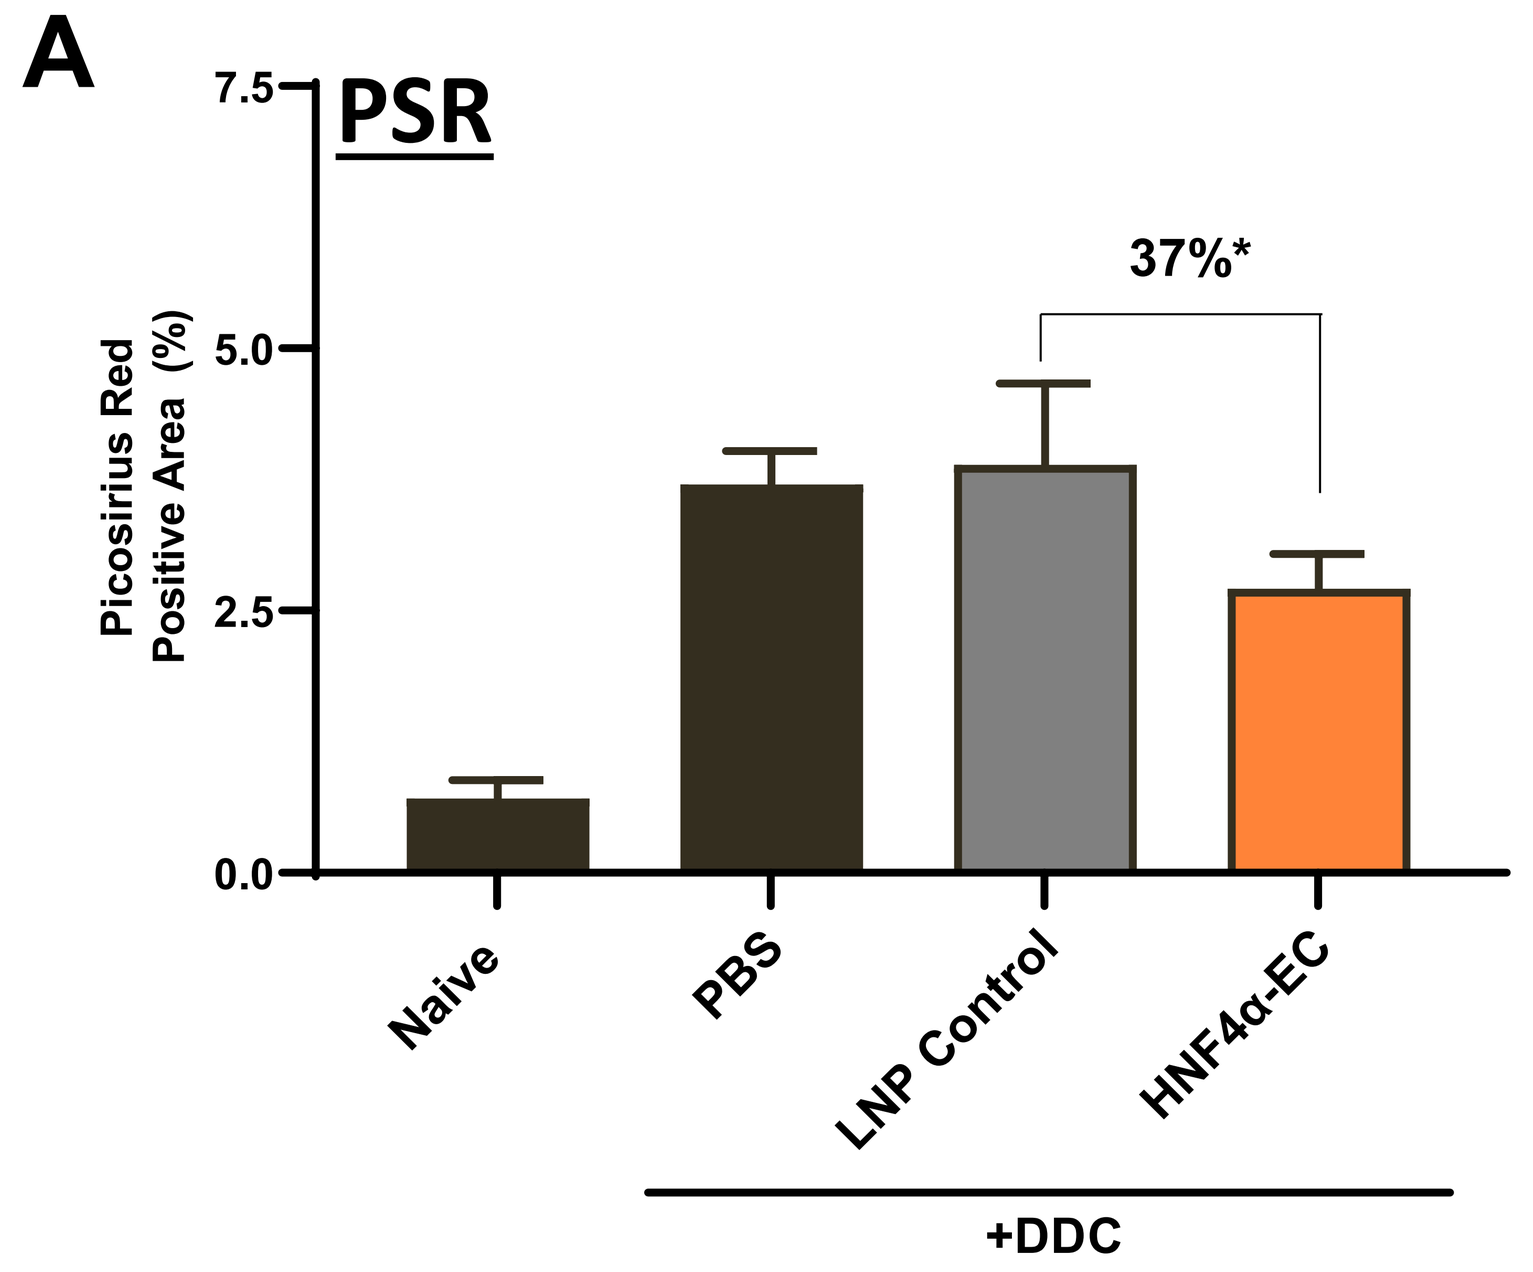
A
PSR
Picosirius Red Positive Area (%)
7.5
5.0
2.5
0.0
37%*
Naive
PBS
LNP Control
HNF4α-EC
+DDC

Figure 9. HNF4α-EC treatment upregulates HNF4α expression and reduces fibrosis in DDC mouse model. Mice fed the 3,5-diethoxycarbonyl-1,4-dihydrocollidine (DDC) diet develop liver injury with fibrosis. In our study, mice were placed on DDC diet and administered 1mpk murine HNF4α-EC Q5d for 42 days. (**A**) Picosirius red staining was performed on section of DDC mouse livers and staining was quantified to measure deposited collagen. (**B**) Hydroxyproline assay was conducted to measure collagen content of DDC mouse livers. The mRNA levels of αSMA (*acta2*) (**C**), HNF4α (**D**), and the HNF4α target gene, HNF1α (**E**) were measured in DDC mouse livers by qPCR. *1-way ANOVA, P<0.05

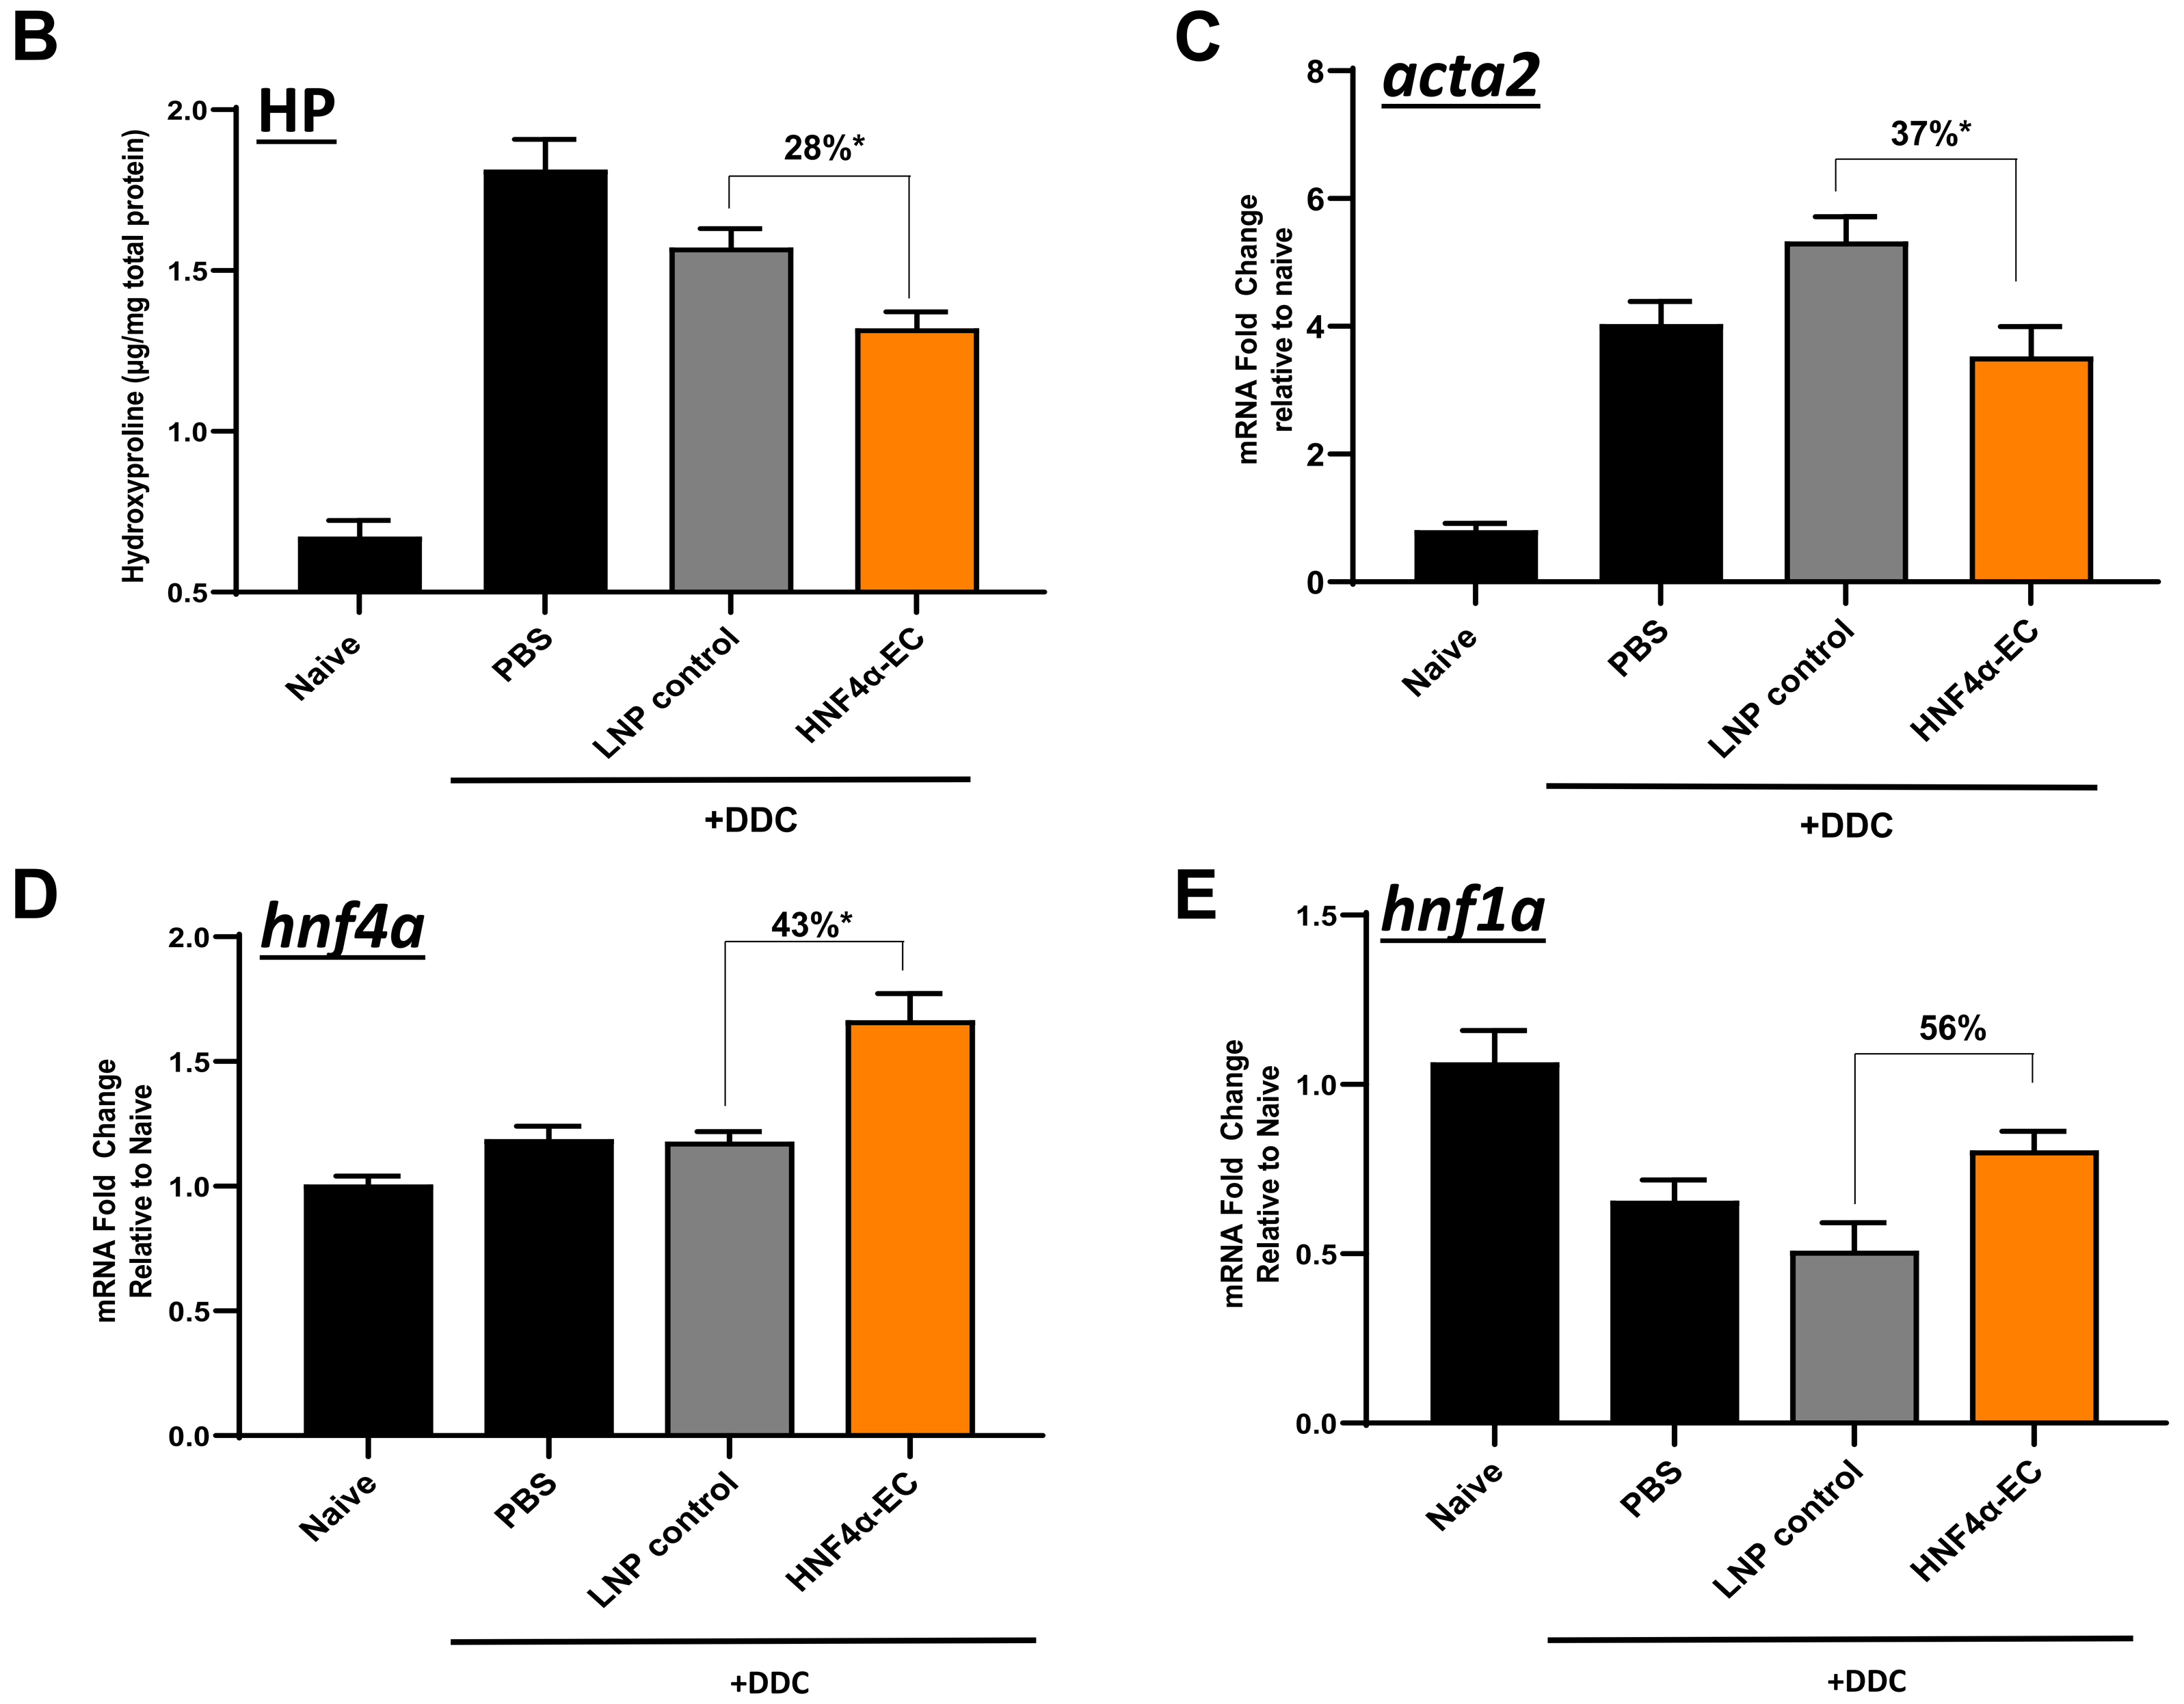
B
HP
Hydroxyproline (µg/mg total protein)
28%*
C
acta2
mRNA Fold Change relative to naive
37%*
D
hnf4a
mRNA Fold Change Relative to Naive
43%*
E
hnf1a
mRNA Fold Change Relative to Naive
56%
Naive
PBS
LNP control
HNF4α-EC
+DDC

Summary and Conclusions

We demonstrate the ability of an Epigenomic Controller (EC) to durably increase HNF4α expression both *in vitro* and *in vivo*. In addition, this modulation preferentially induces the upregulation HNF4α P1 promoter isoforms, resulting in inhibition of fibrotic responses *in vitro*. Significant upregulation of HNF4α is observed following EC treatment in a humanized liver mouse model. We then tested a murine HNF4α-EC in the DDC fibrosis model and saw increased HNF4α mRNA levels, increased levels of a downstream target of HNF4α, and a decreased fibrotic signature, supporting the therapeutic potential of epigenetic targeting of HNF4α in fibrotic liver disease. (Omega thanks Acuitas for the preparation of LNPs utilized in the *in vivo* expression and efficacy studies.)

Contact Information

Omega Therapeutics | 140 First Street | Cambridge MA 02141 | amccurley@omegatx.com