Controlled epigenomic upregulation of CXCL9 and CXCL10 in hepatocellular carcinoma promotes T-cell recruitment

Eugine Lee, Maddy Pacaro, Houda Belaghzal, Cameron Vergato, Daniel F. G. Costa, Pranjali Rumale, Snigdha Kosana, Sam Mildrum, Steve Siecinski, Defne Yarar, Graeme Hodgson

Omega Therapeutics, Cambridge, MA, USA

2024 SITC ANNUAL MEETING
Poster number: 1360

Introduction

Despite improved therapeutic outcomes for cancer patients with the advent of immunotherapy, not all patients respond to these agents. Some patients exhibit an 'immune-cold' or 'excluded' phenotype, with little to no infiltration of T cells into the tumor body. Lack of T-cell infiltration may result from an absence of T-cell recruiting chemokines in the tumor and its surrounding environment, such as CXCR3 ligands CXCL9 and CXCL10. Recent pan-cancer analyses have shown a strong positive correlation between levels of CXCL9 and CXCL10 in tumors and the response to checkpoint inhibitors (CPIs). Moreover, studies in animal models have demonstrated that increasing the levels of CXCL9 and/or CXCL10 can enhance anti-tumor activity, with or without concurrent CPI treatment. Here we explore the utility of epigenomic controllers to tunably upregulate CXCL9 and CXCL10 expression in hepatocellular carcinoma (HCC) and tissue resident cells (hepatocytes) as a potential strategy to create an immunologically-hot HCC tumor microenvironment (TME).

We have developed epigenomic controllers (ECs), which are programmable epigenomic mRNA therapeutics, that are designed to selectively target regulatory elements of target genes such as CXCL9 and CXCL10 and upregulate their expression. Our initial screening of CXCL9- and CXCL10-ECs encoding for various epigenetic activators revealed a broad spectrum of upregulation, demonstrating the broad upregulation capability of our platform. Treatment of human primary hepatocytes with CXCL9- and CXCL10-ECs resulted in the desired epigenetic modifications, leading to robust upregulation of CXCL9 and CXCL10 mRNAs and proteins that induced *in vitro* T-cell migration. Murine surrogate mCXCL10-EC was developed, and *in vitro* tests demonstrated effective upregulation of mouse cxcl10 expression in mouse HCC cells and primary hepatocytes.

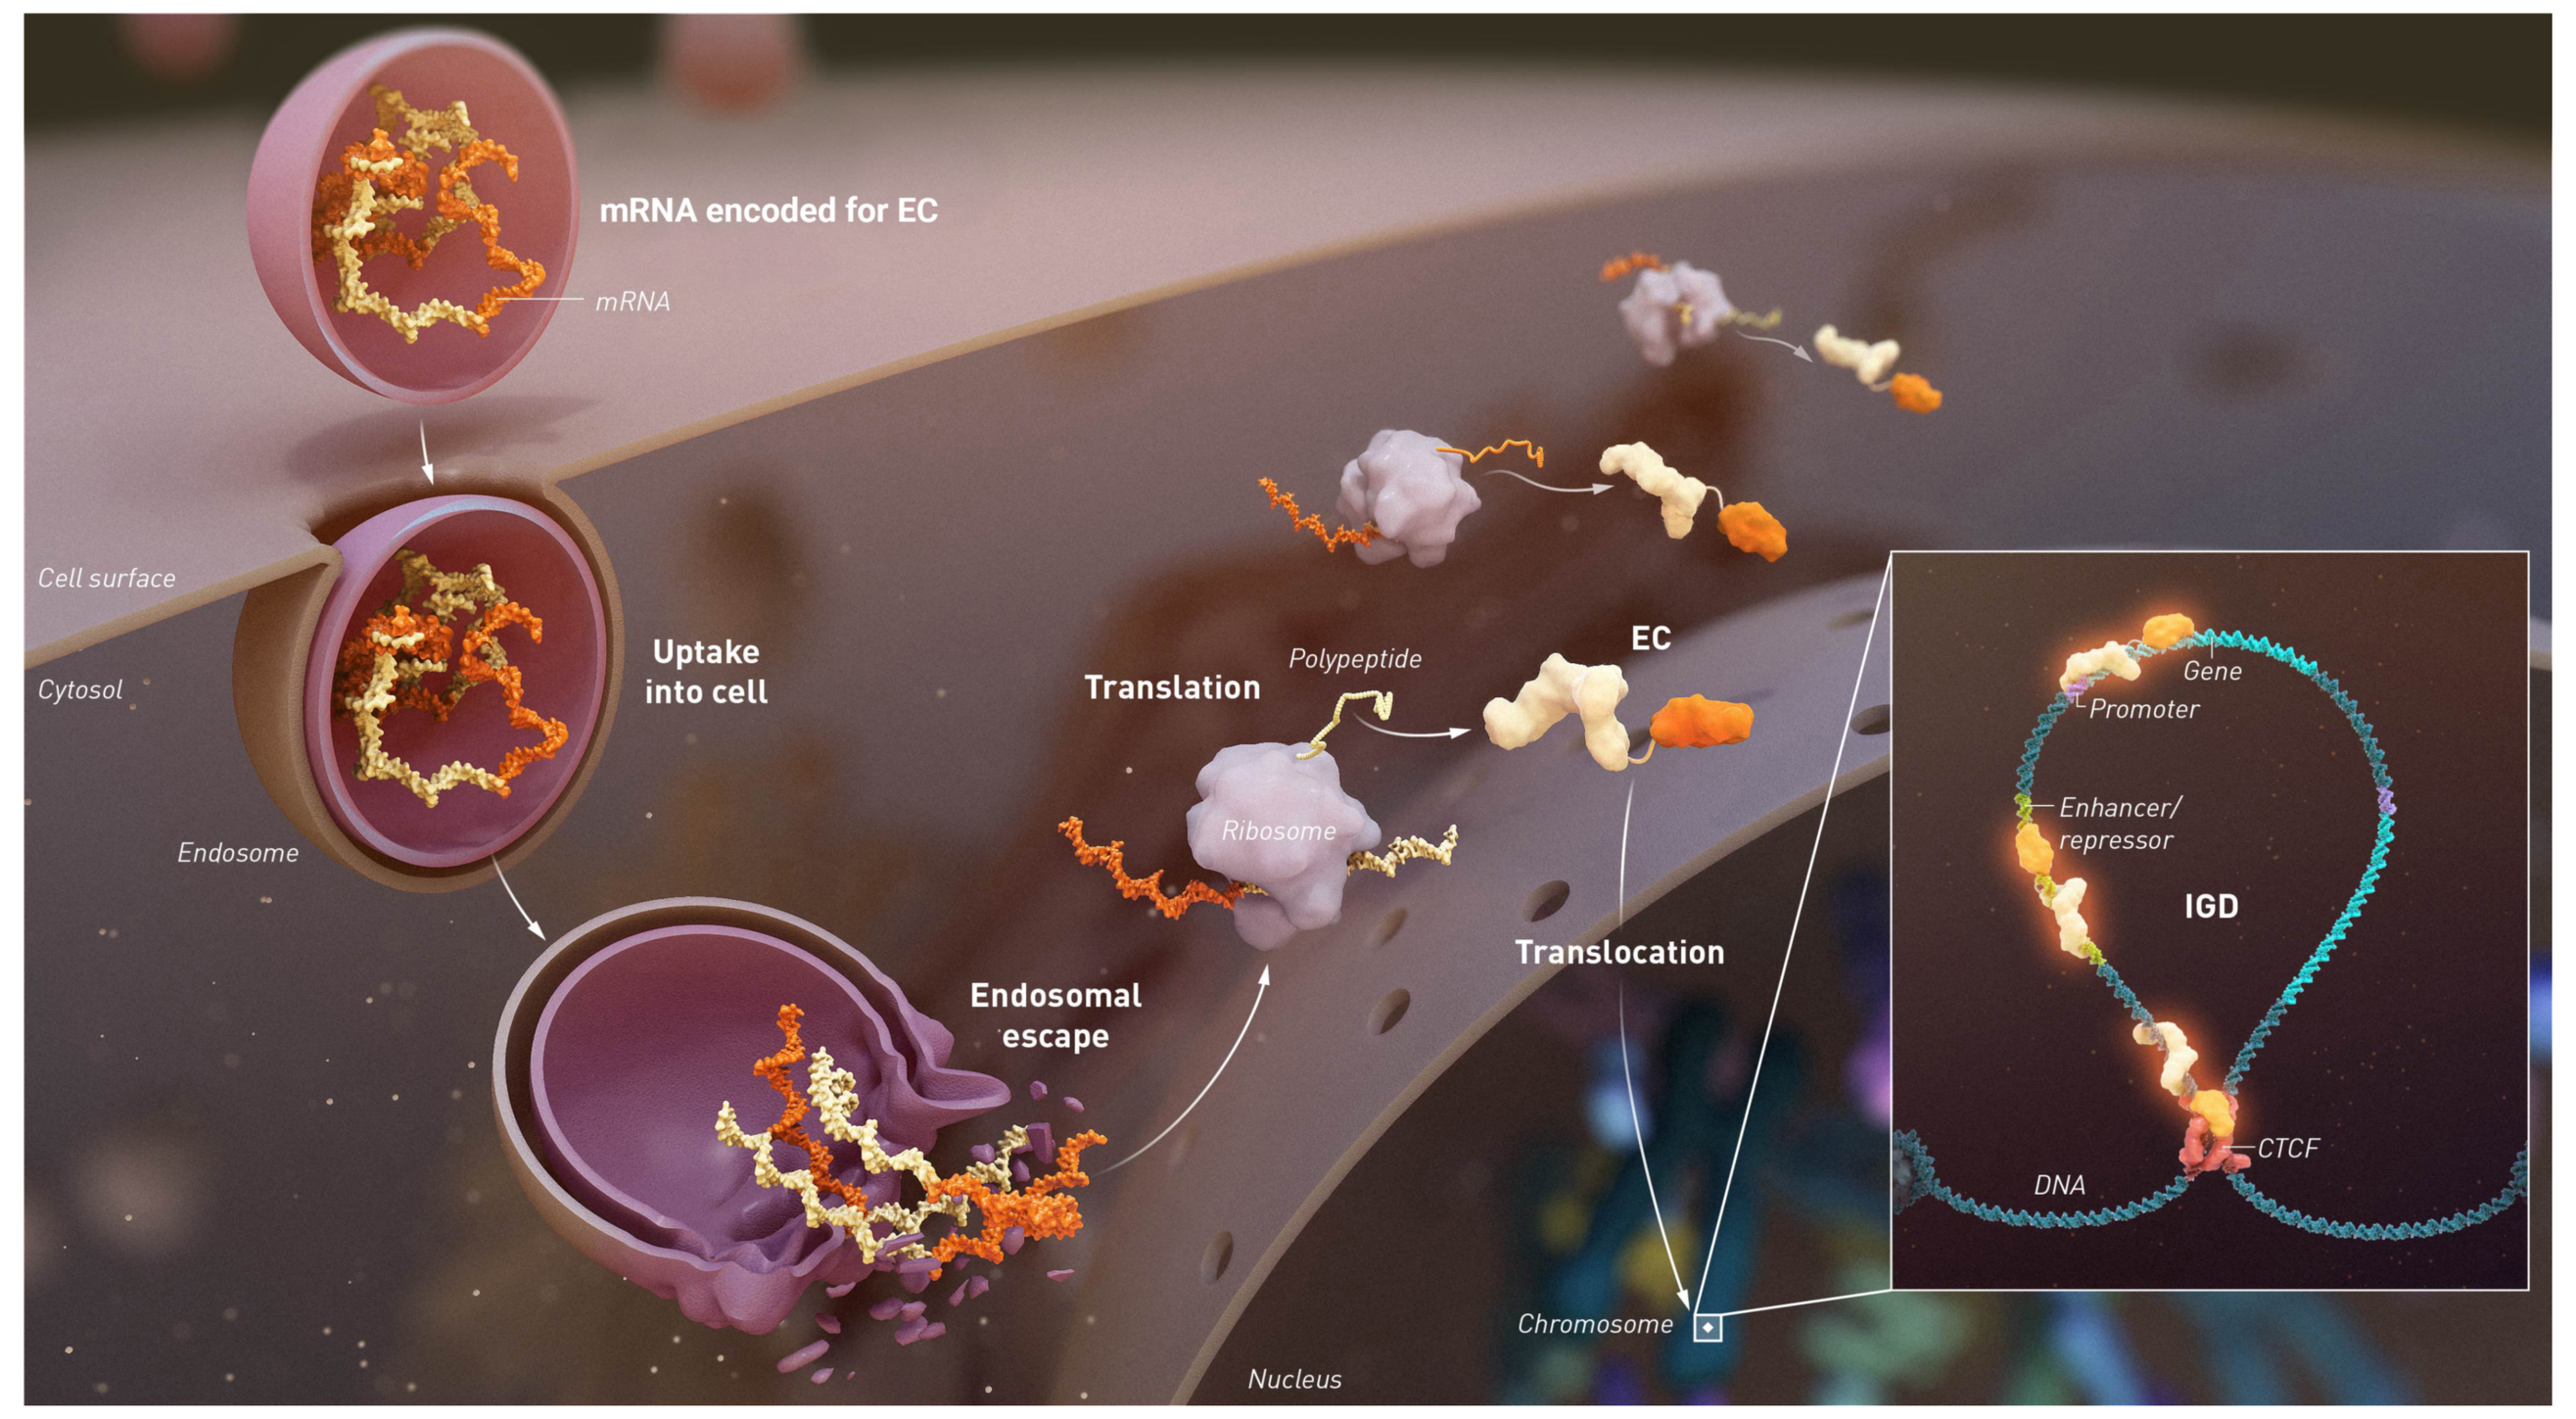
mRNA encoded for EC
mRNA
Cell surface
Cytosol
Uptake into cell
Endosome
Endosomal escape
Translation
Polypeptide
Ribosome
EC
Translocation
Gene
Promoter
Enhancer/ repressor
IGD
CTCF
DNA
Chromosome
Nucleus

Figure 1. Structure (right) and Mechanism of Action (left) of ECs

Treatment with mCXCL10-EC inhibited tumor growth in syngeneic mouse model of HCC and promoted the recruitment of CD8+ T-cells into tumors, providing in vivo proof-of-concept for this approach.

Screening of CXCL9- and CXCL10-ECs with various epigenomic activators reveals a broad spectrum of upregulation, demonstrating the tunability of Omega's platform

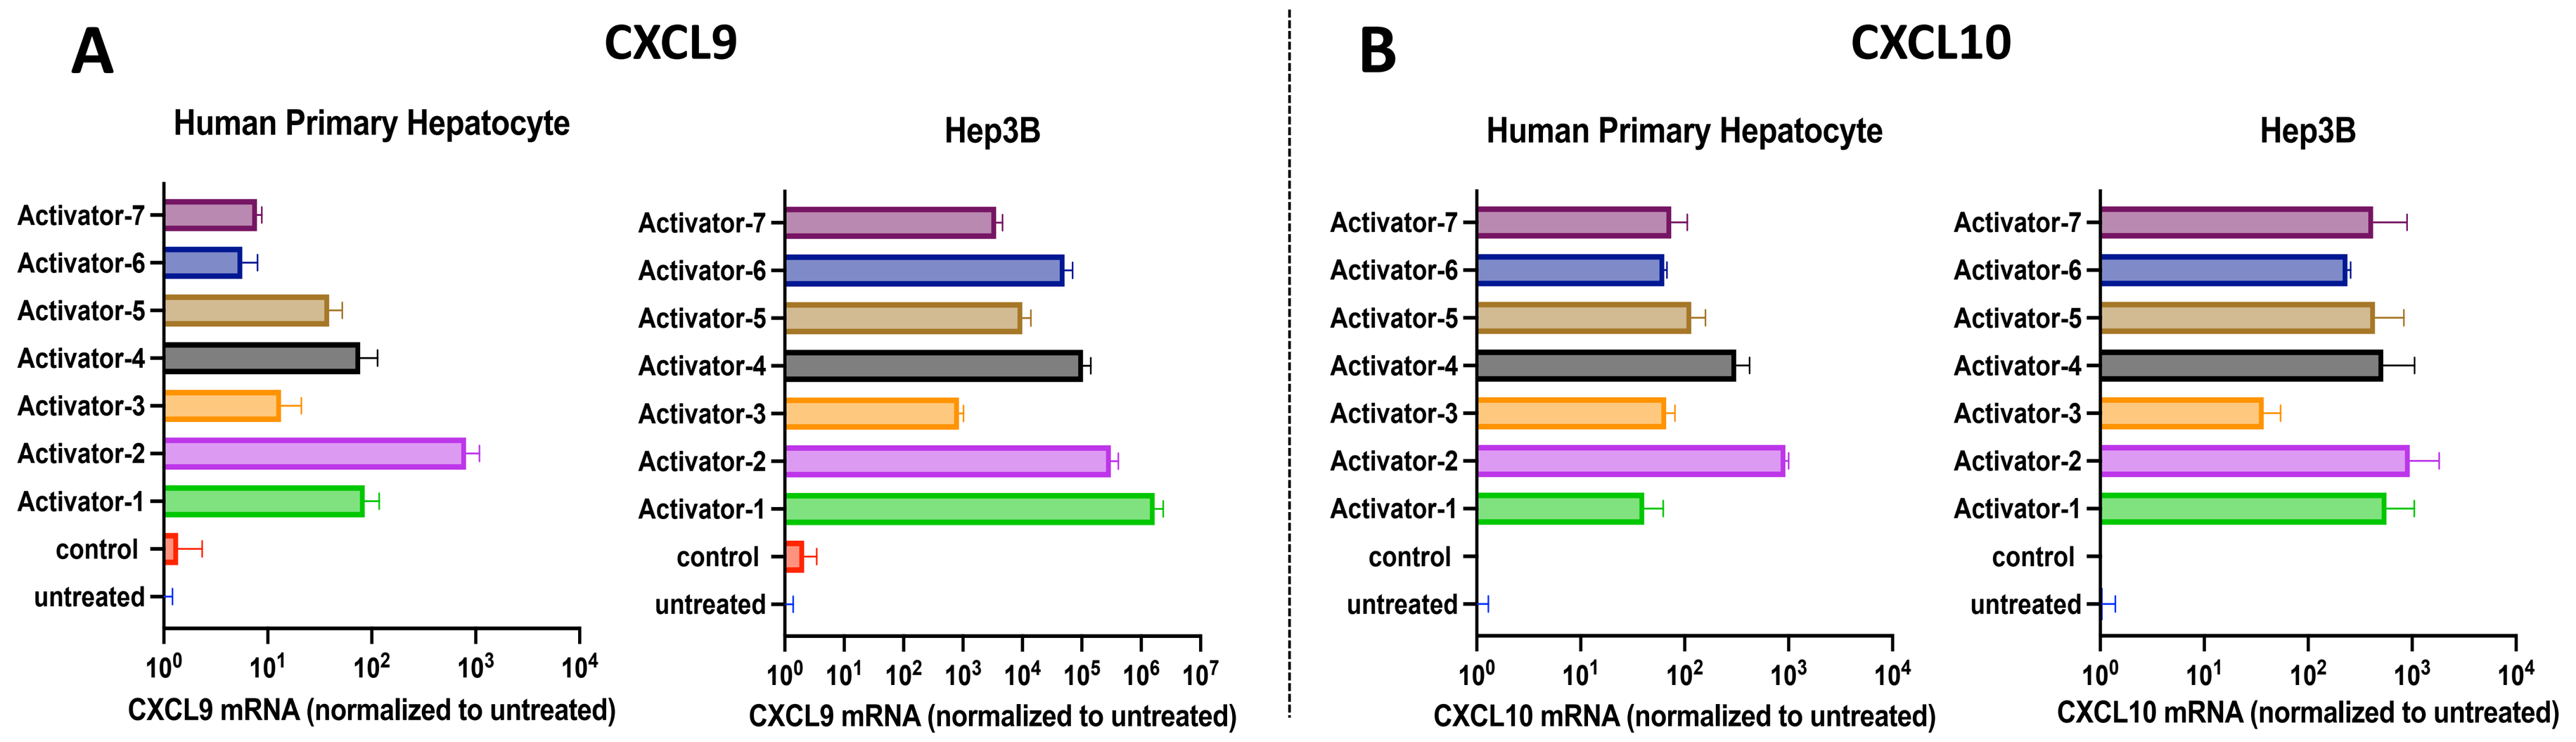
A
CXCL9
Human Primary Hepatocyte
Hep3B
Activator-7
Activator-6
Activator-5
Activator-4
Activator-3
Activator-2
Activator-1
control
untreated
CXCL9 mRNA (normalized to untreated)
B
CXCL10
CXCL10 mRNA (normalized to untreated)

Figure 2. Treatment of human primary hepatocytes or Hep3B cells with CXCL9- or CXCL10-ECs encoding different epigenomic activators shows wide range of CXCL9 or CXCL10 upregulation, respectively, ranging from 10- to 10^6-fold. Cells were treated with PBS (untreated), 1.8 μg/ml of control mRNA, or 1.8 μg/ml of CXCL9-ECs (A) or CXCL10-ECs (B) for 48 hours and were subjected to RT-qPCR. CXCL9- and CXCL10-ECs with Activator-2 were selected for further assessment.

Treatment of human primary hepatocytes with CXCL9- or CXCL10-EC induces robust upregulation of CXCL9 or CXCL10 expression, respectively

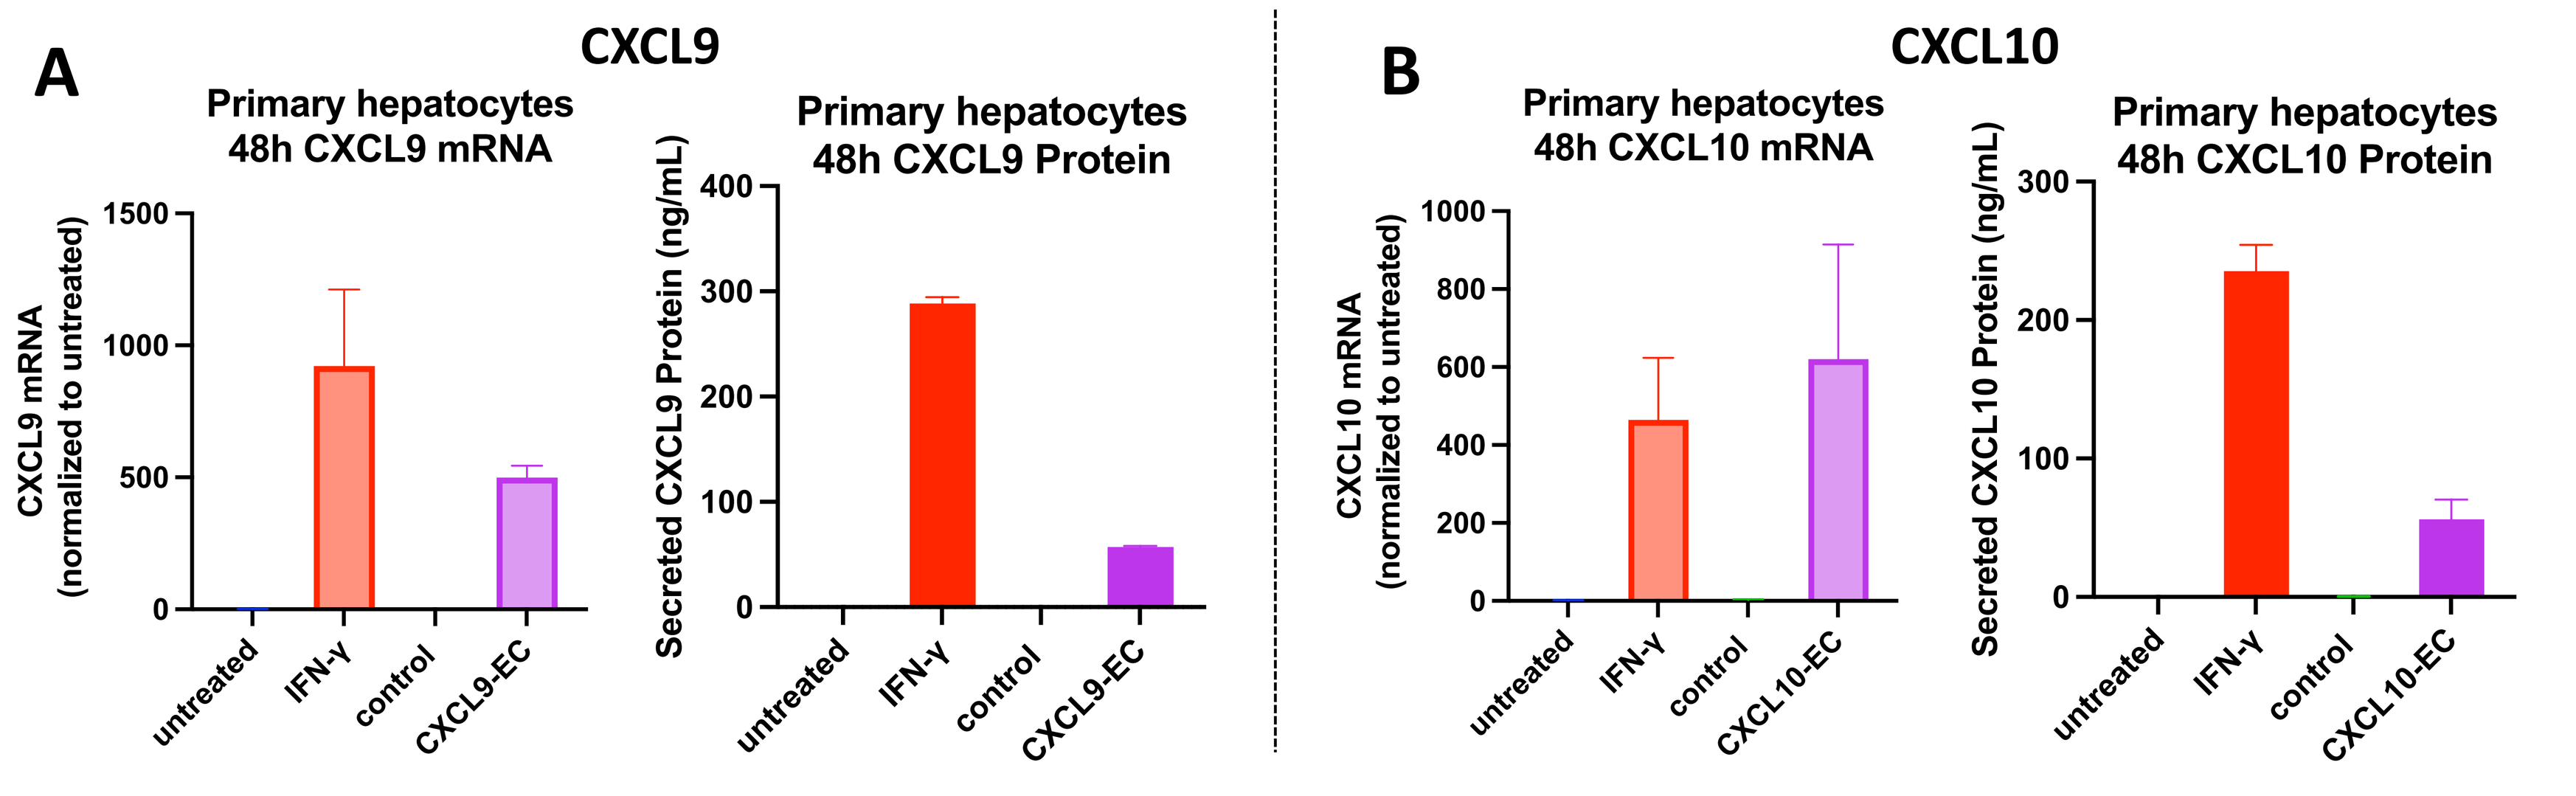
A
CXCL9
Primary hepatocytes 48h CXCL9 mRNA
CXCL9 mRNA (normalized to untreated)
Primary hepatocytes 48h CXCL9 Protein
Secreted CXCL9 Protein (ng/mL)
untreated
IFN-γ
control
CXCL9-EC
B
CXCL10
Primary hepatocytes 48h CXCL10 mRNA
CXCL10 mRNA (normalized to untreated)
Primary hepatocytes 48h CXCL10 Protein
Secreted CXCL10 Protein (ng/mL)
CXCL10-EC

Figure 3: Treatment of human primary hepatocytes with CXCL9- or CXCL10-EC upregulates CXCL9 or CXCL10 expression, respectively, to a level comparable to that induced by IFN-γ treatment. Cells were treated with PBS (untreated), 5 ng/ml of IFN-γ, 1.8 μg/ml of control mRNA, or 1.8 μg/ml of CXCL9-EC (A) or CXCL10-EC (B) for 48 hours and were subjected RT-qPCR. Supernatant was collected to quantify secreted CXCL9 and CXCL10 protein levels by ELISA.

Treatment of human primary hepatocytes with CXCL9- or CXCL10-EC induces the expected epigenetic modifications on their target sites

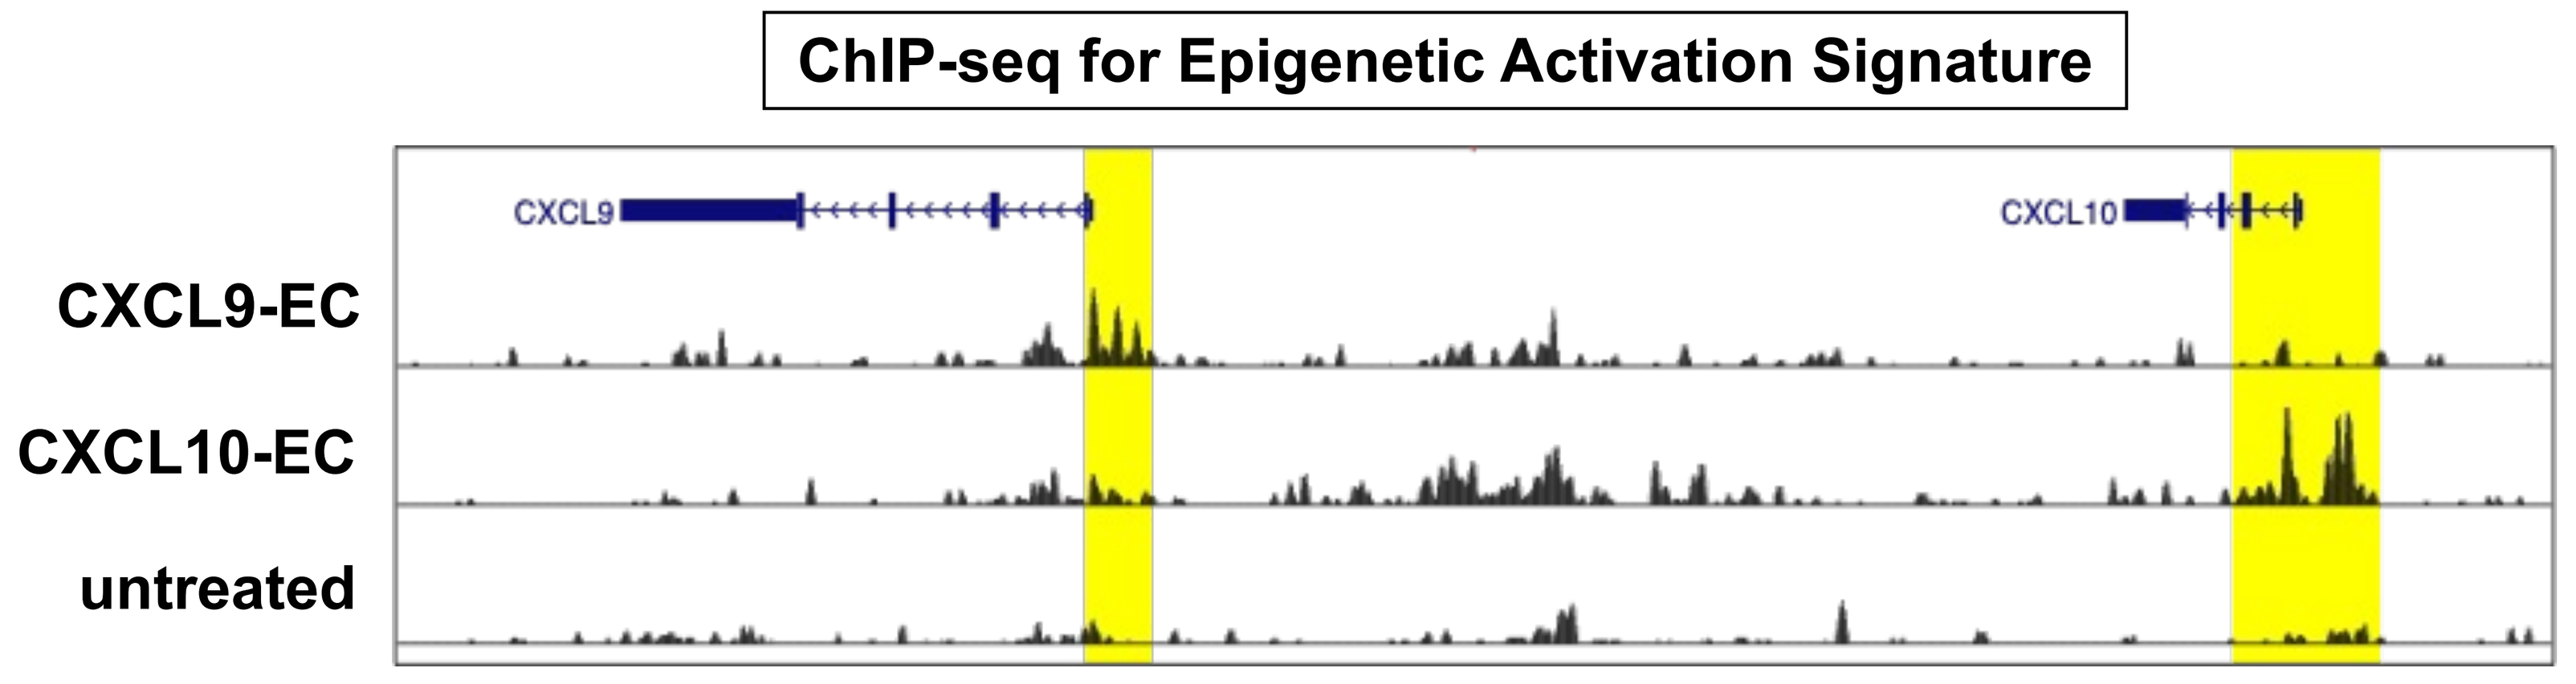
ChIP-seq for Epigenetic Activation Signature
CXCL9
CXCL10
CXCL9-EC
CXCL10-EC
untreated

Figure 4: Treatment of human primary hepatocytes with CXCL9- or CXCL10-EC increases the level of epigenetic activation signature on CXCL9 or CXCL10 transcription start site, respectively. Cells were treated with PBS (untreated), or 1.8 μg/ml of CXCL9-EC or CXCL10-EC for 48 hours and were subjected to ChIP-sequencing.

EC-mediated upregulation of CXCL9 or CXCL10 promotes T-cell recruitment *in vitro*

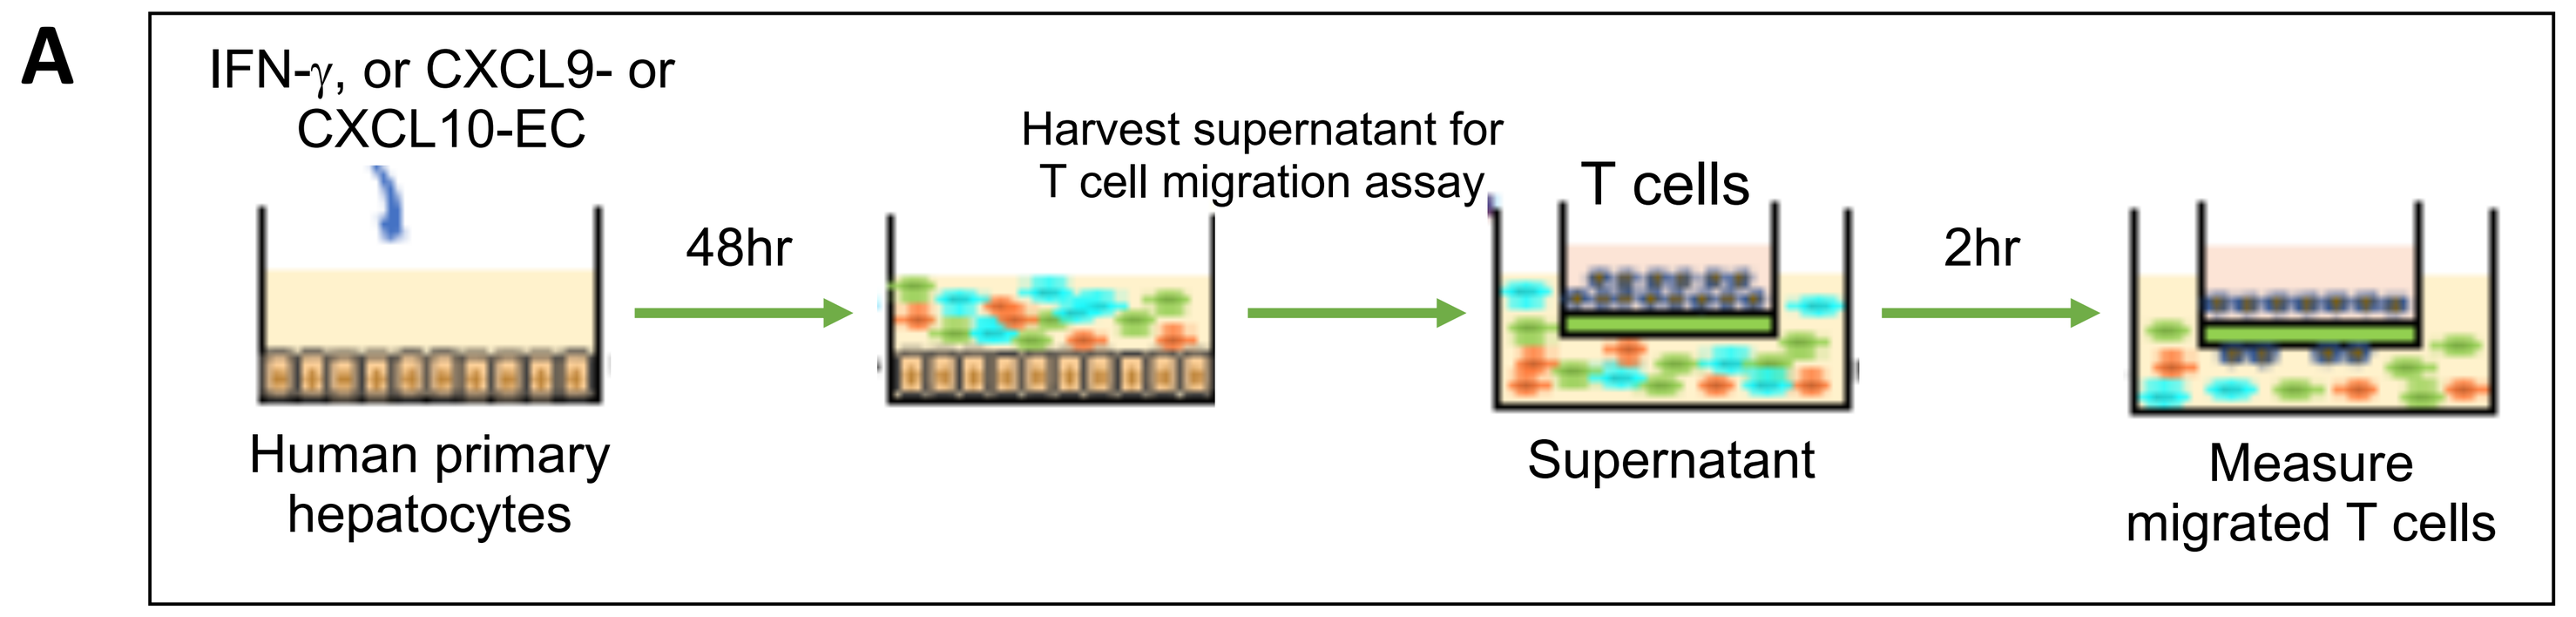
A
IFN-γ, or CXCL9- or CXCL10-EC
48hr
Harvest supernatant for T cell migration assay
T cells
2hr
Human primary hepatocytes
Supernatant
Measure migrated T cells

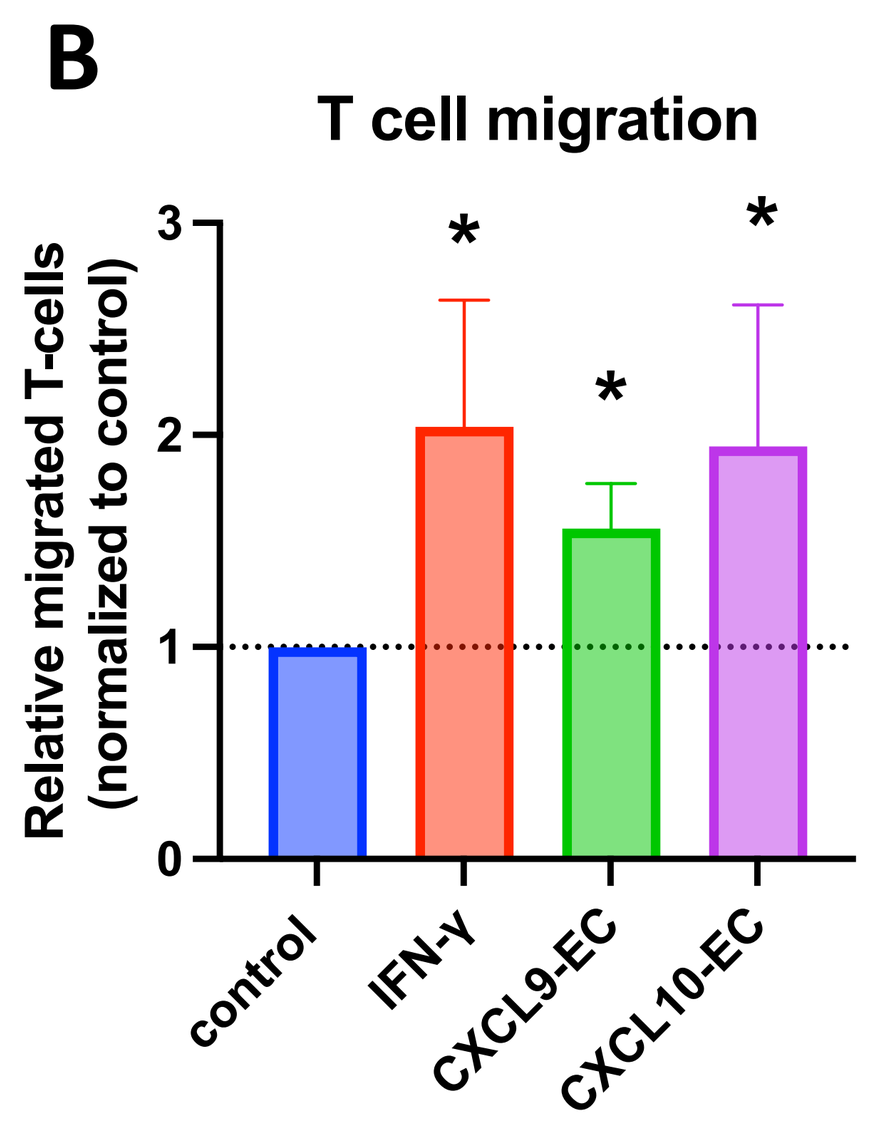
B
T cell migration
Relative migrated T-cells (normalized to control)
control
IFN-γ
CXCL9-EC
CXCL10-EC

Figure 5: Upregulation of CXCL9 or CXCL10 in human primary hepatocytes by CXCL9- or CXCL10-EC treatment induces T-cell migration *in vitro*. (A) Supernatant from IFN-γ or EC-treated primary human hepatocytes are used as the chemoattractant for activated T-cells to migrate across a Boyden chamber. Representative illustration of assay shown. (B) Supernatant from 1.8 μg/ml of CXCL9- or CXCL10-EC treated primary human hepatocytes promoted T-cell migration to a level comparable to that induced by 5 ng/ml IFN-γ, when compared to 1.8 μg/ml of control mRNA treatment. Significant differences between control and selected groups were analyzed using one-way ANOVA. (*) $p < 0.05$.

Treatment of mouse HCC cells or primary hepatocytes with mouse surrogate mCXCL10-EC induces robust upregulation of mouse cxcl10 expression

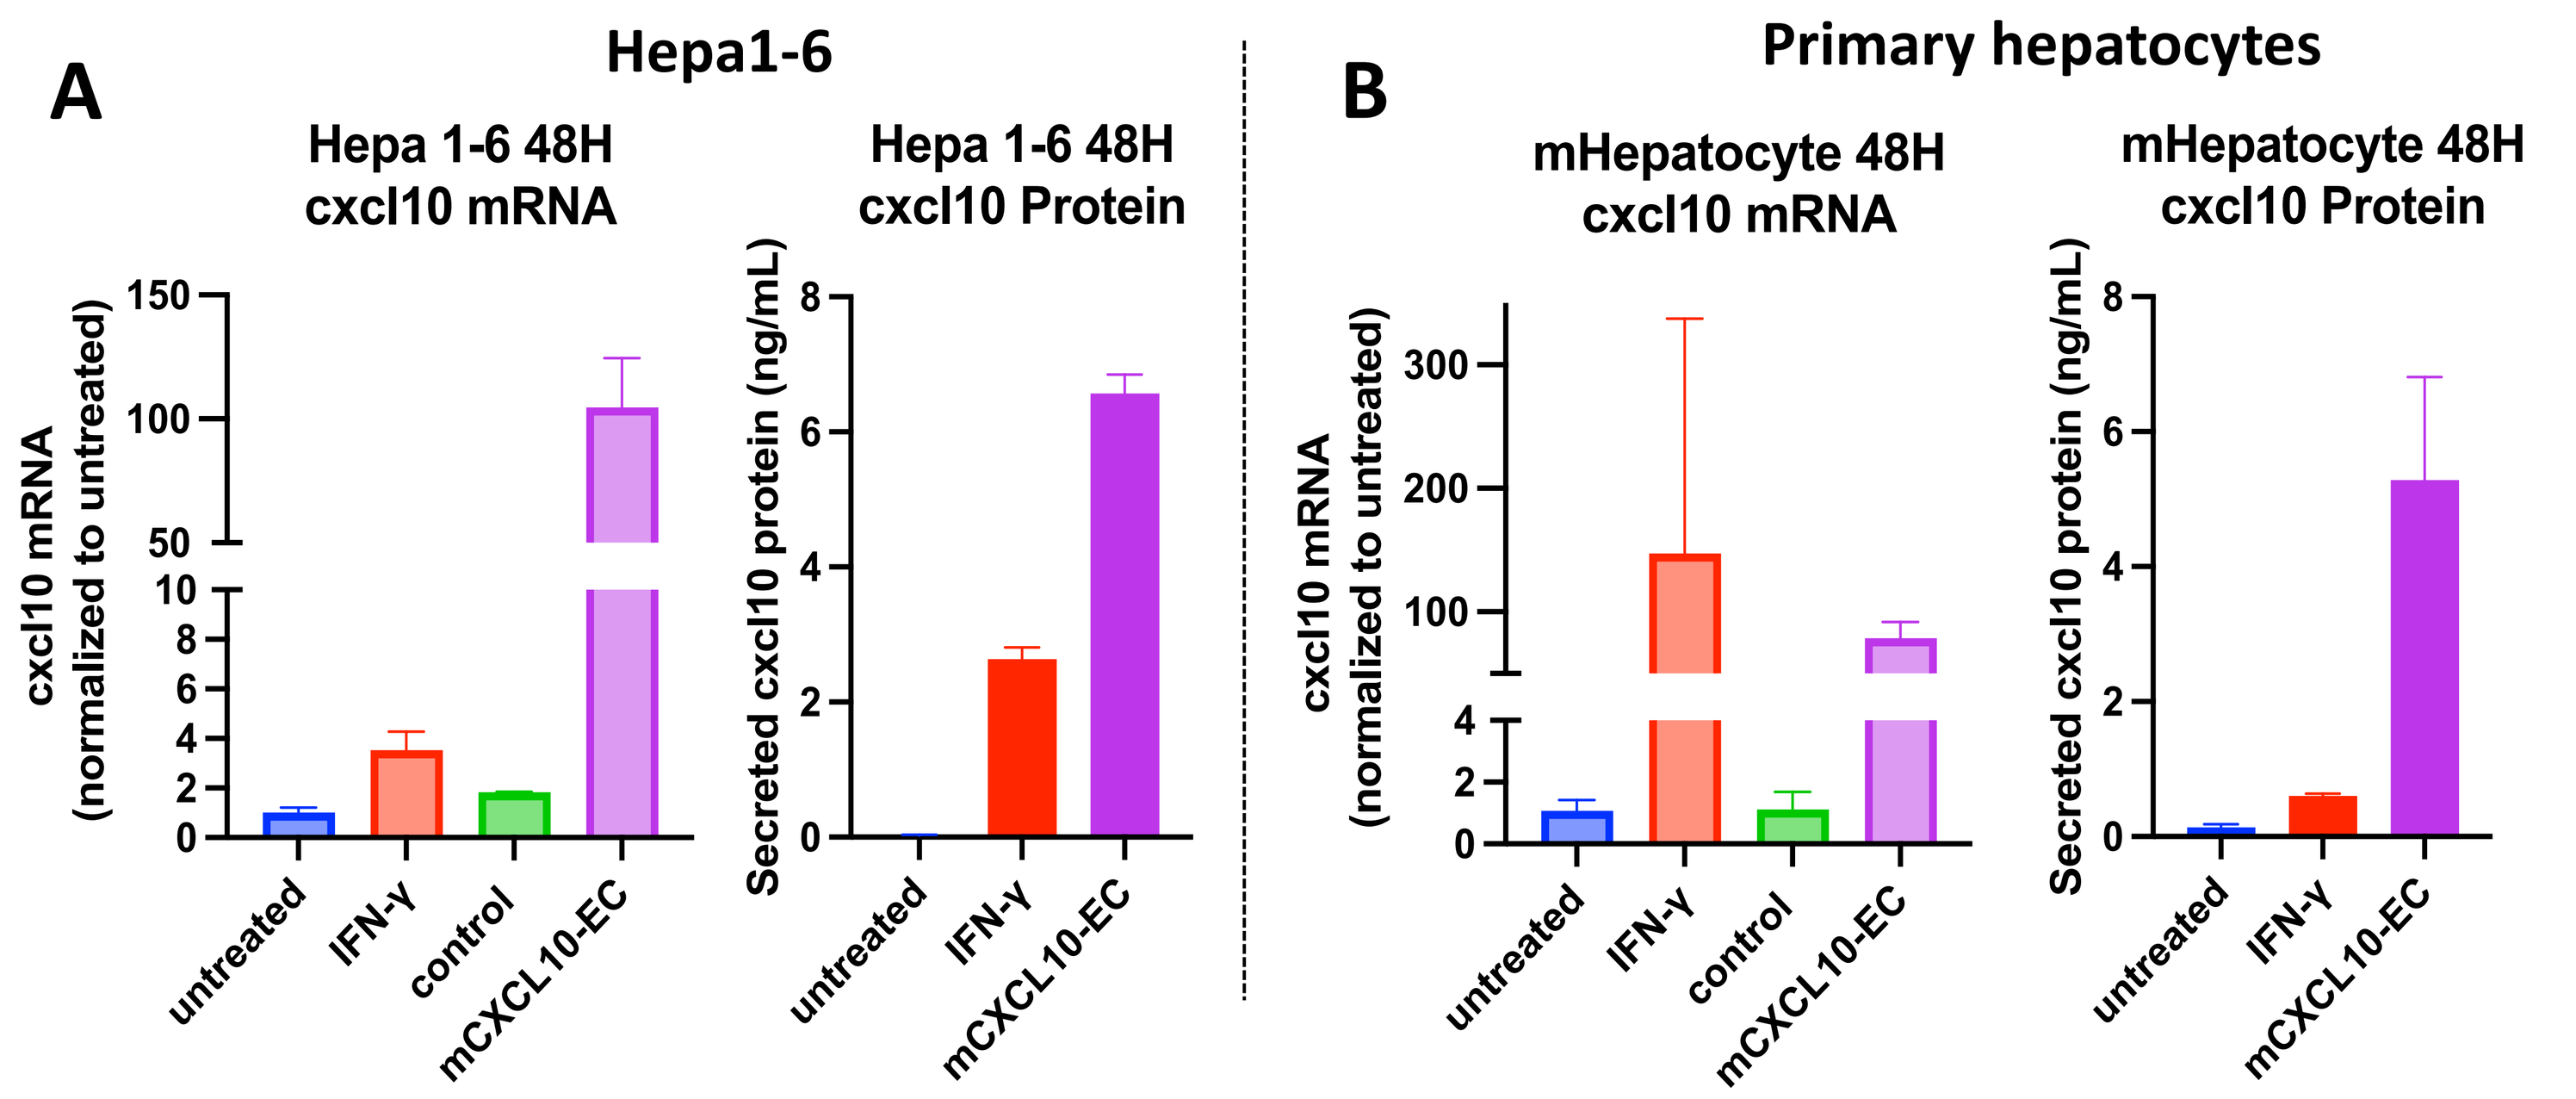
A
Hepa1-6
Hepa 1-6 48H cxcl10 mRNA
cxcl10 mRNA (normalized to untreated)
Hepa 1-6 48H cxcl10 Protein
Secreted cxcl10 protein (ng/mL)
B
Primary hepatocytes
mHepatocyte 48H cxcl10 mRNA
mHepatocyte 48H cxcl10 Protein
untreated
IFN-γ
control
mCXCL10-EC

Figure 6: Treatment of Hepa1-6 cells (A) or mouse primary hepatocytes (B) with mCXCL10-EC upregulates cxcl10 expression. Cells were treated with PBS (untreated), 50 ng/ml of mouse IFN-γ, or 1.8 μg/ml of control mRNA or mCXCL10-EC for 48 hours and were subjected to RT-qPCR. Supernatant was collected to quantify secreted cxcl10 protein levels by ELISA.

Mouse surrogate mCXCL10-EC reduces tumor growth and enhances CD8+ T-cell recruitment in a syngeneic mouse model of HCC *in vivo*

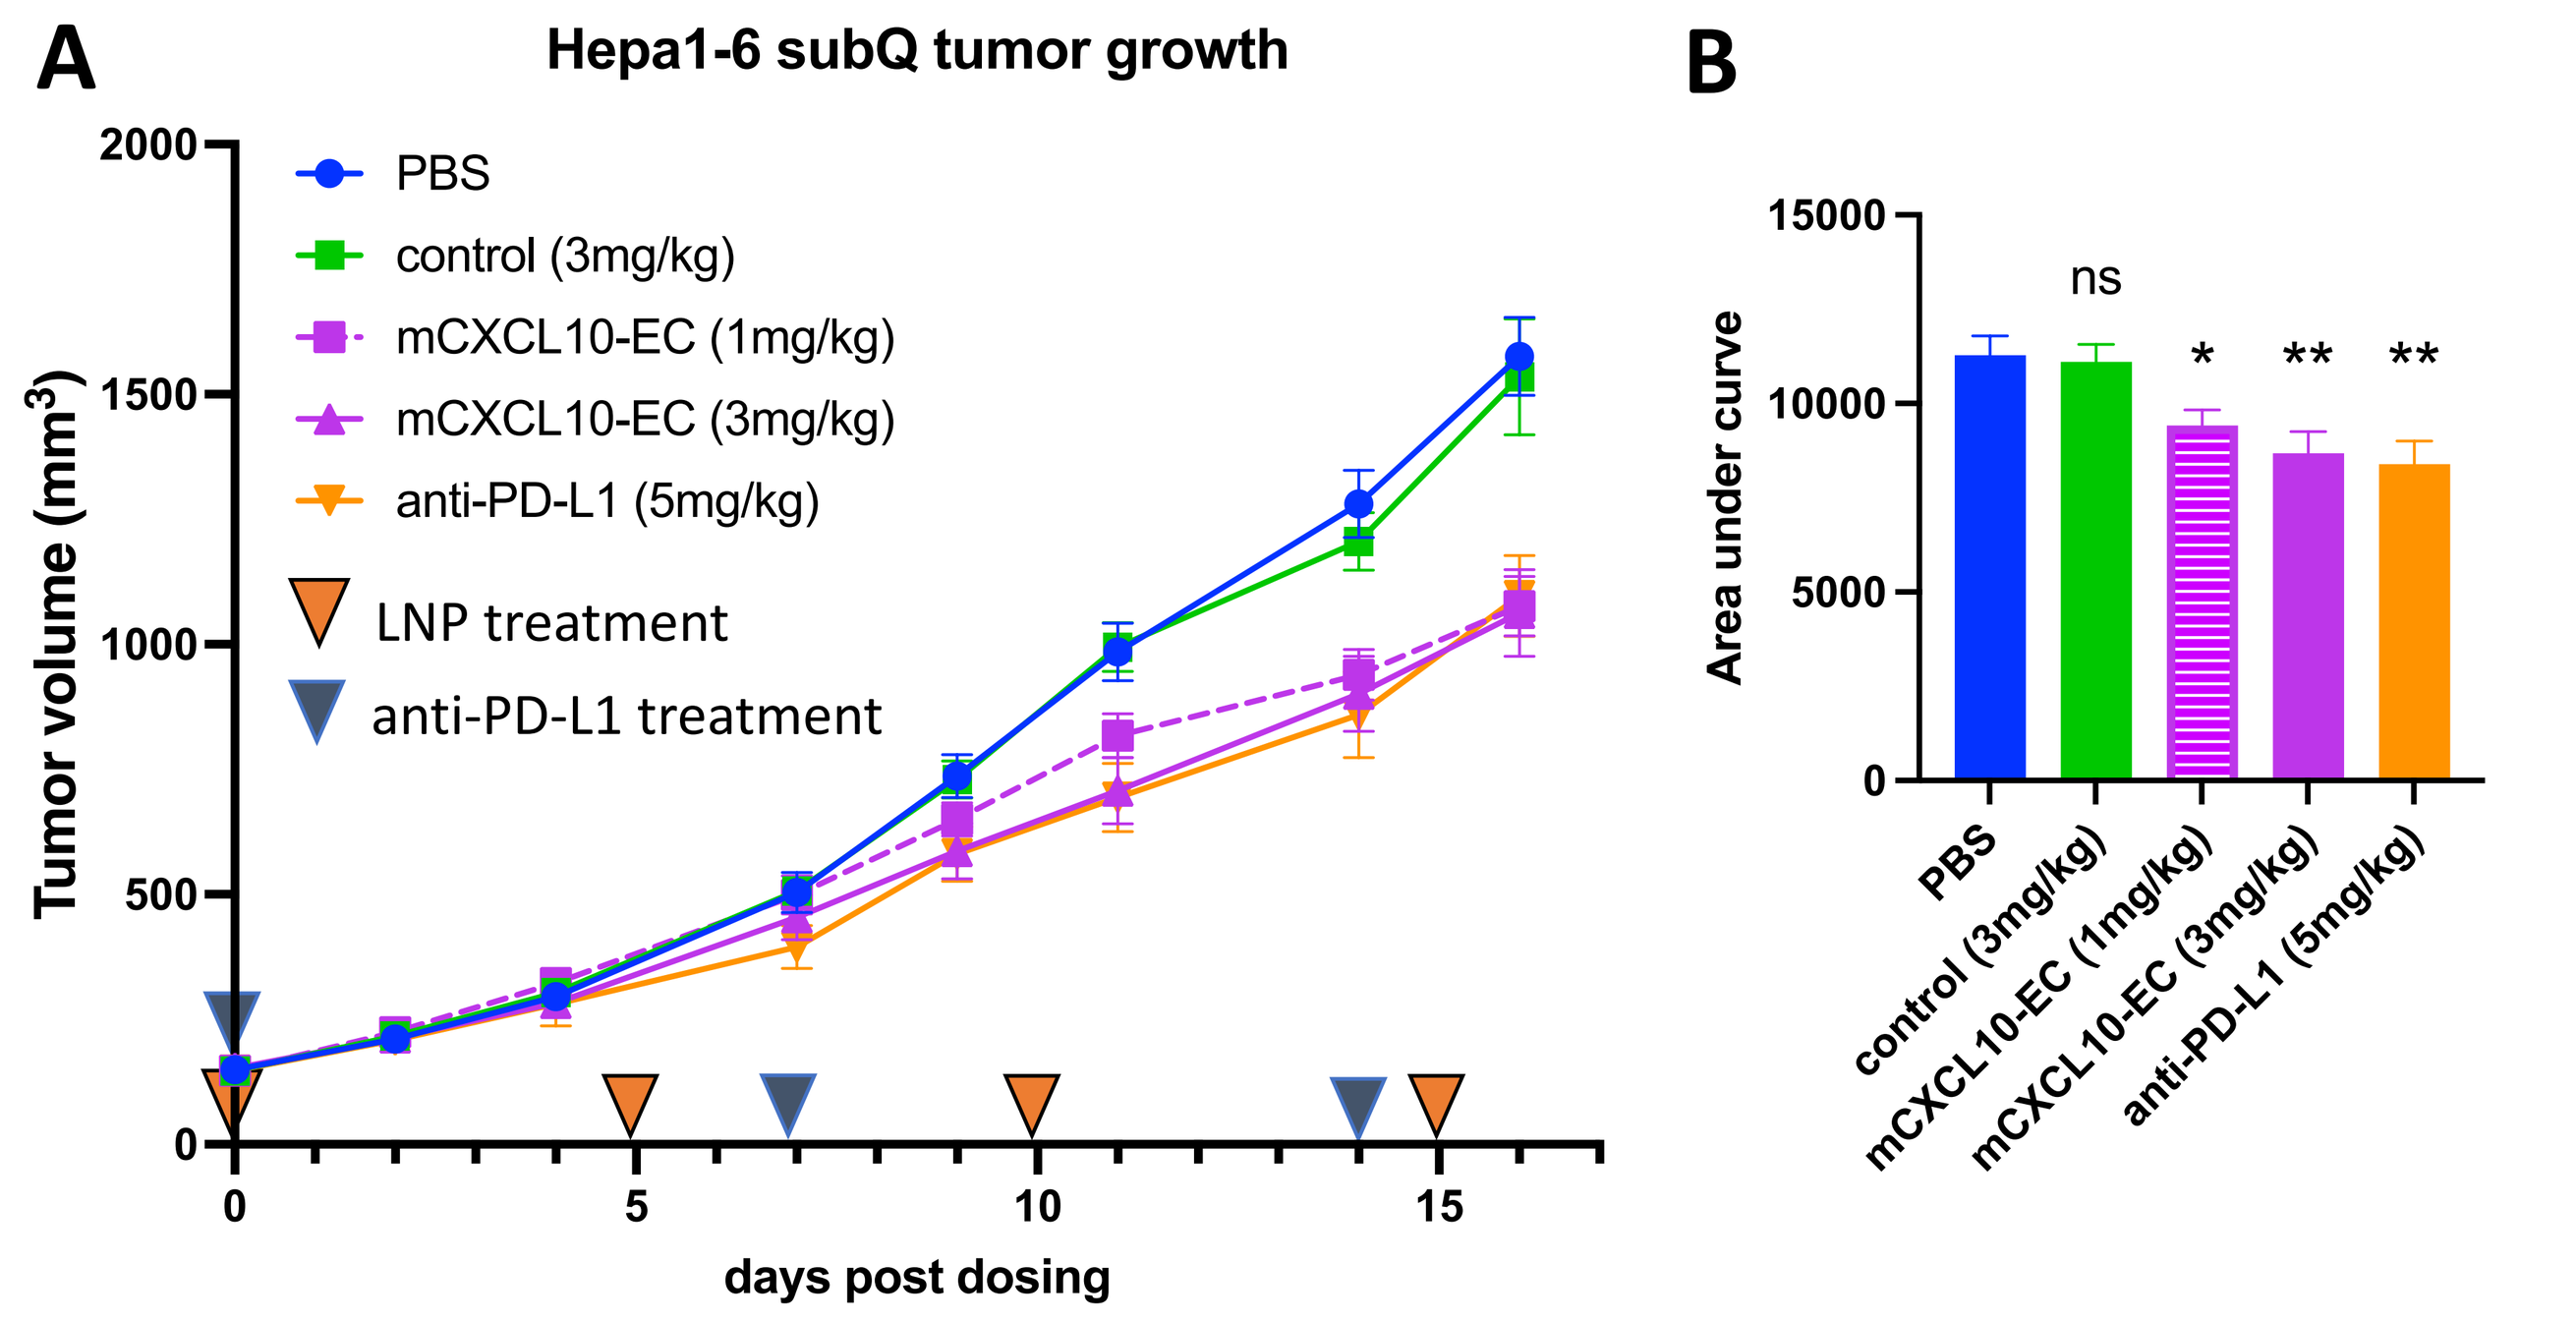
A
Hepa1-6 subQ tumor growth
Tumor volume (mm³)
days post dosing
PBS
control (3mg/kg)
mCXCL10-EC (1mg/kg)
mCXCL10-EC (3mg/kg)
anti-PD-L1 (5mg/kg)
LNP treatment
anti-PD-L1 treatment
B
Area under curve
ns
*
**
**

Figure 7: mCXCL10-EC treatment reduces growth of syngeneic mouse HCC tumors *in vivo.* (A) Mice bearing Hepa1-6 subcutaneous tumors were treated with PBS, 1 mg/kg of control mRNA-LNP, 1 mg/kg or 3 mg/kg of mCXCL10-EC-LNP, or 5 mg/kg of anti-PD-L1. LNPs were dosed Q5D and anti-PD-L1 QW. (B) Significant differences in the area under the curve between PBS and selected groups were analyzed using one-way ANOVA. '**' denotes $p < 0.01$, '*' denotes $p < 0.05$, and 'ns' denotes not significant.

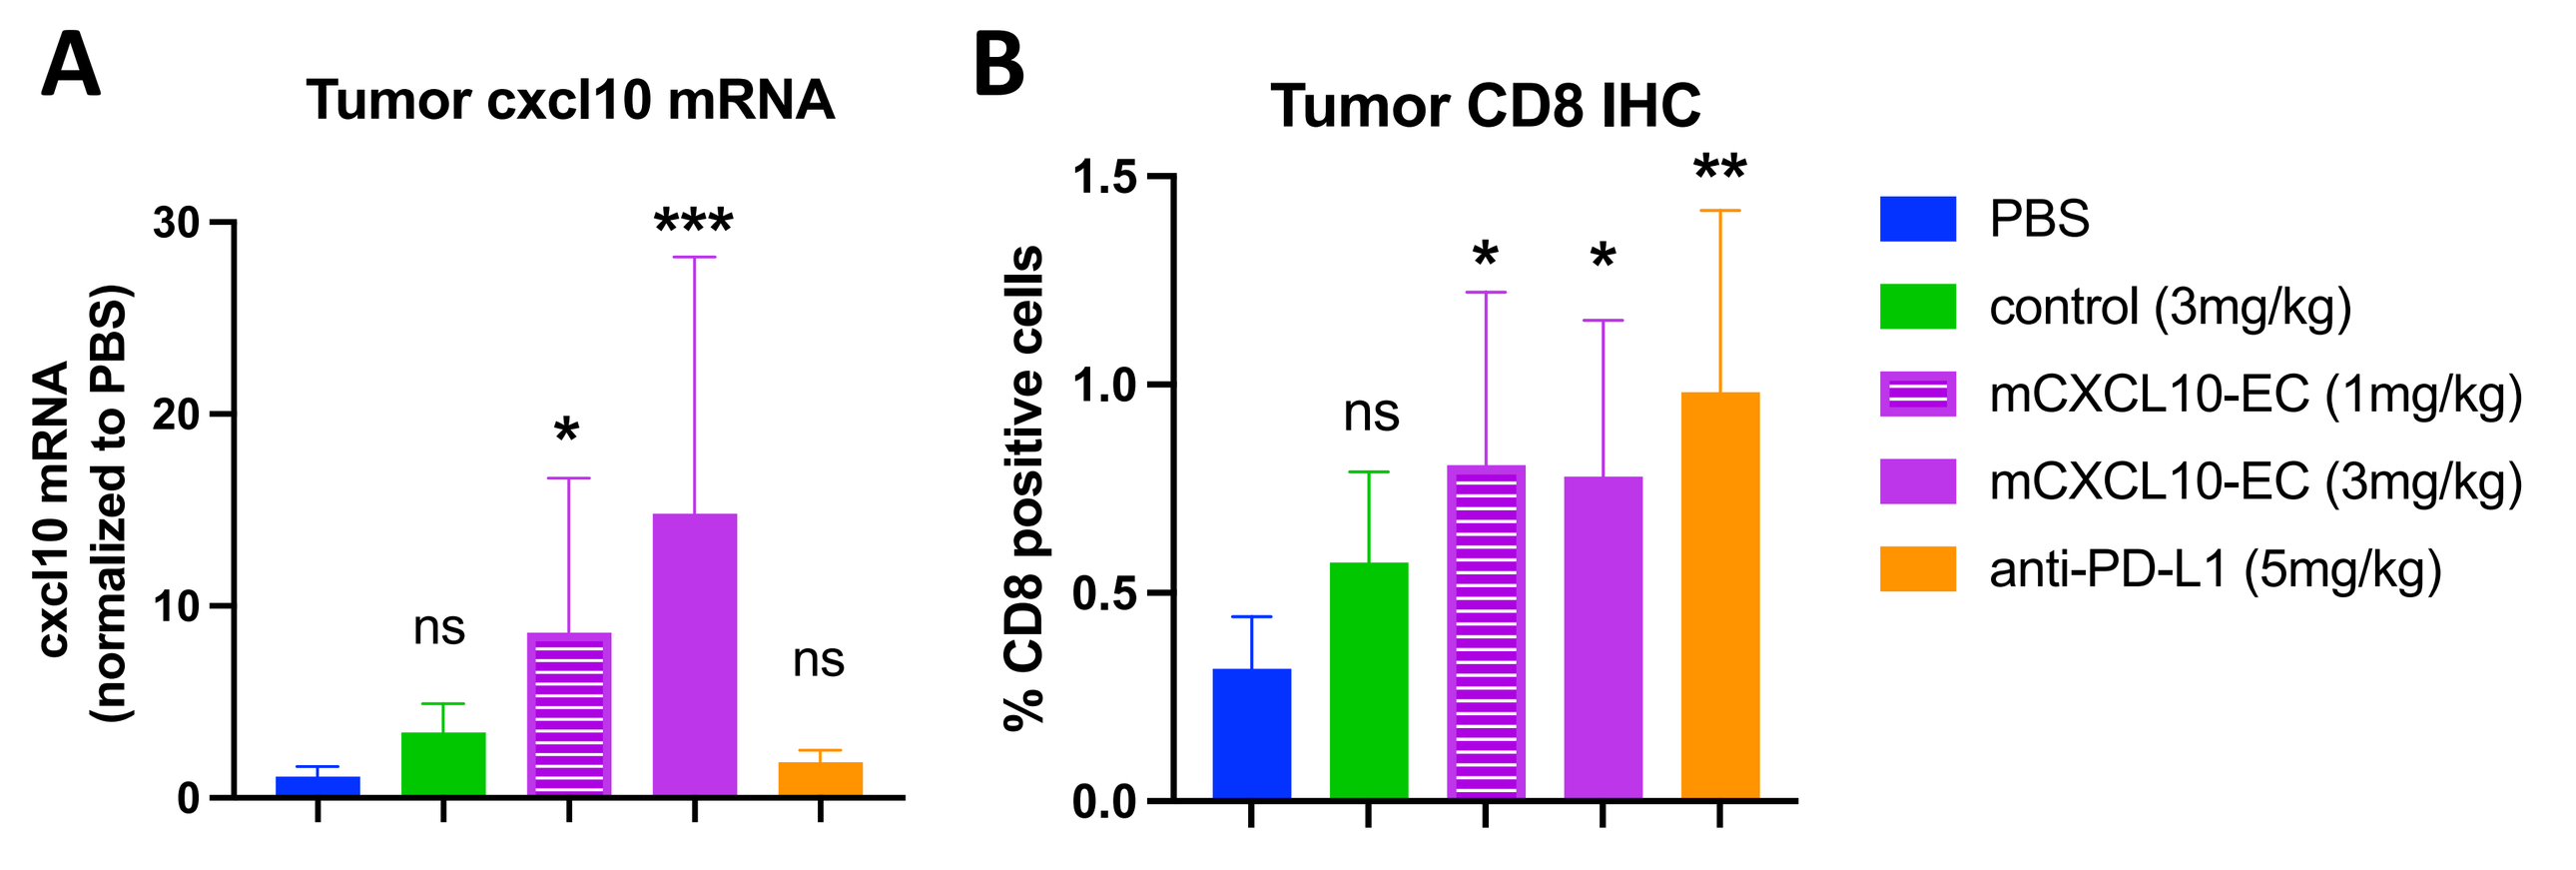
A
Tumor cxcl10 mRNA
cxcl10 mRNA (normalized to PBS)
B
Tumor CD8 IHC
% CD8 positive cells
PBS
control (3mg/kg)
mCXCL10-EC (1mg/kg)
mCXCL10-EC (3mg/kg)
anti-PD-L1 (5mg/kg)

Figure 8: mCXCL10-EC treatment upregulates cxcl10 expression and CD8+ T-cell infiltration in Hepa1-6 subcutaneous tumors. Tumors in Figure 7A were collected at 24 hours after 4th dose of LNP or 48 hours after 3rd dose of anti-PD-L1 and were subjected to qRT-PCR (A) or Immunohistochemistry (B). Significant differences between PBS and selected groups were analyzed using one-way ANOVA. (***) $p < 0.001$, (**) $p < 0.01$, (*) $p < 0.05$, (ns) not significant.

Conclusions

- Robust and tunable upregulation of genes of interest demonstrated by screening and choice of various epigenomic controllers
- Epigenomic upregulation of T-cell-recruiting chemokines CXCL9 and CXCL10 in human primary hepatocytes promotes T-cell recruitment *in vitro*
- Epigenomic upregulation of mouse cxcl10 inhibits tumor growth and enhances CD8+ T-cell tumor infiltration in syngeneic mouse HCC tumor xenografts in vivo
- These results provide preclinical proof-of-concept for epigenomic upregulation of T-cell attracting chemokines as a potential anti-tumor strategy and support continued evaluation of this approach through further EC characterization and mechanistic studies